Pegozafermin Treatment Ameliorates a Subclinical Increase in Spleen Volume, That Is Associated With a Worsening Metabolic Profile, in Non-cirrhotic NASH in Correlation With Change in Liver Fat and Inflammatory Markers in a Phase 1b/2a, Placebo-Controlled, Double-Blind Proof of Concept Study

JUAN FRIAS, MD[1], LARS JOHANSSON, PHD[2], R WILL CHARLTON, MD[3], SHIBAO FENG, PHD[3], GERMAINE D. AGOLLAH, PHD[3], HANK MANSBACH, MD[3], MAYA MARGALIT, MD[3]

[1]VELOCITY CLINICAL RESEARCH, LOS ANGELES, CA, USA [2]ANTAROS MEDICAL, MÖLNDAL, SWEDEN [3]89BIO INC., SAN FRANCISCO, CA, USA

INTRODUCTION

- FGF21 is an endogenous hormone that regulates carbohydrate, lipid, and energy metabolism. FGF21 analogs have demonstrated improvements in liver and metabolic abnormalities in non-alcoholic steatohepatitis (NASH).
- Pegozafermin (previously BIO89-100) is a glycoPEGylated FGF21 analog in development for treatment of NASH and SHTG.
- In addition to liver-related abnormalities, NASH is commonly associated with metabolic derangements, including insulin resistance and abnormal lipids.
- In a phase 1b/2a POC study in subjects with NASH, pegozafermin led to significant reductions in liver fat by MRI-PDFF and volume by MRI, with concurrent metabolic benefits and a favorable safety and tolerability profile.

BACKGROUND

- Portal venous pressure may begin to rise due to steatosis-induced changes in sinusoidal homeostasis in early stages of non-alcoholic fatty liver disease (NAFLD), which may contribute to progression of fibrosis and portal hypertension.[1]
- Emerging data suggest spleen elastography may be helpful in detection of liver fibrosis stage and portal hypertension in patients with NAFLD.
- A correlation between spleen elastography[1] and spleen volume (SV) assessments and hepatic venous pressure gradient has previously been demonstrated.
- However, whether an association exists between SV and extra-hepatic metabolic parameters has not been systematically assessed.

[1]Colecchia A et al, *Gastroenterology* 2012; 143: 646.

OBJECTIVE

The objective of this post-hoc sub-study was to examine the effect of pegozafermin (PGZ) vs placebo on SV and assess potential correlations of SV with metabolic disarrangements in subjects with NASH.

METHODS

Double-Blind, Placebo-Controlled, Phase 1B/2A Multiple Ascending Dose Study (NCT4048135)

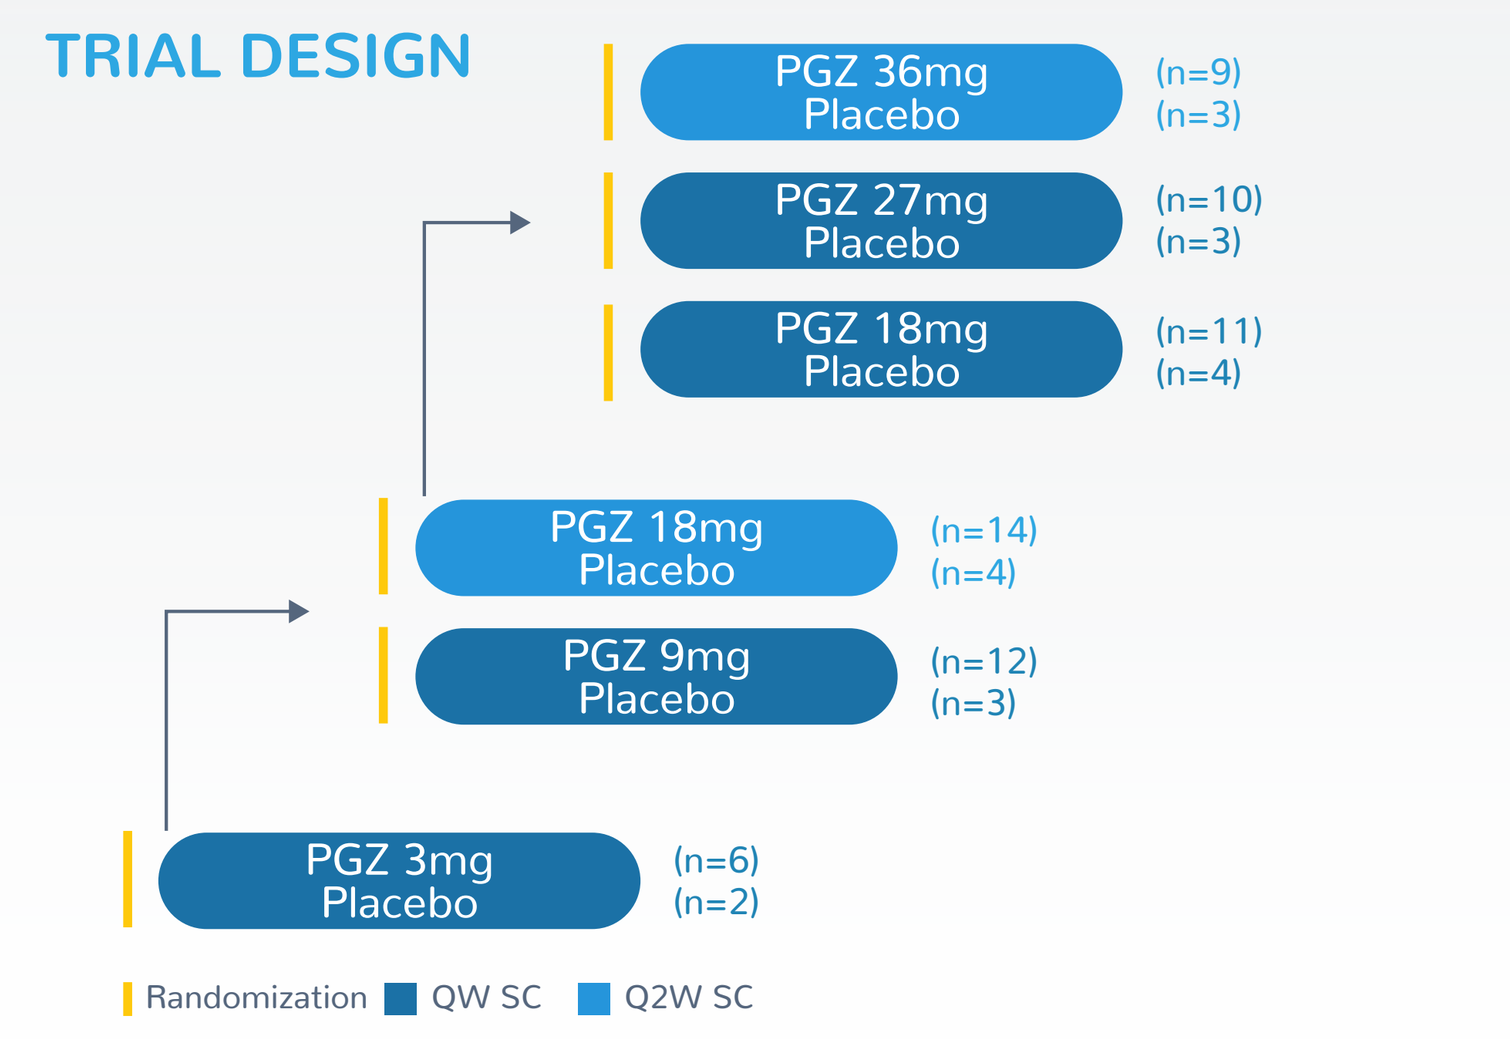
TRIAL DESIGN
PGZ 36mg Placebo (n=9) (n=3)
PGZ 27mg Placebo (n=10) (n=3)
PGZ 18mg Placebo (n=11) (n=4)
PGZ 18mg Placebo (n=14) (n=4)
PGZ 9mg Placebo (n=12) (n=3)
PGZ 3mg Placebo (n=6) (n=2)
Randomization QW SC Q2W SC

KEY INCLUSION CRITERIA

- NASH* or phenotypic NASH (PNASH)#
- PDFF ≥10%

*Subjects with biopsy-proven F1-F3
#Central obesity plus T2DM or evidence of liver injury

KEY TRIAL ENDPOINTS

- Safety, PK
- Relative changes in liver fat
- Serum lipids, liver, and metabolic markers

- 12-week treatment duration + 4-week safety follow-up
- Placebo (n=19) combined across cohorts for analysis

- Randomized, pharmacodynamic (PD) and safety analysis set n=81; study completers n=71
- MRI analysis set=75 (subjects with post-baseline MRI)

METHODS

Spleen Volume Post Hoc Analysis

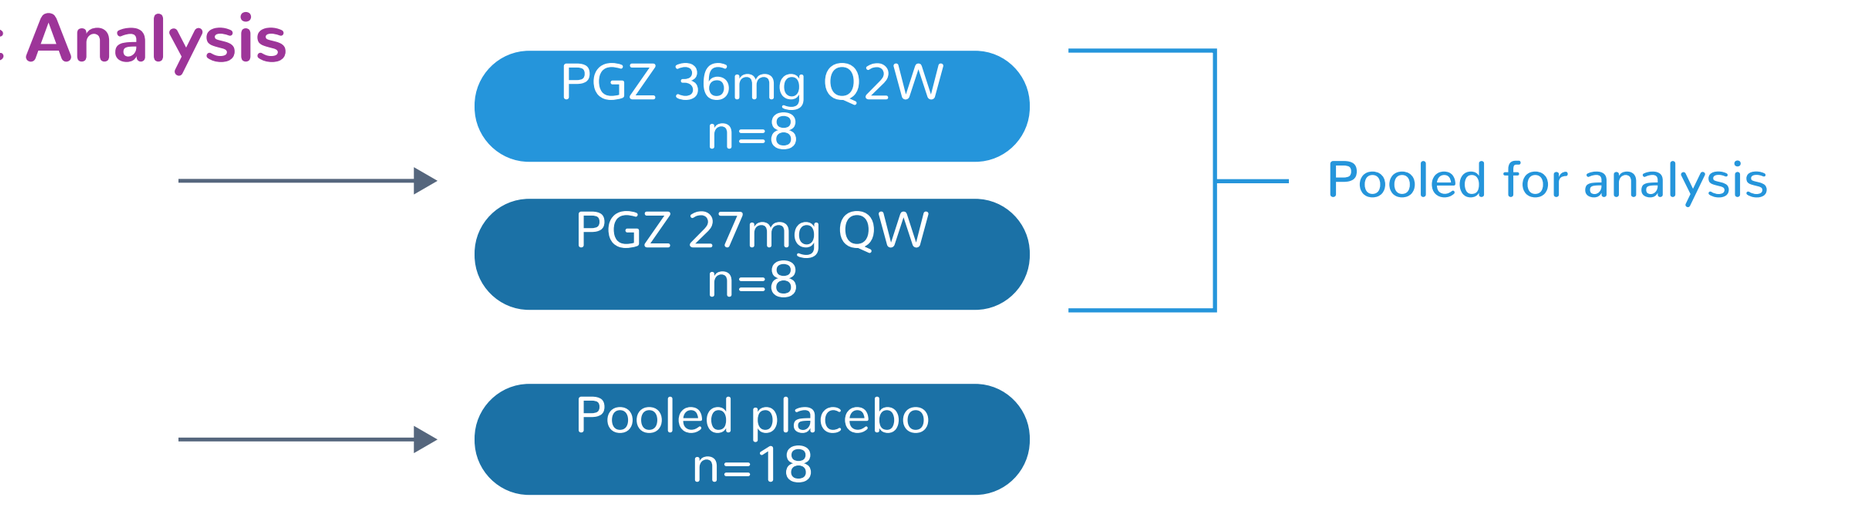
PGZ 36mg Q2W n=8
PGZ 27mg QW n=8
Pooled for analysis
Pooled placebo n=18

- SV was assessed by MRI at Baseline, Day 50, and Day 92.
- Correlation of baseline SV and change in SV to various clinical and lab parameters was investigated.

RESULTS

Baseline Characteristics

CHARACTERISTICS	PLACEBO (n=18) MEDIAN [RANGE]	PEGOZAFERMIN (n=16) MEDIAN [RANGE]
Age (years)	56.5 [37.4, 66.3]	47.6 [37.7, 66.6]
Male gender (%)	38.9	56.3
T2DM (%)	61%	25%
BMI	33.5 [29.5, 39.8]	36.6 [28.8, 44.8]
MRI-PDFF (%)	19.7 [10.5, 39.5]	19.3 [12.1, 41.1]
Fibroscan VCTE Score (kPa)	7.05 [2.9, 12.0]	8.0 [4.0, 11.4]
Liver volume (L)	2.1 [1.30, 2.83]	2.4 [1.5, 3.5]
Spleen volume (mL)*	170.9 [107.6, 366.9]	232.6 [137.6, 504.4]
ALT (U/L)	29 [14, 95]	53 [17, 178]
CK-18 (U/L)	113 [39, 1078]	179 [39, 1548]
Platelet count (baseline)	265.5 [184, 339]	245 [188, 328]
HDL-C (mg/dL)	44.8 [31.0, 60.5]	38.8 [26.5, 76.0]
HOMA-IR	10.9 [1.5, 26.1]	13.8 [5.1, 24.5]
ADIPO-IR	10.1 [4.8, 42.5]	13.2 [5.1, 39.1]
Adiponectin (ug/mL)	4.6 [1.2, 17.1]	3.8 [0.8, 10.8]

*Within normal range; Linguraru et al. *Acad Radiol* 2013; 20(6): 675

Baseline Spleen Volume Correlates With Liver Volume, VCTE Score, BMI, and Platelet Count

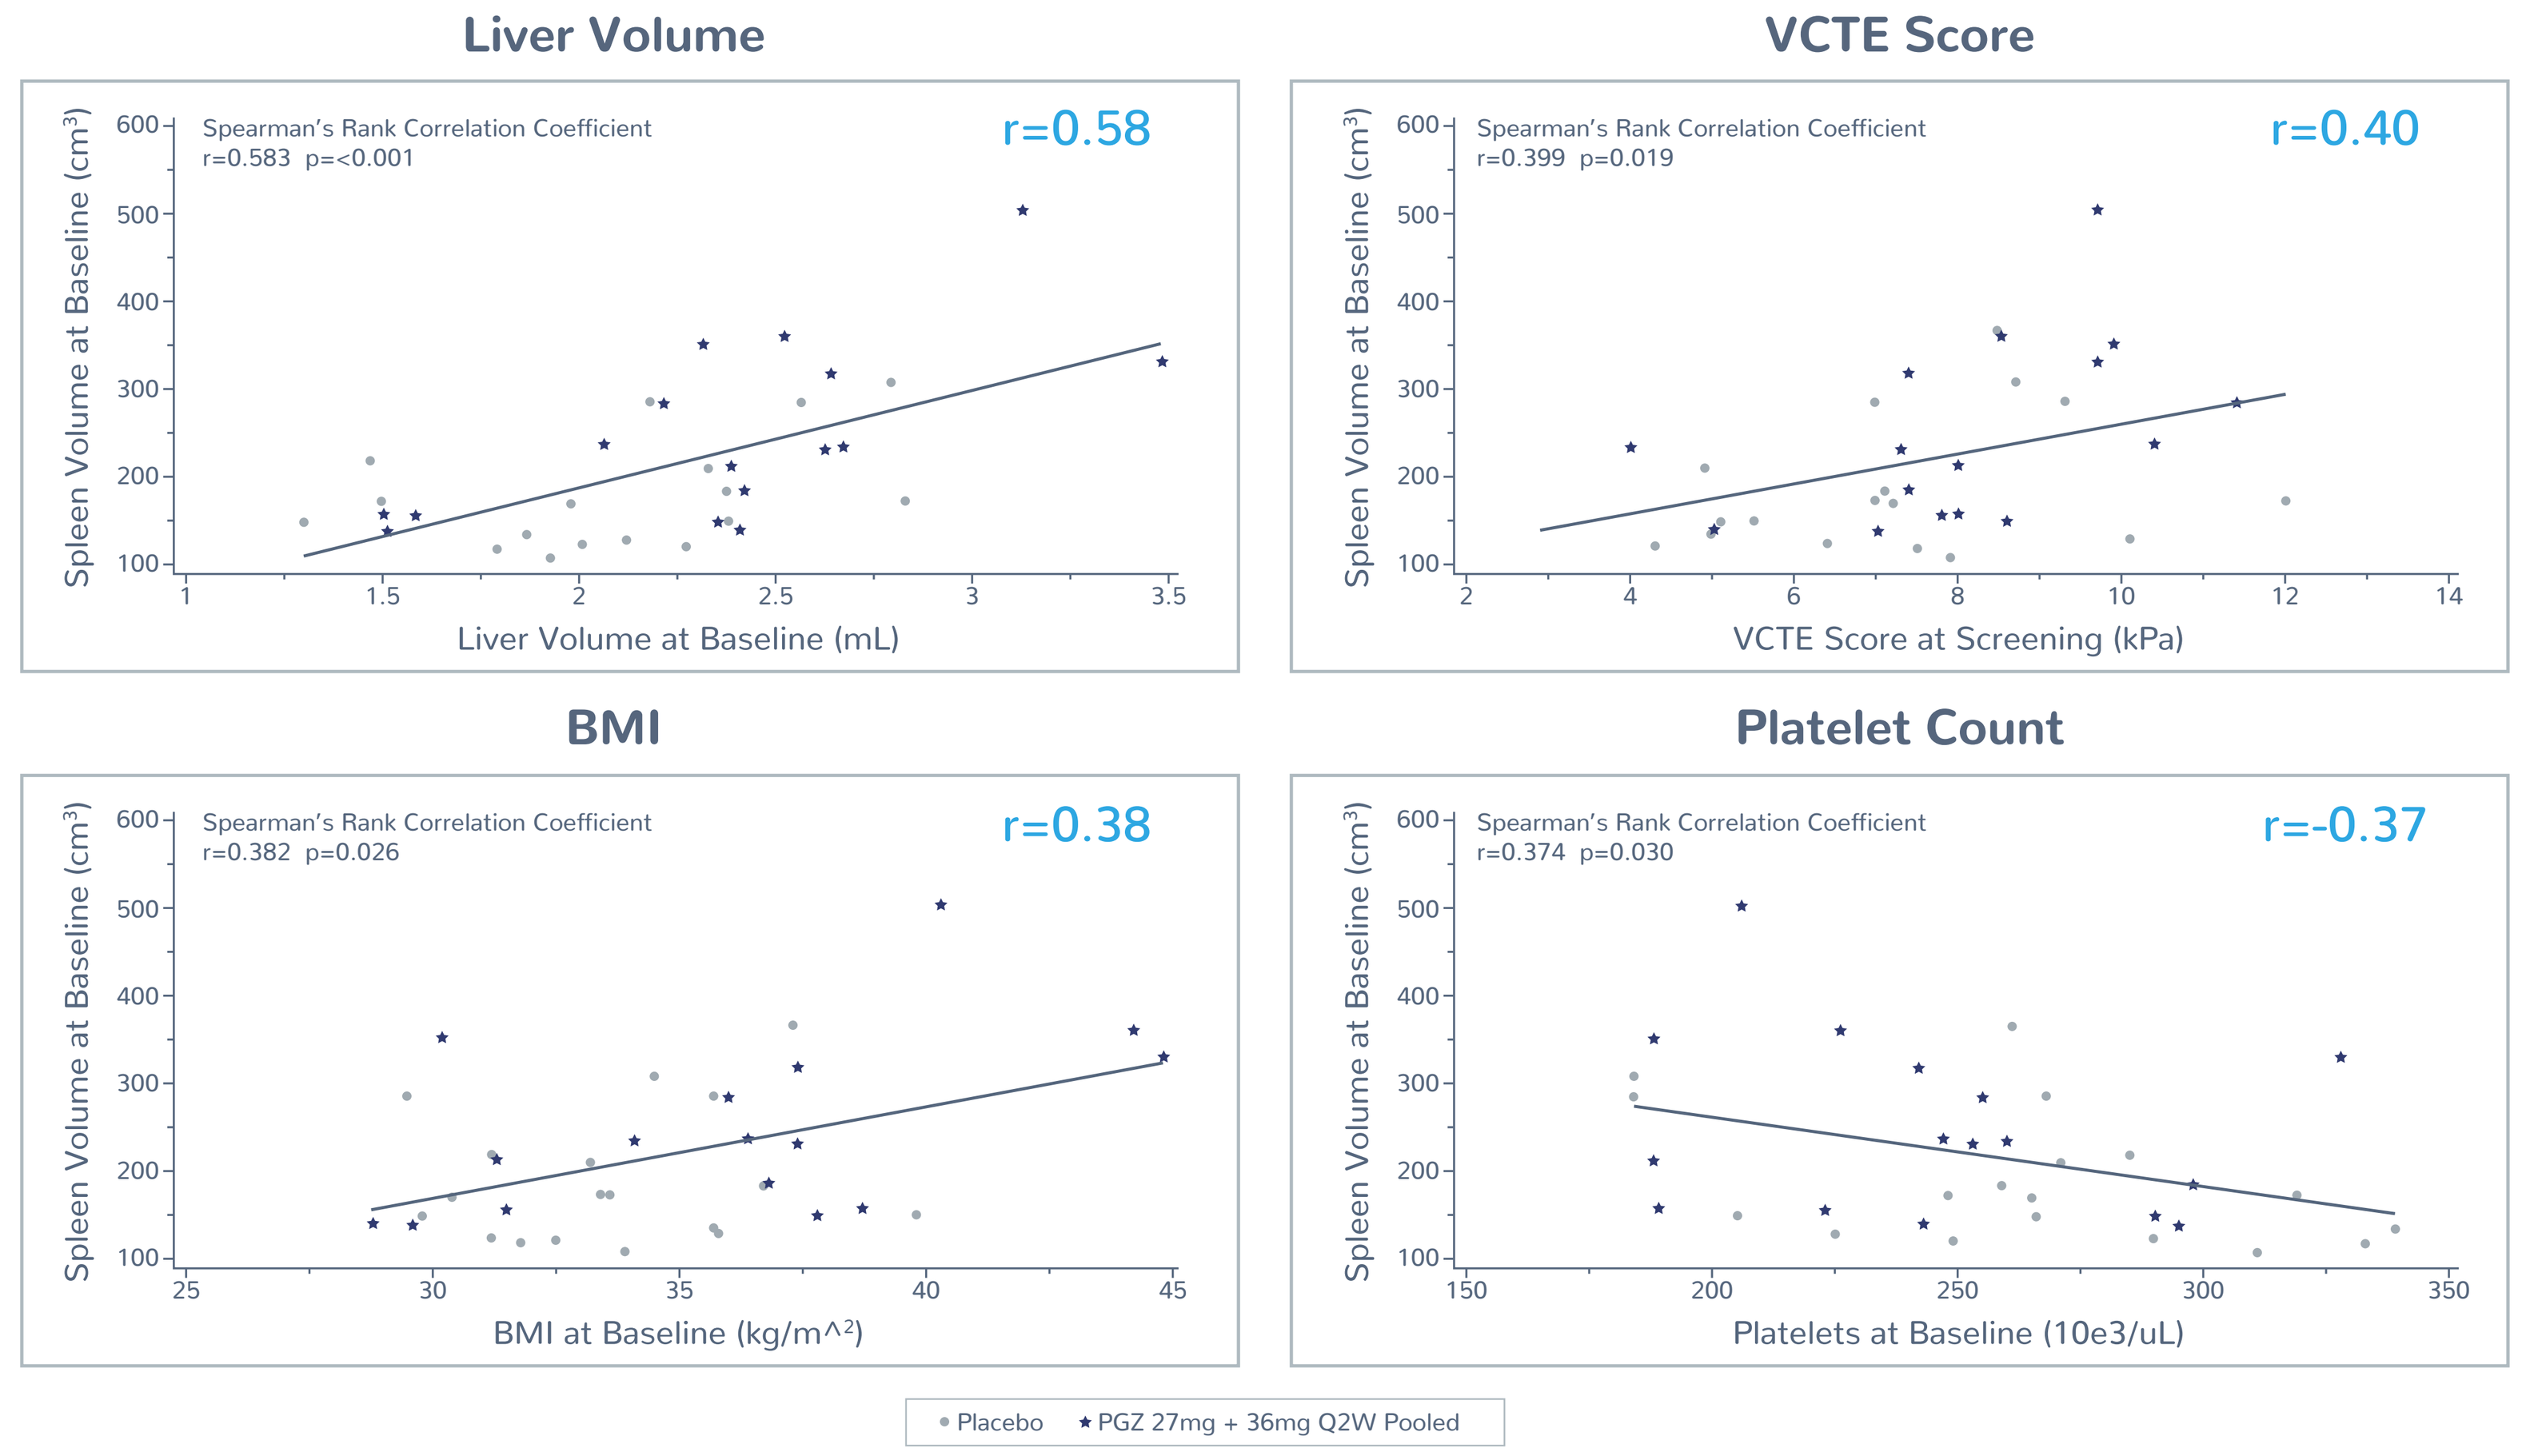
Liver Volume
Spearman's Rank Correlation Coefficient r=0.583 p=<0.001
r=0.58
Spleen Volume at Baseline (cm³)
Liver Volume at Baseline (mL)
VCTE Score
Spearman's Rank Correlation Coefficient r=0.399 p=0.019
r=0.40
VCTE Score at Screening (kPa)
BMI
Spearman's Rank Correlation Coefficient r=0.382 p=0.026
r=0.38
BMI at Baseline (kg/m^2)
Platelet Count
Spearman's Rank Correlation Coefficient r=0.374 p=0.030
r=-0.37
Platelets at Baseline (10e3/uL)
Placebo PGZ 27mg + 36mg Q2W Pooled

RESULTS

Baseline Spleen Volume Correlates With HOMA-IR, Adipo-IR, Adiponectin and HDL-cholesterol

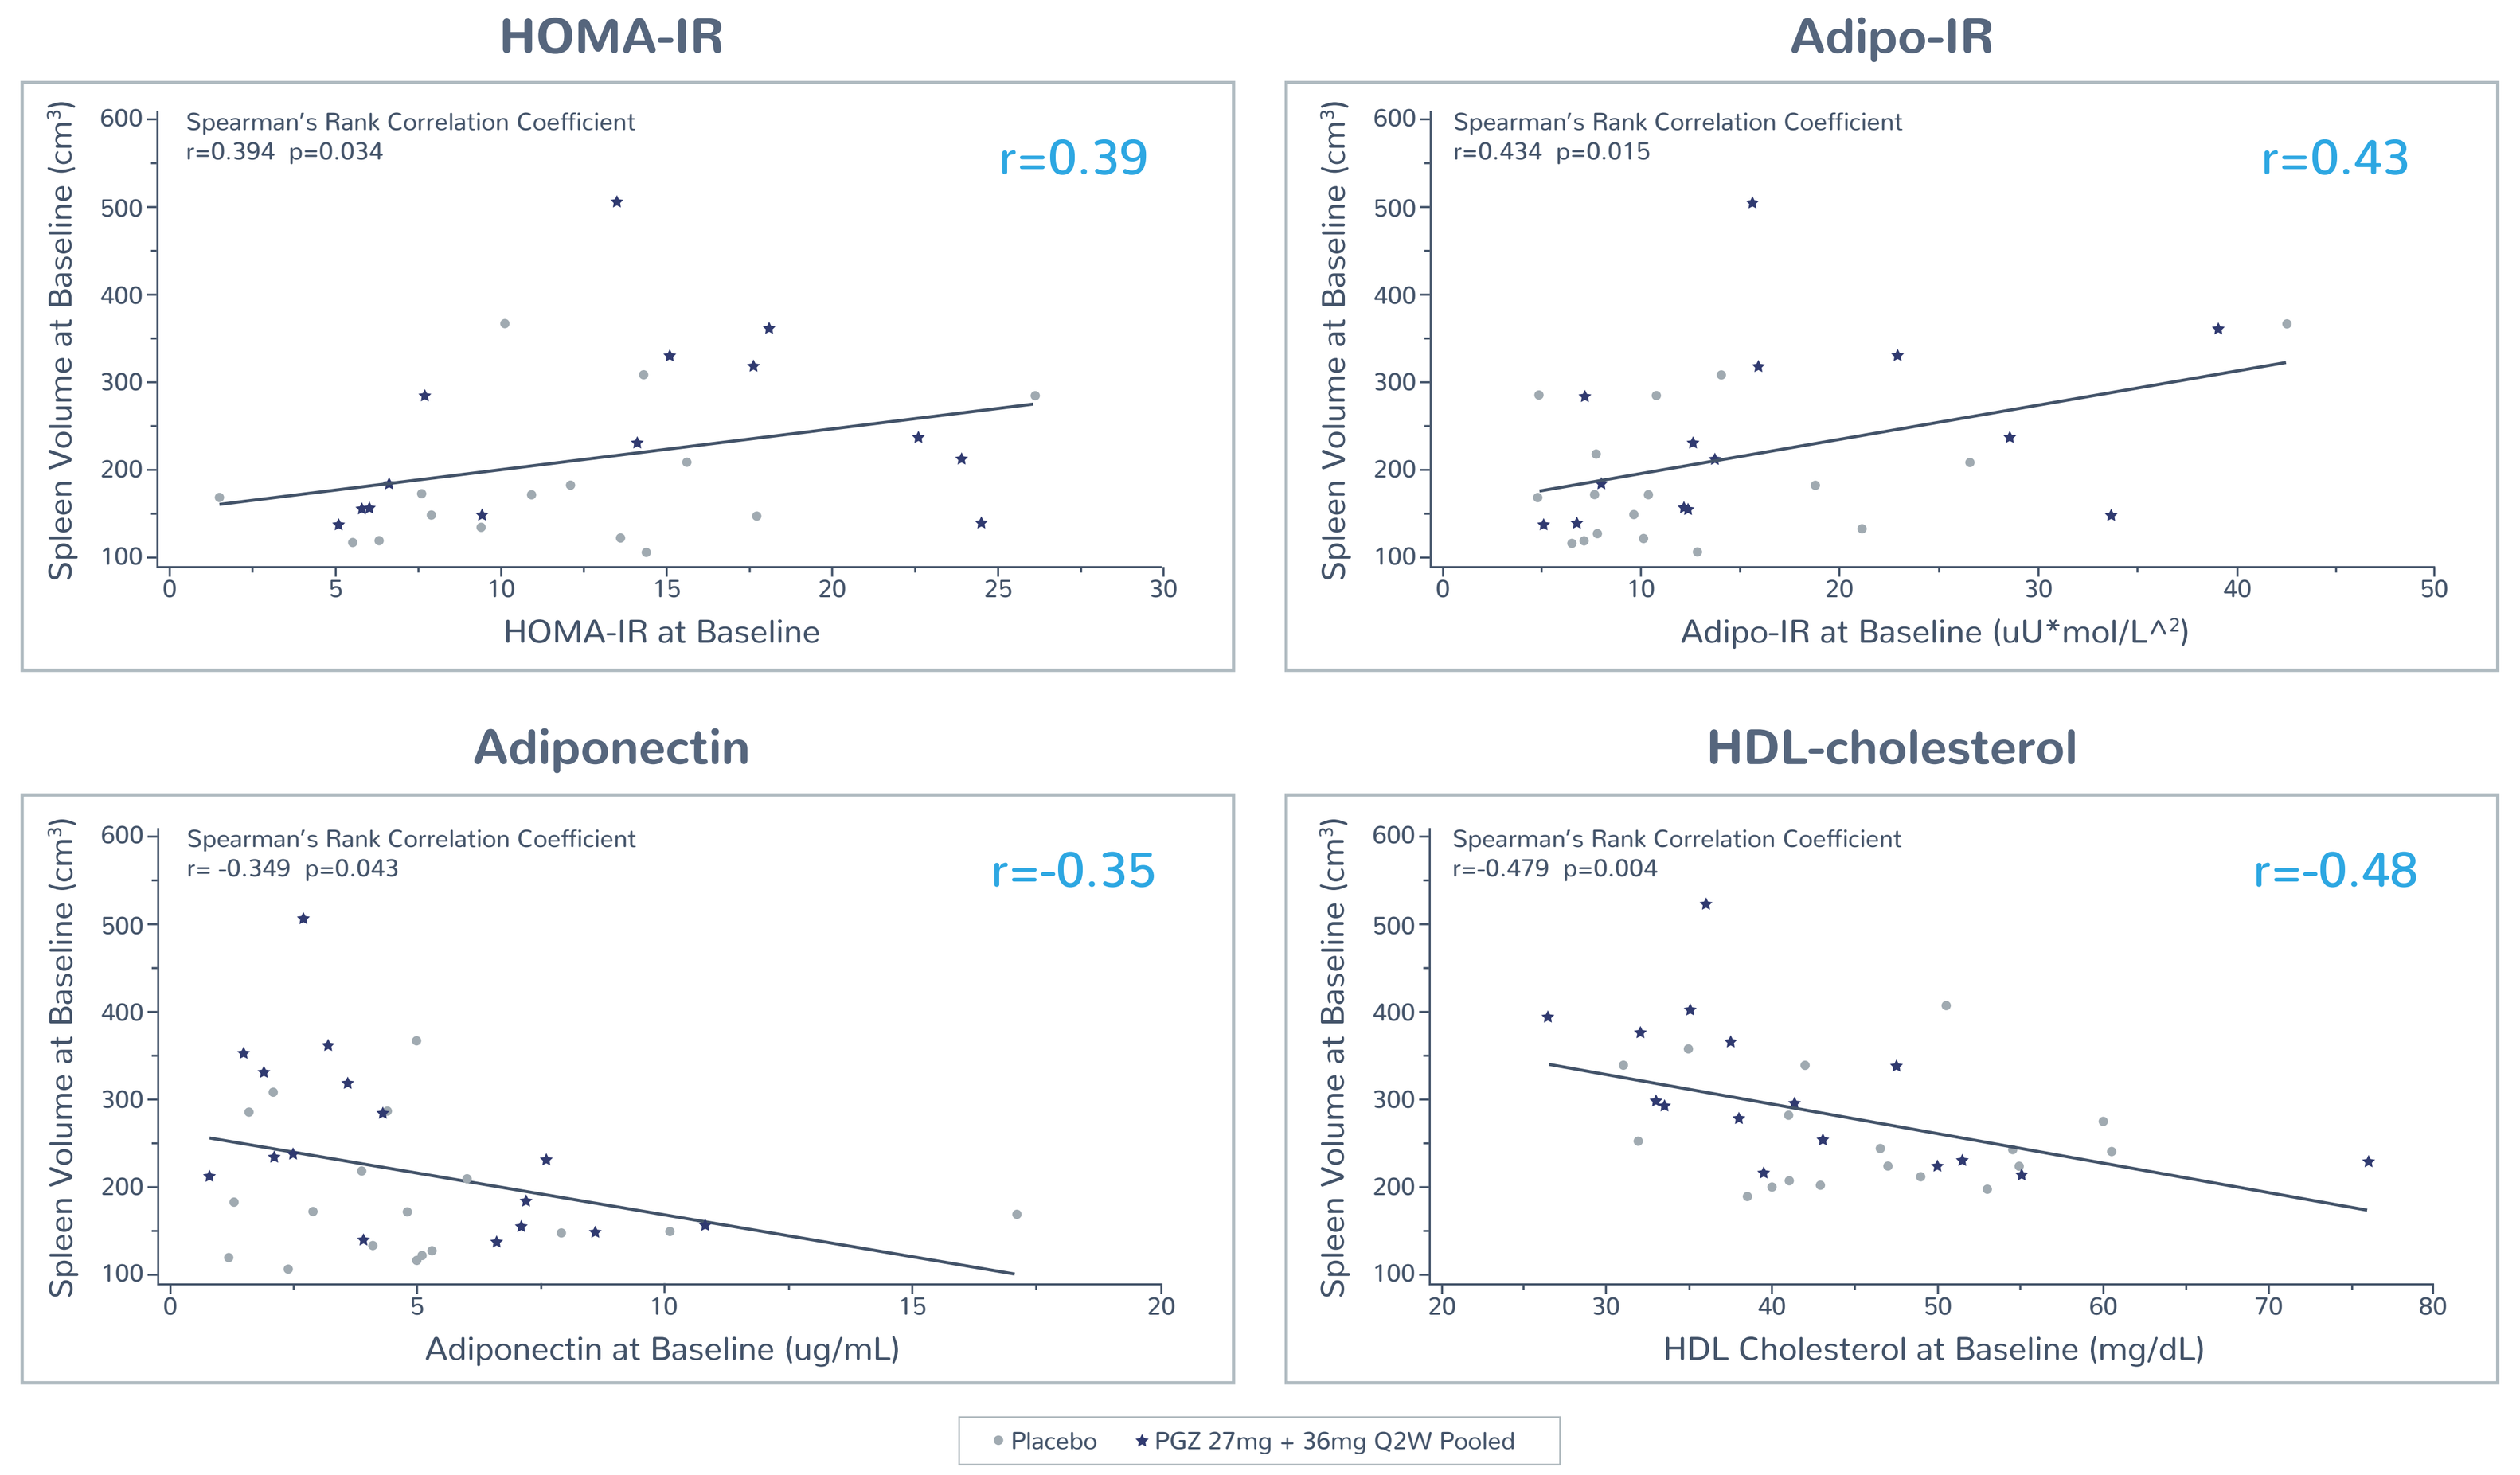
HOMA-IR
Spearman's Rank Correlation Coefficient r=0.394 p=0.034
r=0.39
Spleen Volume at Baseline (cm³)
HOMA-IR at Baseline
Adipo-IR
Spearman's Rank Correlation Coefficient r=0.434 p=0.015
r=0.43
Adipo-IR at Baseline (uU*mol/L^2)
Adiponectin
Spearman's Rank Correlation Coefficient r= -0.349 p=0.043
r=-0.35
Adiponectin at Baseline (ug/mL)
HDL-cholesterol
Spearman's Rank Correlation Coefficient r=-0.479 p=0.004
r=-0.48
HDL Cholesterol at Baseline (mg/dL)
Placebo PGZ 27mg + 36mg Q2W Pooled

Significant Reduction in Spleen Volume With Pegozafermin

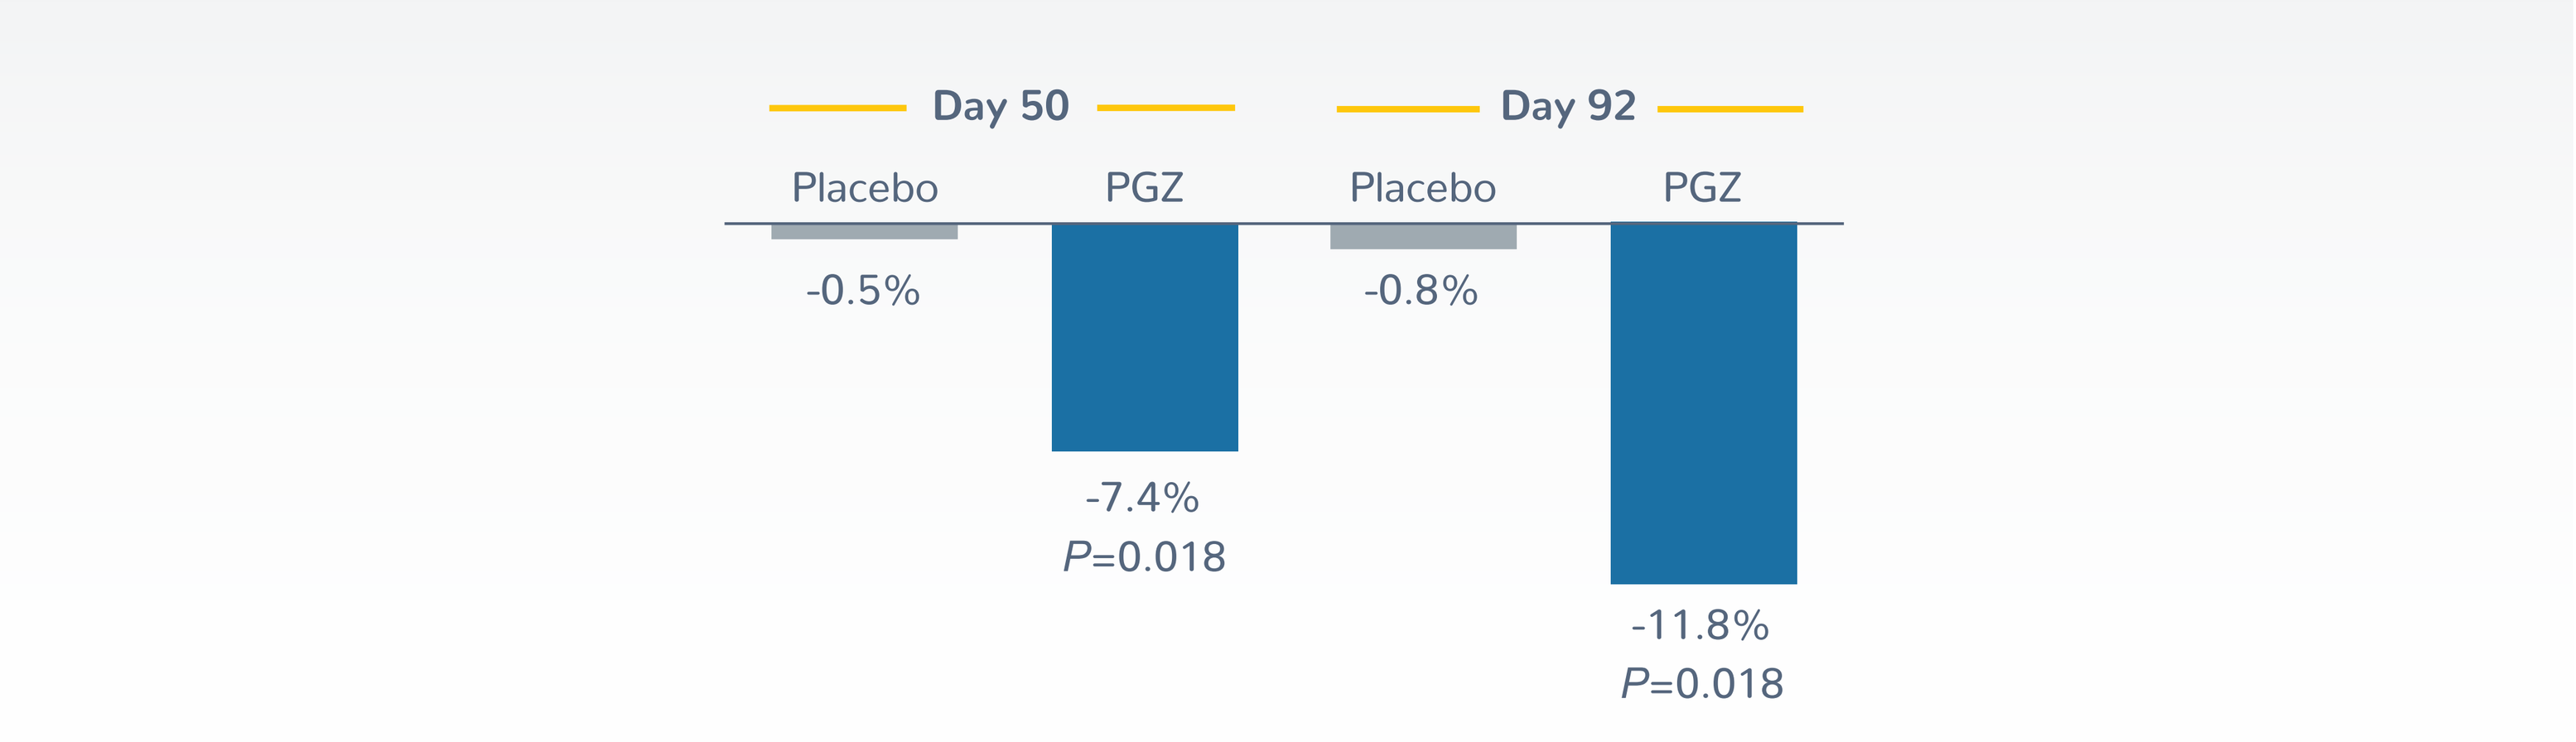
Day 50
Day 92
Placebo
PGZ
Placebo
PGZ
-0.5%
-7.4%
P=0.018
-0.8%
-11.8%
P=0.018

Percent changes from baseline to D50 and D92 were estimated using ANCOVA model adjusted for baseline SV.

Spleen Volume Change Correlated to Change in Liver Fat and Liver Fat Volume at Day 92

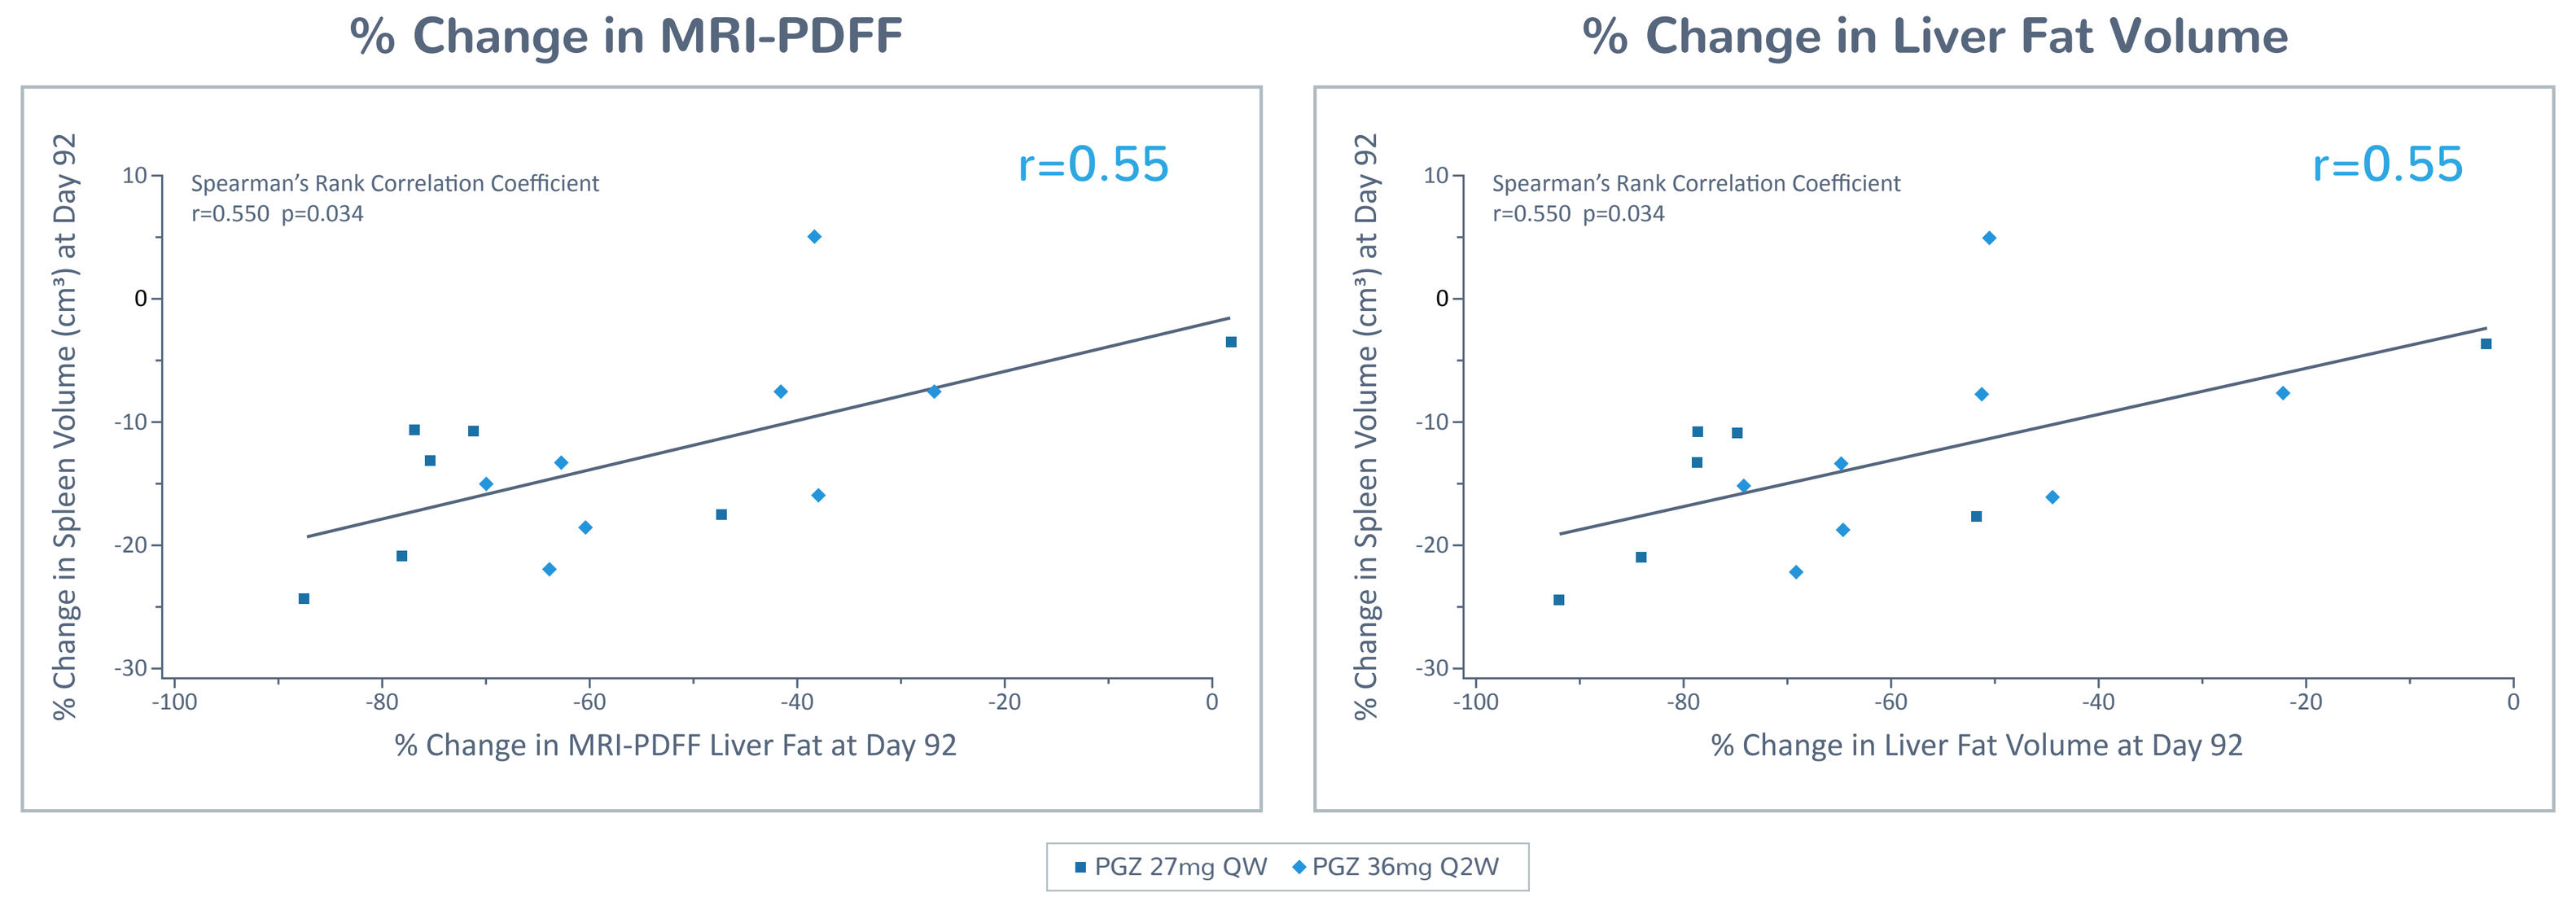
% Change in MRI-PDFF
Spearman's Rank Correlation Coefficient r=0.550 p=0.034
r=0.55
% Change in Spleen Volume (cm³) at Day 92
% Change in MRI-PDFF Liver Fat at Day 92
% Change in Liver Fat Volume
Spearman's Rank Correlation Coefficient r=0.550 p=0.034
r=0.55
% Change in Liver Fat Volume at Day 92
PGZ 27mg QW PGZ 36mg Q2W

Spleen Volume Change Correlated With Change of CK-18 and ALT and Negatively Correlated With Platelet Count at Day 92

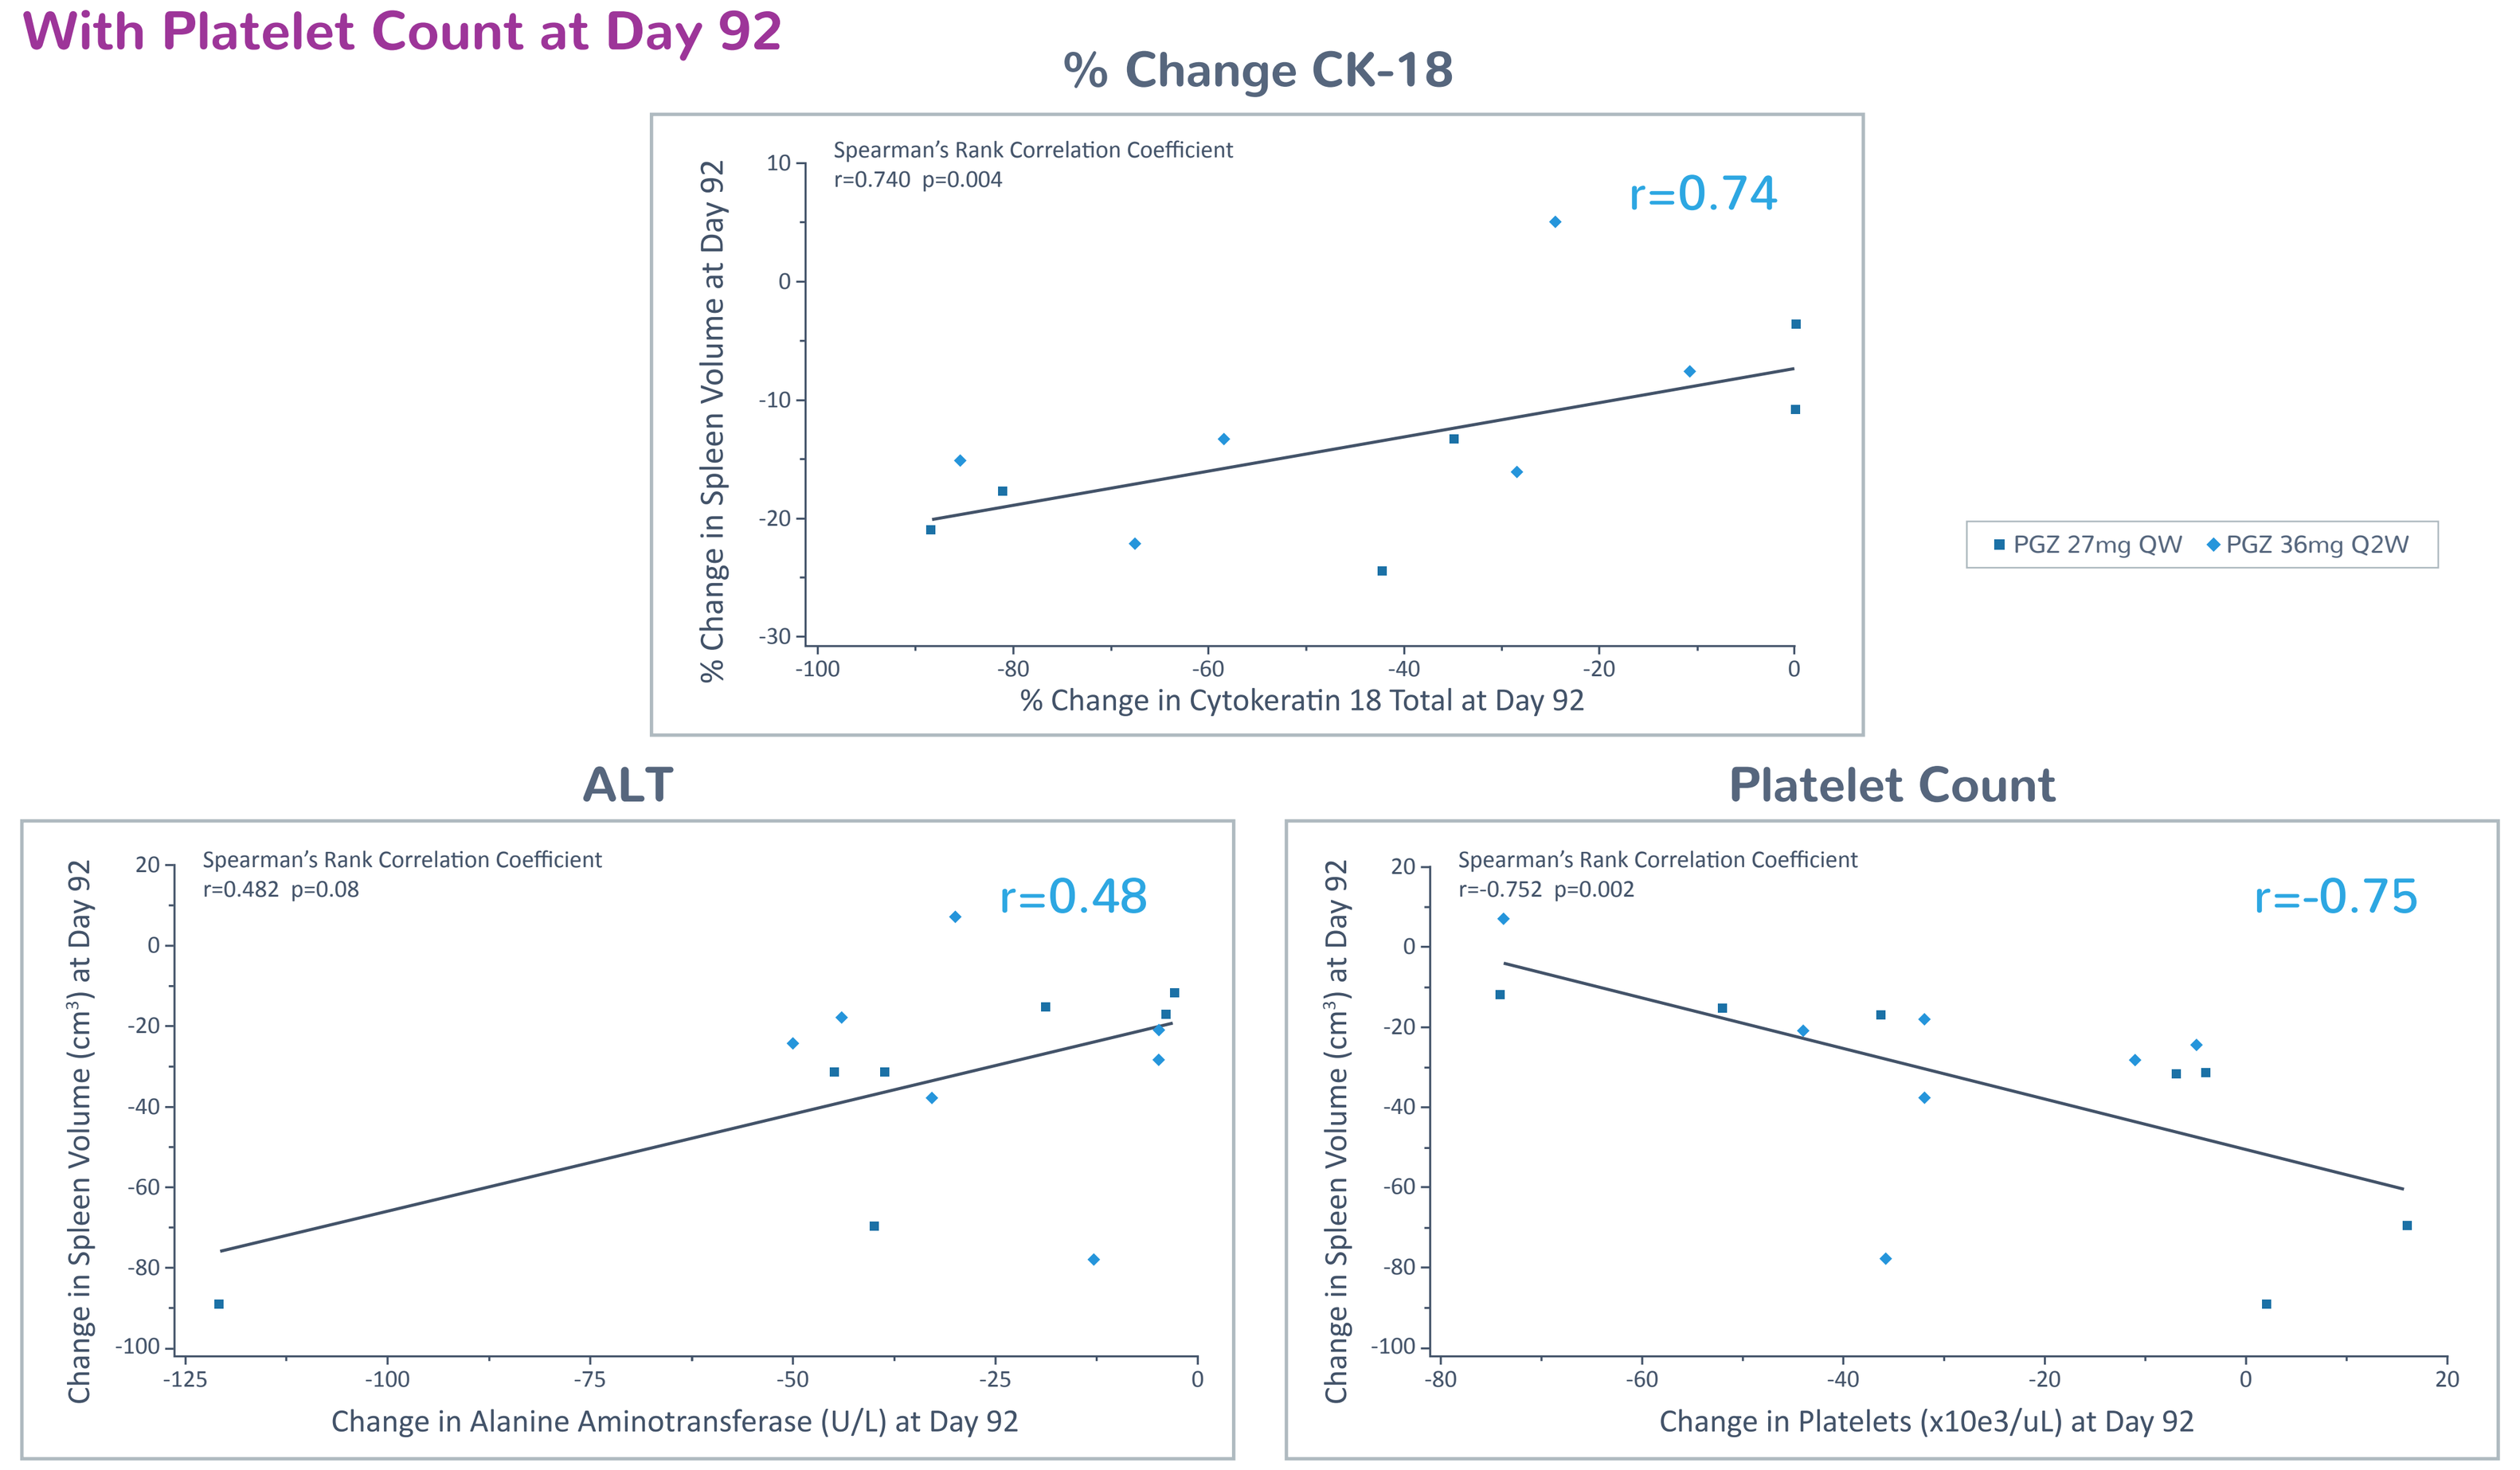
% Change CK-18
Spearman's Rank Correlation Coefficient r=0.740 p=0.004
r=0.74
% Change in Spleen Volume at Day 92
% Change in Cytokeratin 18 Total at Day 92
PGZ 27mg QW PGZ 36mg Q2W
ALT
Spearman's Rank Correlation Coefficient r=0.482 p=0.08
r=0.48
Change in Spleen Volume (cm³) at Day 92
Change in Alanine Aminotransferase (U/L) at Day 92
Platelet Count
Spearman's Rank Correlation Coefficient r=-0.752 p=0.002
r=-0.75
Change in Platelets (x10e3/uL) at Day 92

Reduction in SV at Day 92 appeared not to be correlated with change in VCTE score, ELF or Pro-C3 level, or change in metabolic parameters

Post Hoc Analysis of NASH POC Study Demonstrated Meaningful Reduction in Spleen Volume That Correlated With Common NASH Biomarkers

- SV at baseline was correlated with liver volume, VCTE score, BMI, HOMA-IR, and Adipo-IR, and negatively correlated with platelet count, adiponectin, and HDL-cholesterol.
 - This baseline association of increased SV with both liver-related and metabolic derangements strengthens the position of a NAFLD-related phenomenon.
- Treatment with pegozafermin led to a progressive, statistically significant decrease in SV compared to placebo.
- SV reduction correlated with reductions in liver fat by MRI-PDFF, liver fat volume, CK-18 and ALT, and was negatively correlated with platelet count.
- Limitations include post-hoc analysis, small sample size, and short treatment duration.

CONCLUSIONS

- These preliminary data suggest that in non-cirrhotic NAFLD, a subclinical increase in SV is associated with a worsening metabolic profile, increased liver stiffness and decreased platelet count.
- Reversal of NAFLD by normalization of liver fat and liver volume may be associated with improved portal flow, thus contributing to decreases in spleen volume.
- Further mechanistic studies are needed to confirm this hypothesis.
- These data suggest a potential role for monitoring of SV in assessing treatment response in NASH clinical trials. The utility of SV as a non-invasive tool warrants further investigation.

©2022 89bio, Inc.