28773

89bio

Population Pharmacokinetics (PK) and Pharmacodynamics (PD) of BIO89-100, a Novel GlycoPEGylated FGF21, in a Phase 1b/2a POC Study in Nonalcoholic Steatohepatitis (NASH)

LEO TSENG[1], R WILL CHARLTON[1], MAYA MARGALIT[1], HANK MANSBACH[1], RADA M SAVIC[2]

[1]89bio, Inc.,Clinical Development, 6 Hamada street Herzliya, Israel, and 242 Sansome Street, San Francisco, CA, USA; [2]Department of Bioengineering and Therapeutic Sciences, UCSF.

INTRODUCTION

- BIO89-100 is a long-acting glycoPEGylated analogue of fibroblast growth factor 21 (FGF21) in development for the treatment of NASH and severe hypertriglyceridemia.
- In a Phase 1b/2a placebo-controlled, double-blind, multiple ascending dose study in patients with NASH, administration of BIO89-100 resulted in clinically meaningful reductions in liver fat content (LFC), with a favorable safety and tolerability profile.
- PK and PD data from this study were analyzed using a population modelling approach to characterize the relationship between BIO89-100 dosing, exposure, and effects on LFC reduction.
 – The primary goal of this analysis was to identify an optimal dose range for the Phase 2b (ENLIVEN) trial of this promising molecule in patients with NASH.

MATERIAL AND METHODS

Source Trial Data

- A multicenter, randomized, double-blind, placebo-controlled, Phase 1b/2a multiple ascending dose study over 12 weeks was conducted in 81 patients with biopsy confirmed NASH (BC-NASH) or phenotypic NASH (PNASH) and a magnetic resonance imaging-derived proton density fat fraction (MRI-PDFF) of at least 10%.

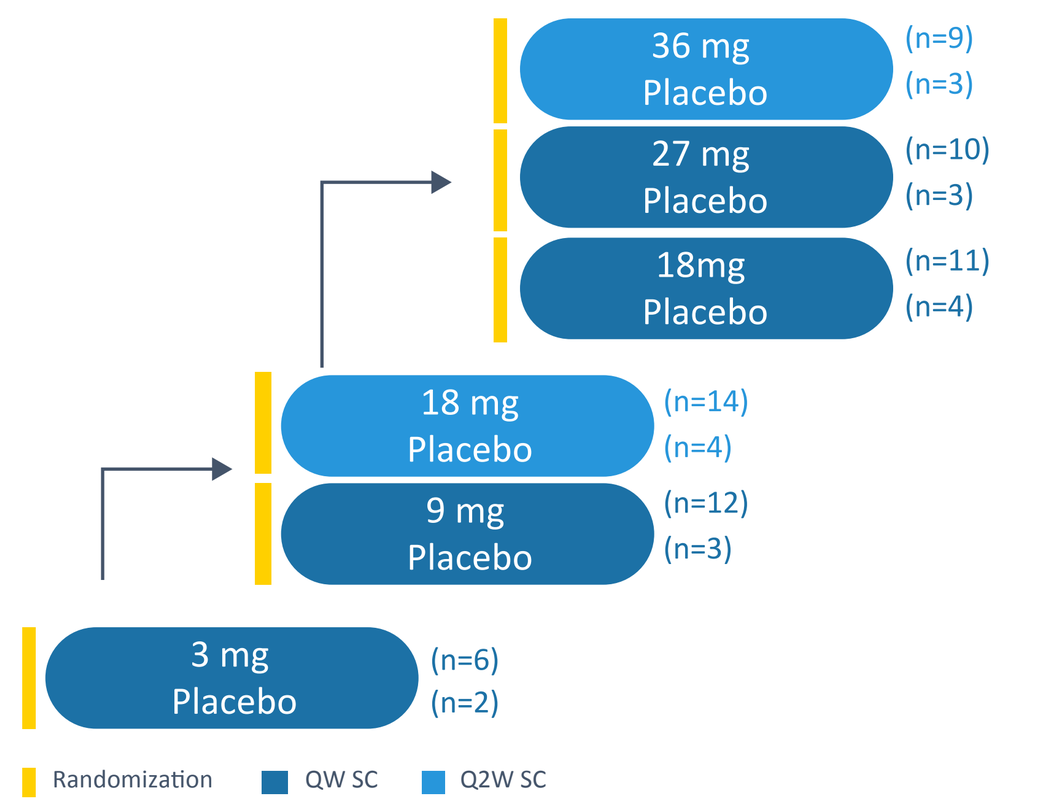
36 mg
Placebo
(n=9)
(n=3)
27 mg
Placebo
(n=10)
(n=3)
18mg
Placebo
(n=11)
(n=4)
18 mg
Placebo
(n=14)
(n=4)
9 mg
Placebo
(n=12)
(n=3)
3 mg
Placebo
(n=6)
(n=2)
Randomization
QW SC
Q2W SC

Clinical trial study design (ClinicalTrials.gov Identifier: NCT04048135). QW, once a week; Q2W, once every 2 weeks.
Phenotypic NASH (was defined as at least one of the following criteria: 1) obesity with T2DM (ie, fasting plasma glucose ≥126 mg/dL, plasma glucose in a 75-g oral glucose tolerance test ≥200 mg/dL, or glycated hemoglobin ≥6.5 %); 2) obesity with evidence of liver injury (either increased ALT [≥40 U/L in males or ≥30 U/L in females] and/or FibroScan vibration-controlled transient elastography score ≥7 KPa); T2DM, Type 2 diabetes mellitus; U, unit; ULN, upper limit of normal.

Exposure-Response Relationship and PK/PD Modeling

- Pooled PK and PD data from all patients receiving BIO89-100 (N = 62) were characterized using a population nonlinear mixed-effects modeling approach and NONMEM 7.4.2 software.
- Exposure (average concentration, C_{avg}) and Response (MRI-PDFF effect at Day 92) relationship were assessed using both logistic regression and E_{max} model.
- Longitudinal PK/PD model:
 – Several model structures for PK disposition (one or two compartment) and various models for the absorption (first order, first order with a lag time, and transit compartment model) were evaluated.
 – PK/PD relationship was described as: $PDFF(t)=Baseline - \frac{(E_{max}\times Cp(t))}{(Cp(t)+EC50)}$
 – Covariate effect was evaluated after final PK/PD model was established.

RESULTS

TABLE 1: BASELINE CHARACTERISTICS PK/PD POPULATION

PARAMETER (MEAN OR PERCENTAGE)	POOLED BIO89-100 (N=62)	3 mg QW (N=6)	9 mg QW (N=12)	18 mg QW (N=11)	27 mg QW (N=10)	18 mg Q2W (N=14)	36 mg Q2W (N=9)
Age (years)	51.7	56.1	49.5	51.5	52	51.2	52.5
Male (%)	38.7	16.7	50	27.3	20	28.6	88.9
Weight (kg)	93.6	87.9	87.2	87.1	94	101.5	101.1
BMI (kg/m²)	34.8	34.3	32.7	32.8	36.8	37	34.8
Type 2 diabetes (%)	40.3	83.3	33.3	63.6	40	21.4	22.2
ALT (U/L)	42.3	45	32.8	38.4	53.3	39.1	50.4
AST (U/L)	31.5	34.5	22.8	30.9	39	28.8	38.1
MRI-PDFF (%)	21.2	22.4	21.4	19.3	22	21.6	20.9
Triglyceride (mg/dL)	174	135	177	226	157	137	207

ALT, alanine aminotransferase; AST, aspartate transaminase; BMI, body-mass index; MRI-PDFF, magnetic resonance imaging derived proton density fat fraction; QW, weekly; Q2W, every 2 weeks.

- Baseline characteristics of key parameters were similar across the dosing groups.

RESULTS

FIGURE 1: EXPOSURE-RESPONSE RELATIONSHIP AS DESCRIBED BY LOGISTIC REGRESSION MODEL

Logistic Regression of MRI-PDFF on Day 92

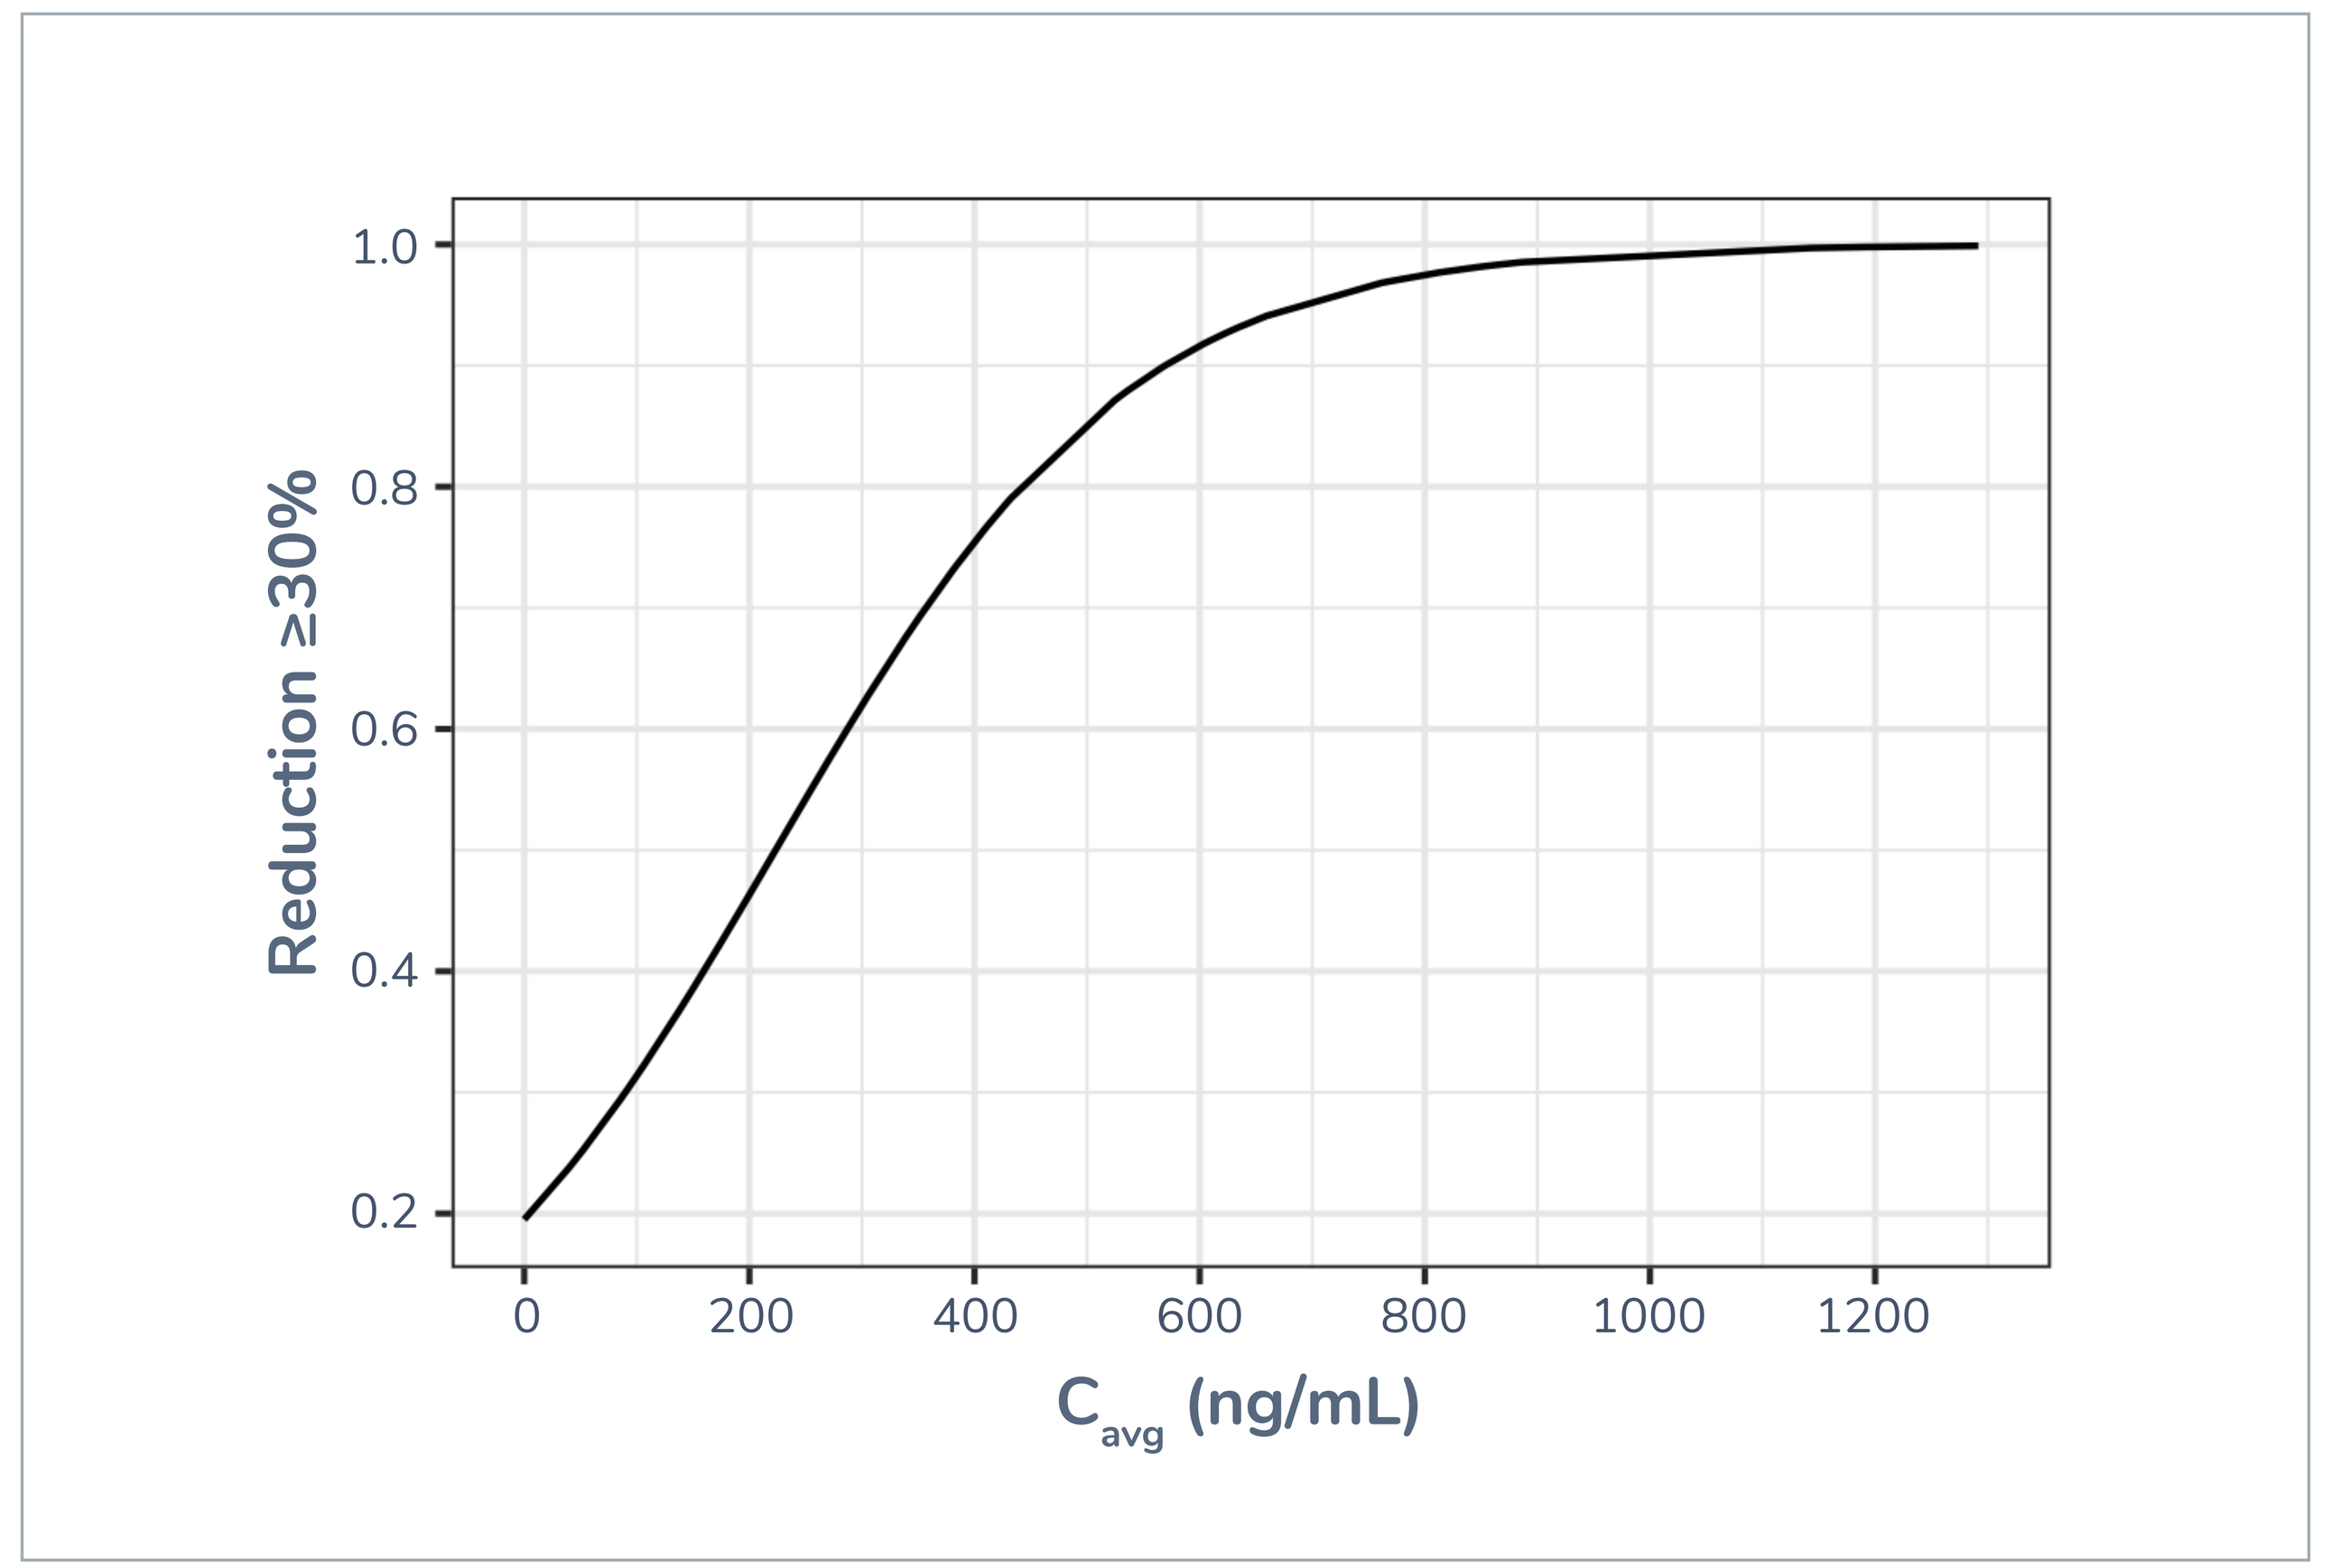
Reduction ≥30%
1.0
0.8
0.6
0.4
0.2
0
200
400
600
800
1000
1200
Cavg (ng/mL)

- Full PD effect, defined as all subjects with ≥30% LFC reduction from baseline, can be reached at approximately C_{avg} ≥800 ng/mL.

FIGURE 2: EXPOSURE-RESPONSE AS DESCRIBED BY E_{MAX} MODEL

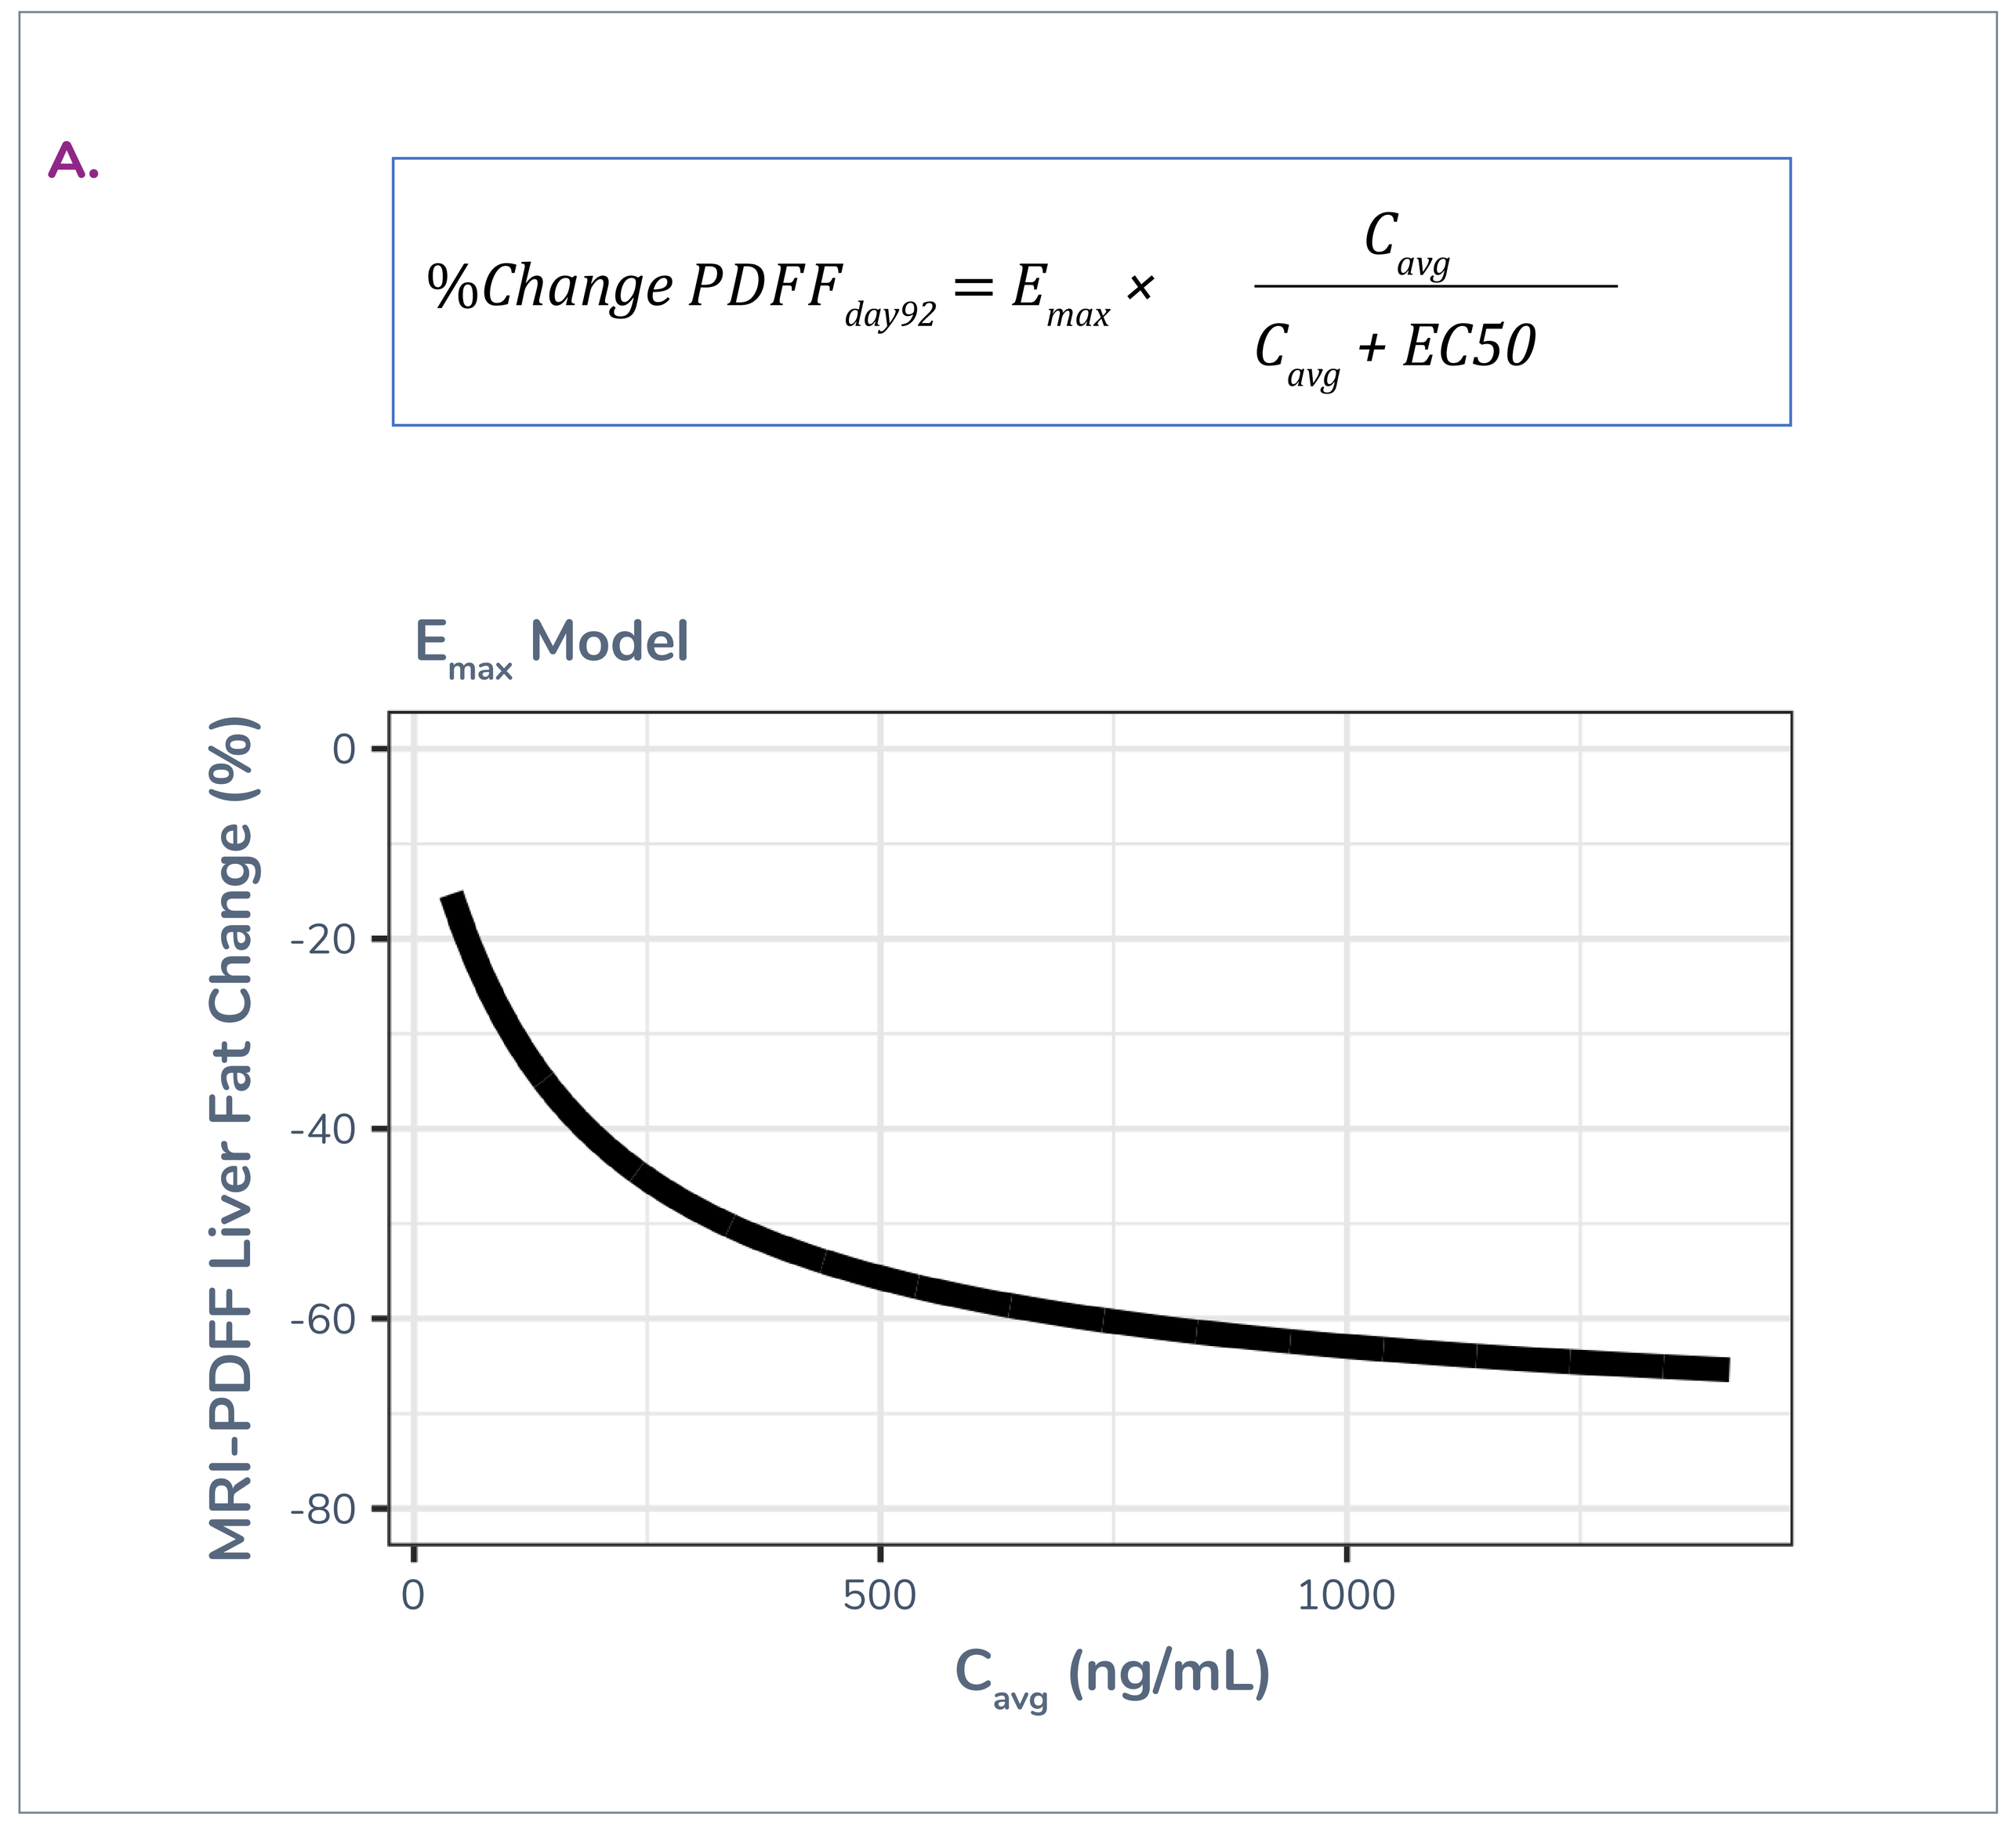
A.
%Change PDFF day92 = Emax × Cavg / (Cavg + EC50)
Emax Model
MRI-PDFF Liver Fat Change (%)
0
-20
-40
-60
-80
0
500
1000
Cavg (ng/mL)

- C_{avg} >~500 ng/mL reached relatively flat range of response based on a E_{max} model.

FIGURE 3: POPULATION PK MODEL WITH COVARIATES

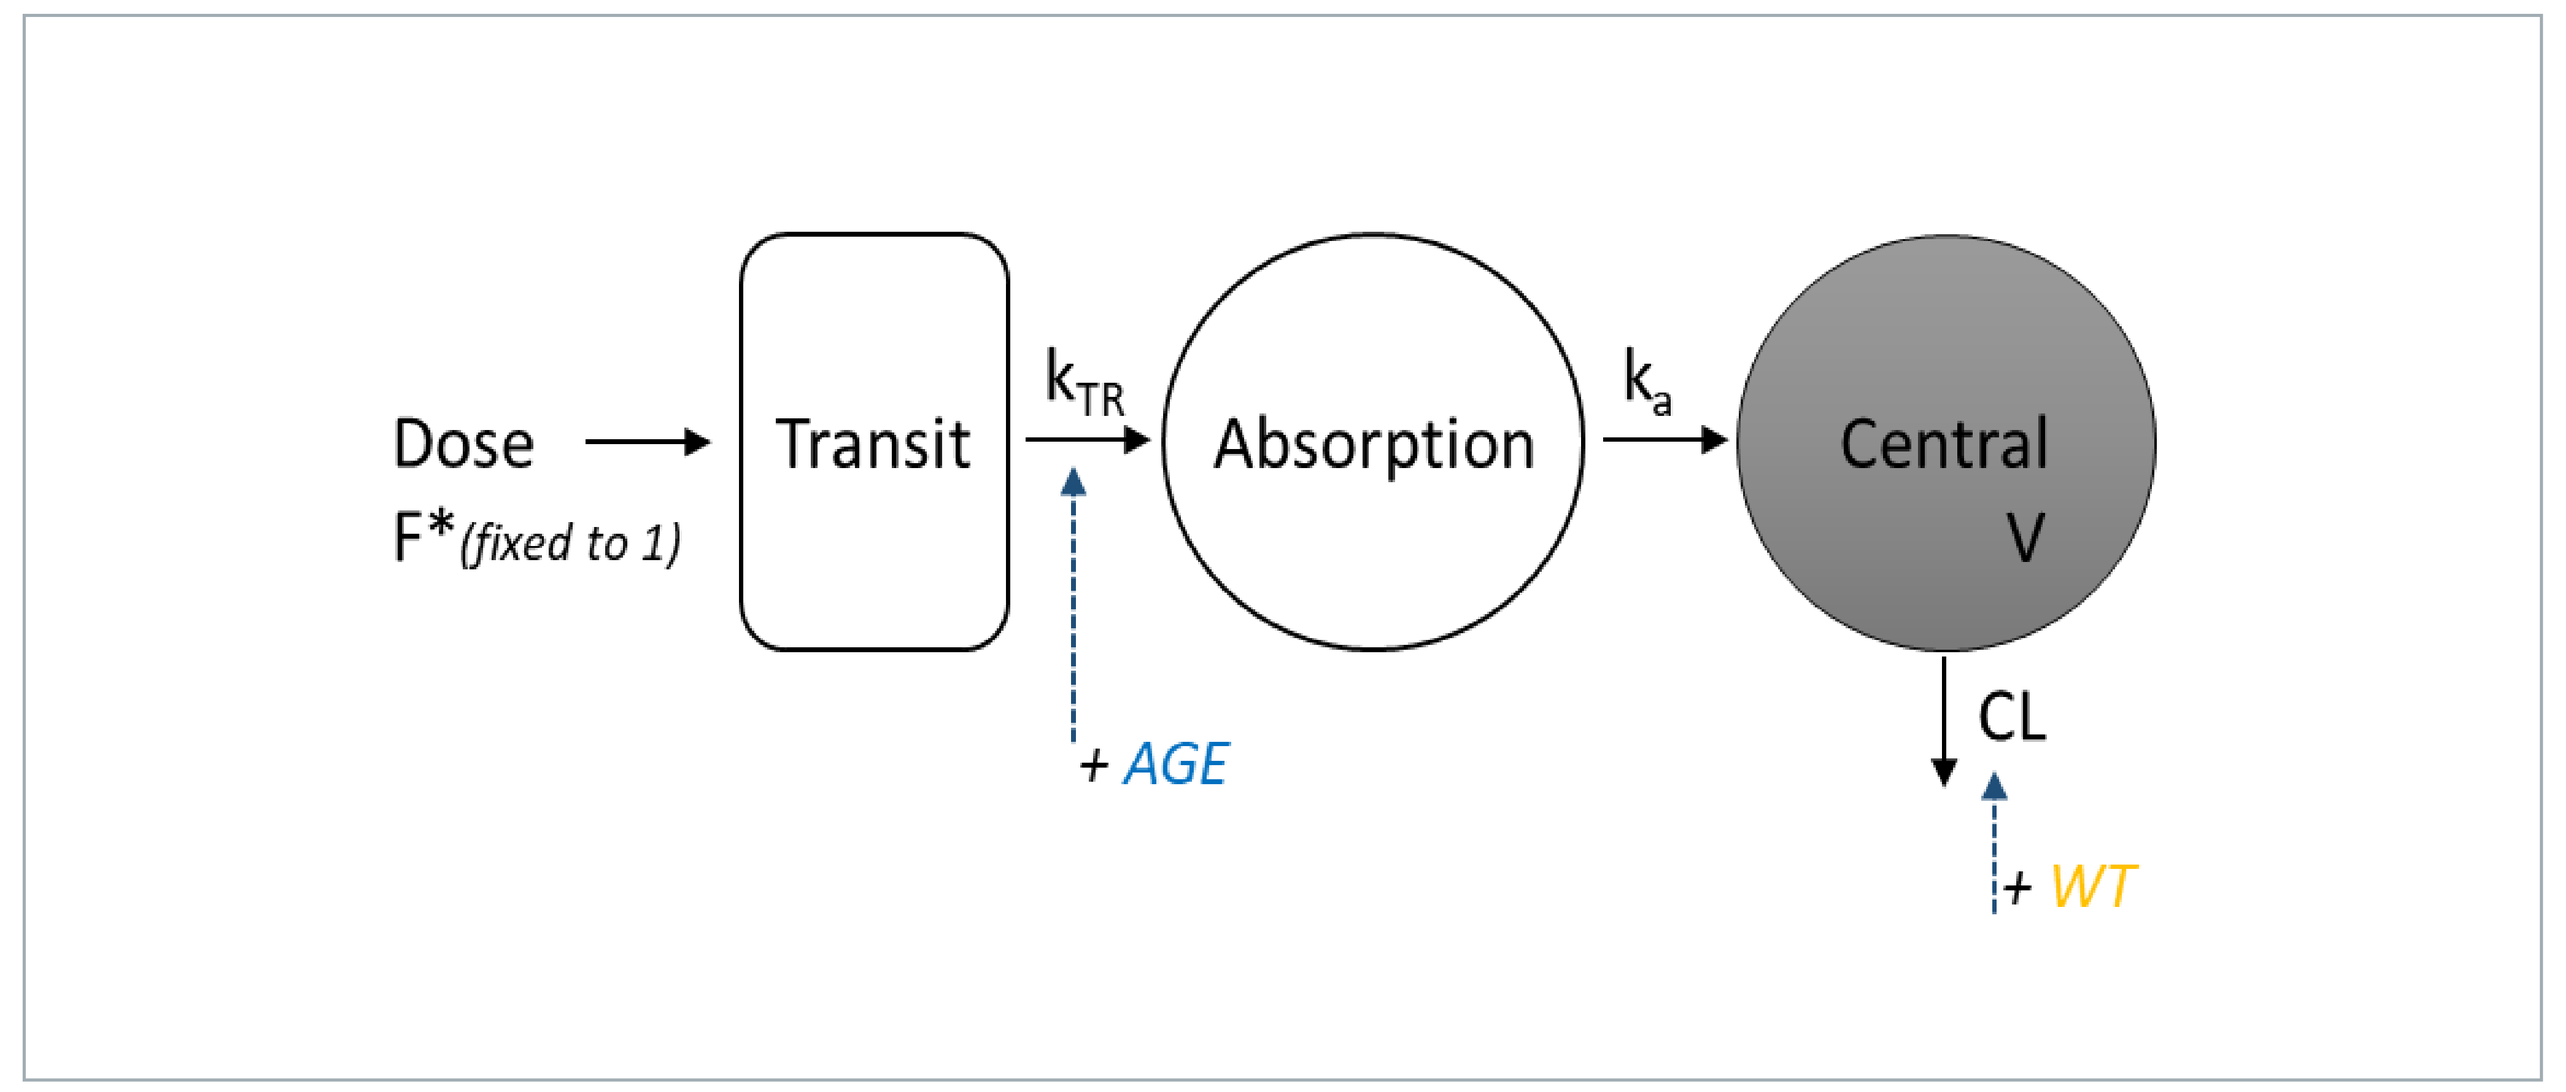
Dose
F*(fixed to 1)
Transit
kTR
Absorption
ka
Central
V
CL
+ AGE
+ WT

BSV, between-subject variability; CL, clearance; CV, coefficient of variation; F, fraction absorbed; K_a, absorption rate constant; K_{TR}, transit rate constant; MTT, mean transit time; RSE, relative standard error; WT, weight; V, volume.

- A one-compartment model with one additional transit absorption compartment was found to adequately describe the PK data, reflecting a delayed absorption profile from the route of SC administration and assumed PK behavior of BIO89-100.
- Covariates (eg, age and body weight) were incorporated into the final model. A 10% increase in drug clearance (CL) was estimated for every 10 kg increase in body weight, while a 13% increase in the absorption mean transit time (MTT) of the drug was estimated for every 10-year increase in age.

FIGURE 4: LONGITUDINAL PK/PD MODEL WITH COVARIATES

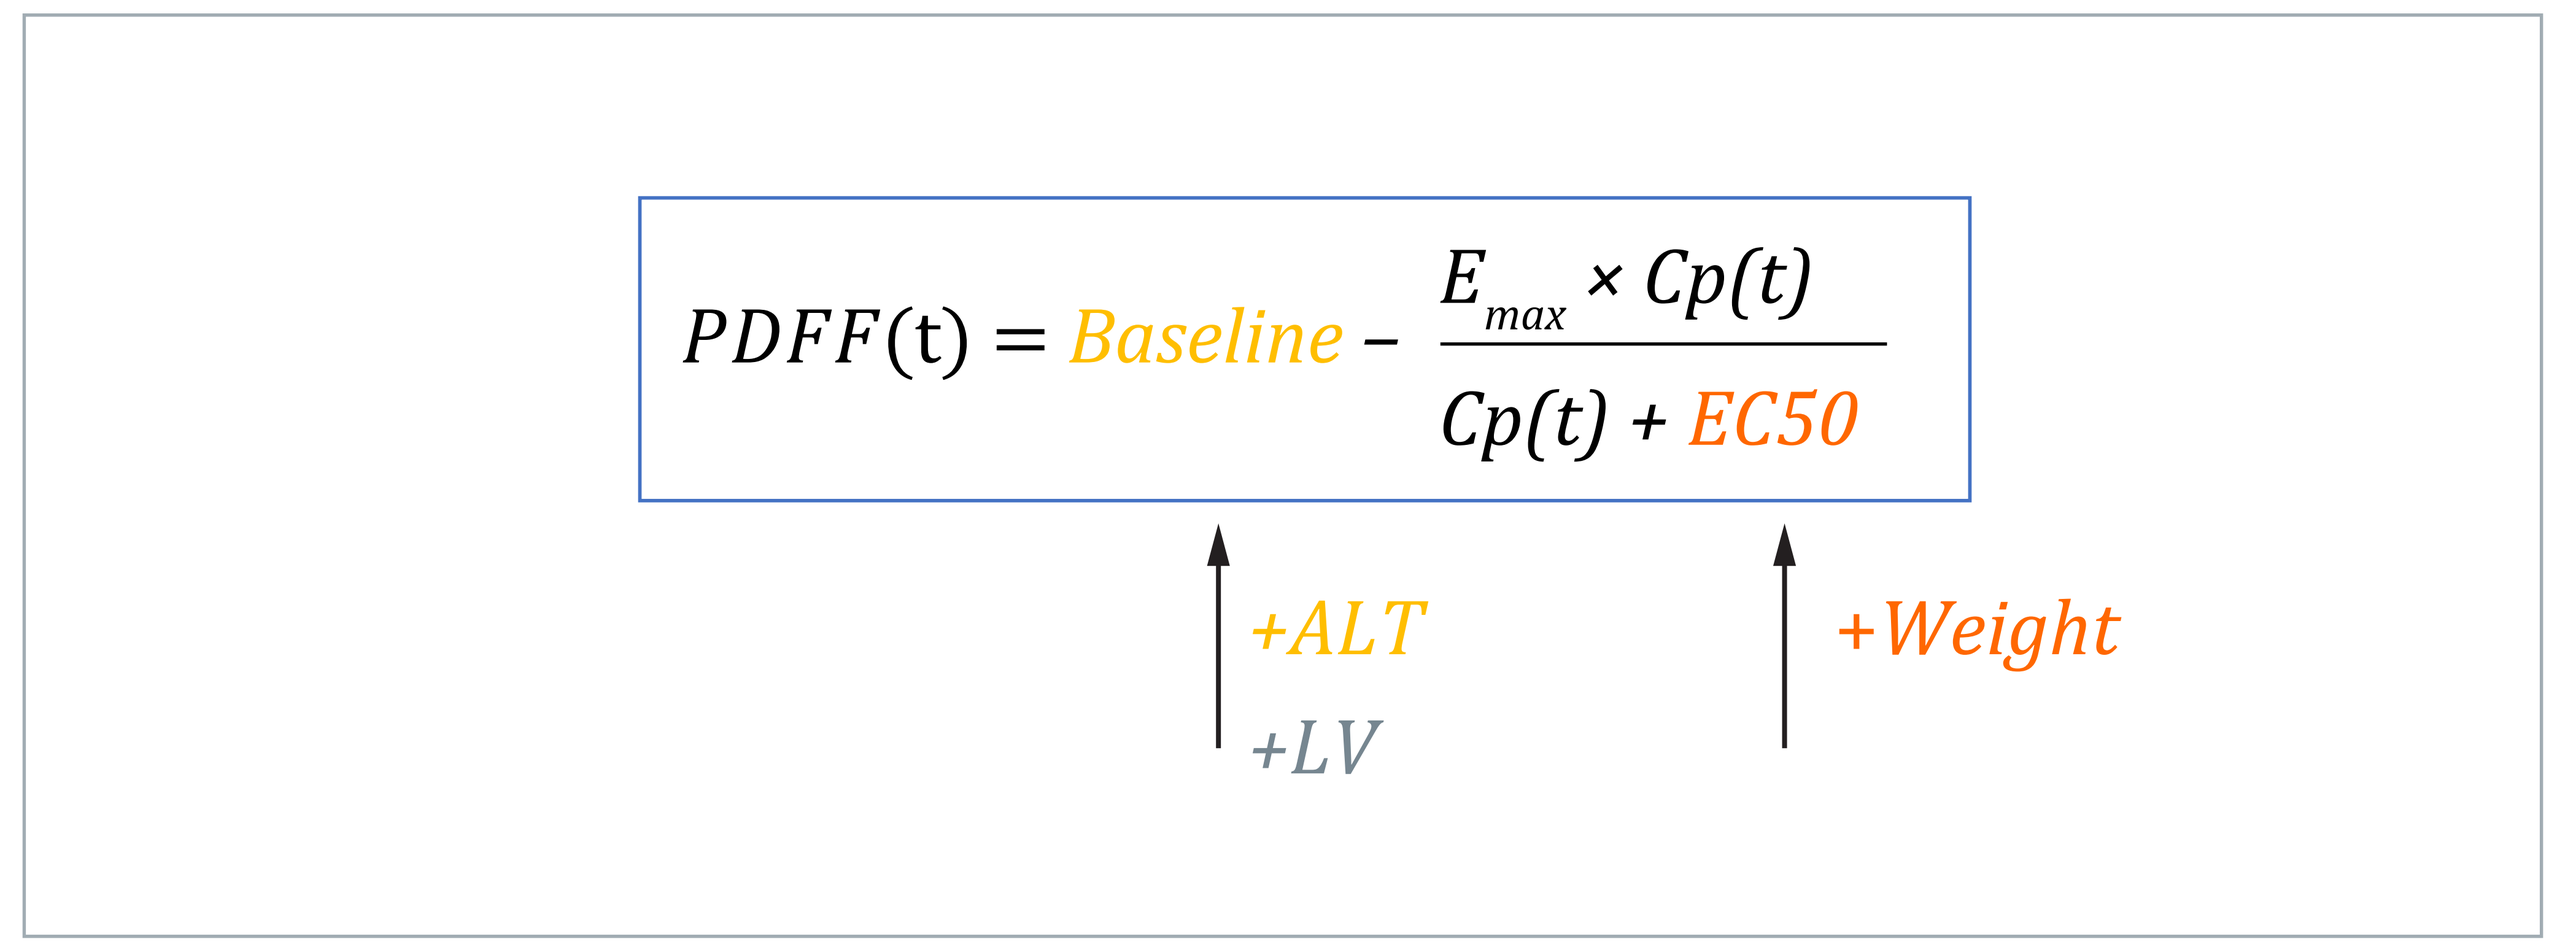
PDFF(t) = Baseline − Emax × Cp(t) / (Cp(t) + EC50)
+ALT
+LV
+Weight

[a]1.3% increase in baseline PDFF for each 10 U/L increase in baseline ALT. [b]10.9% increase in baseline PDFF for each unit increase in baseline LV (L). [c]19.7% increase in EC50 for each 10 kg increase in weight. ALT, alanine aminotransferase; LV, liver volume; PDFF, proton density fat fraction.

- Significant covariates of the model included weight, ALT, and liver volume. Specifically, higher baseline ALT and liver volume were significantly correlated with higher baseline PDFF, while higher baseline weight correlated with higher EC50 values.

FIGURE 5: MODEL SIMULATION FOR THE DOSES IN A FOLLOW-UP PHASE 2B STUDY (ENLIVEN)

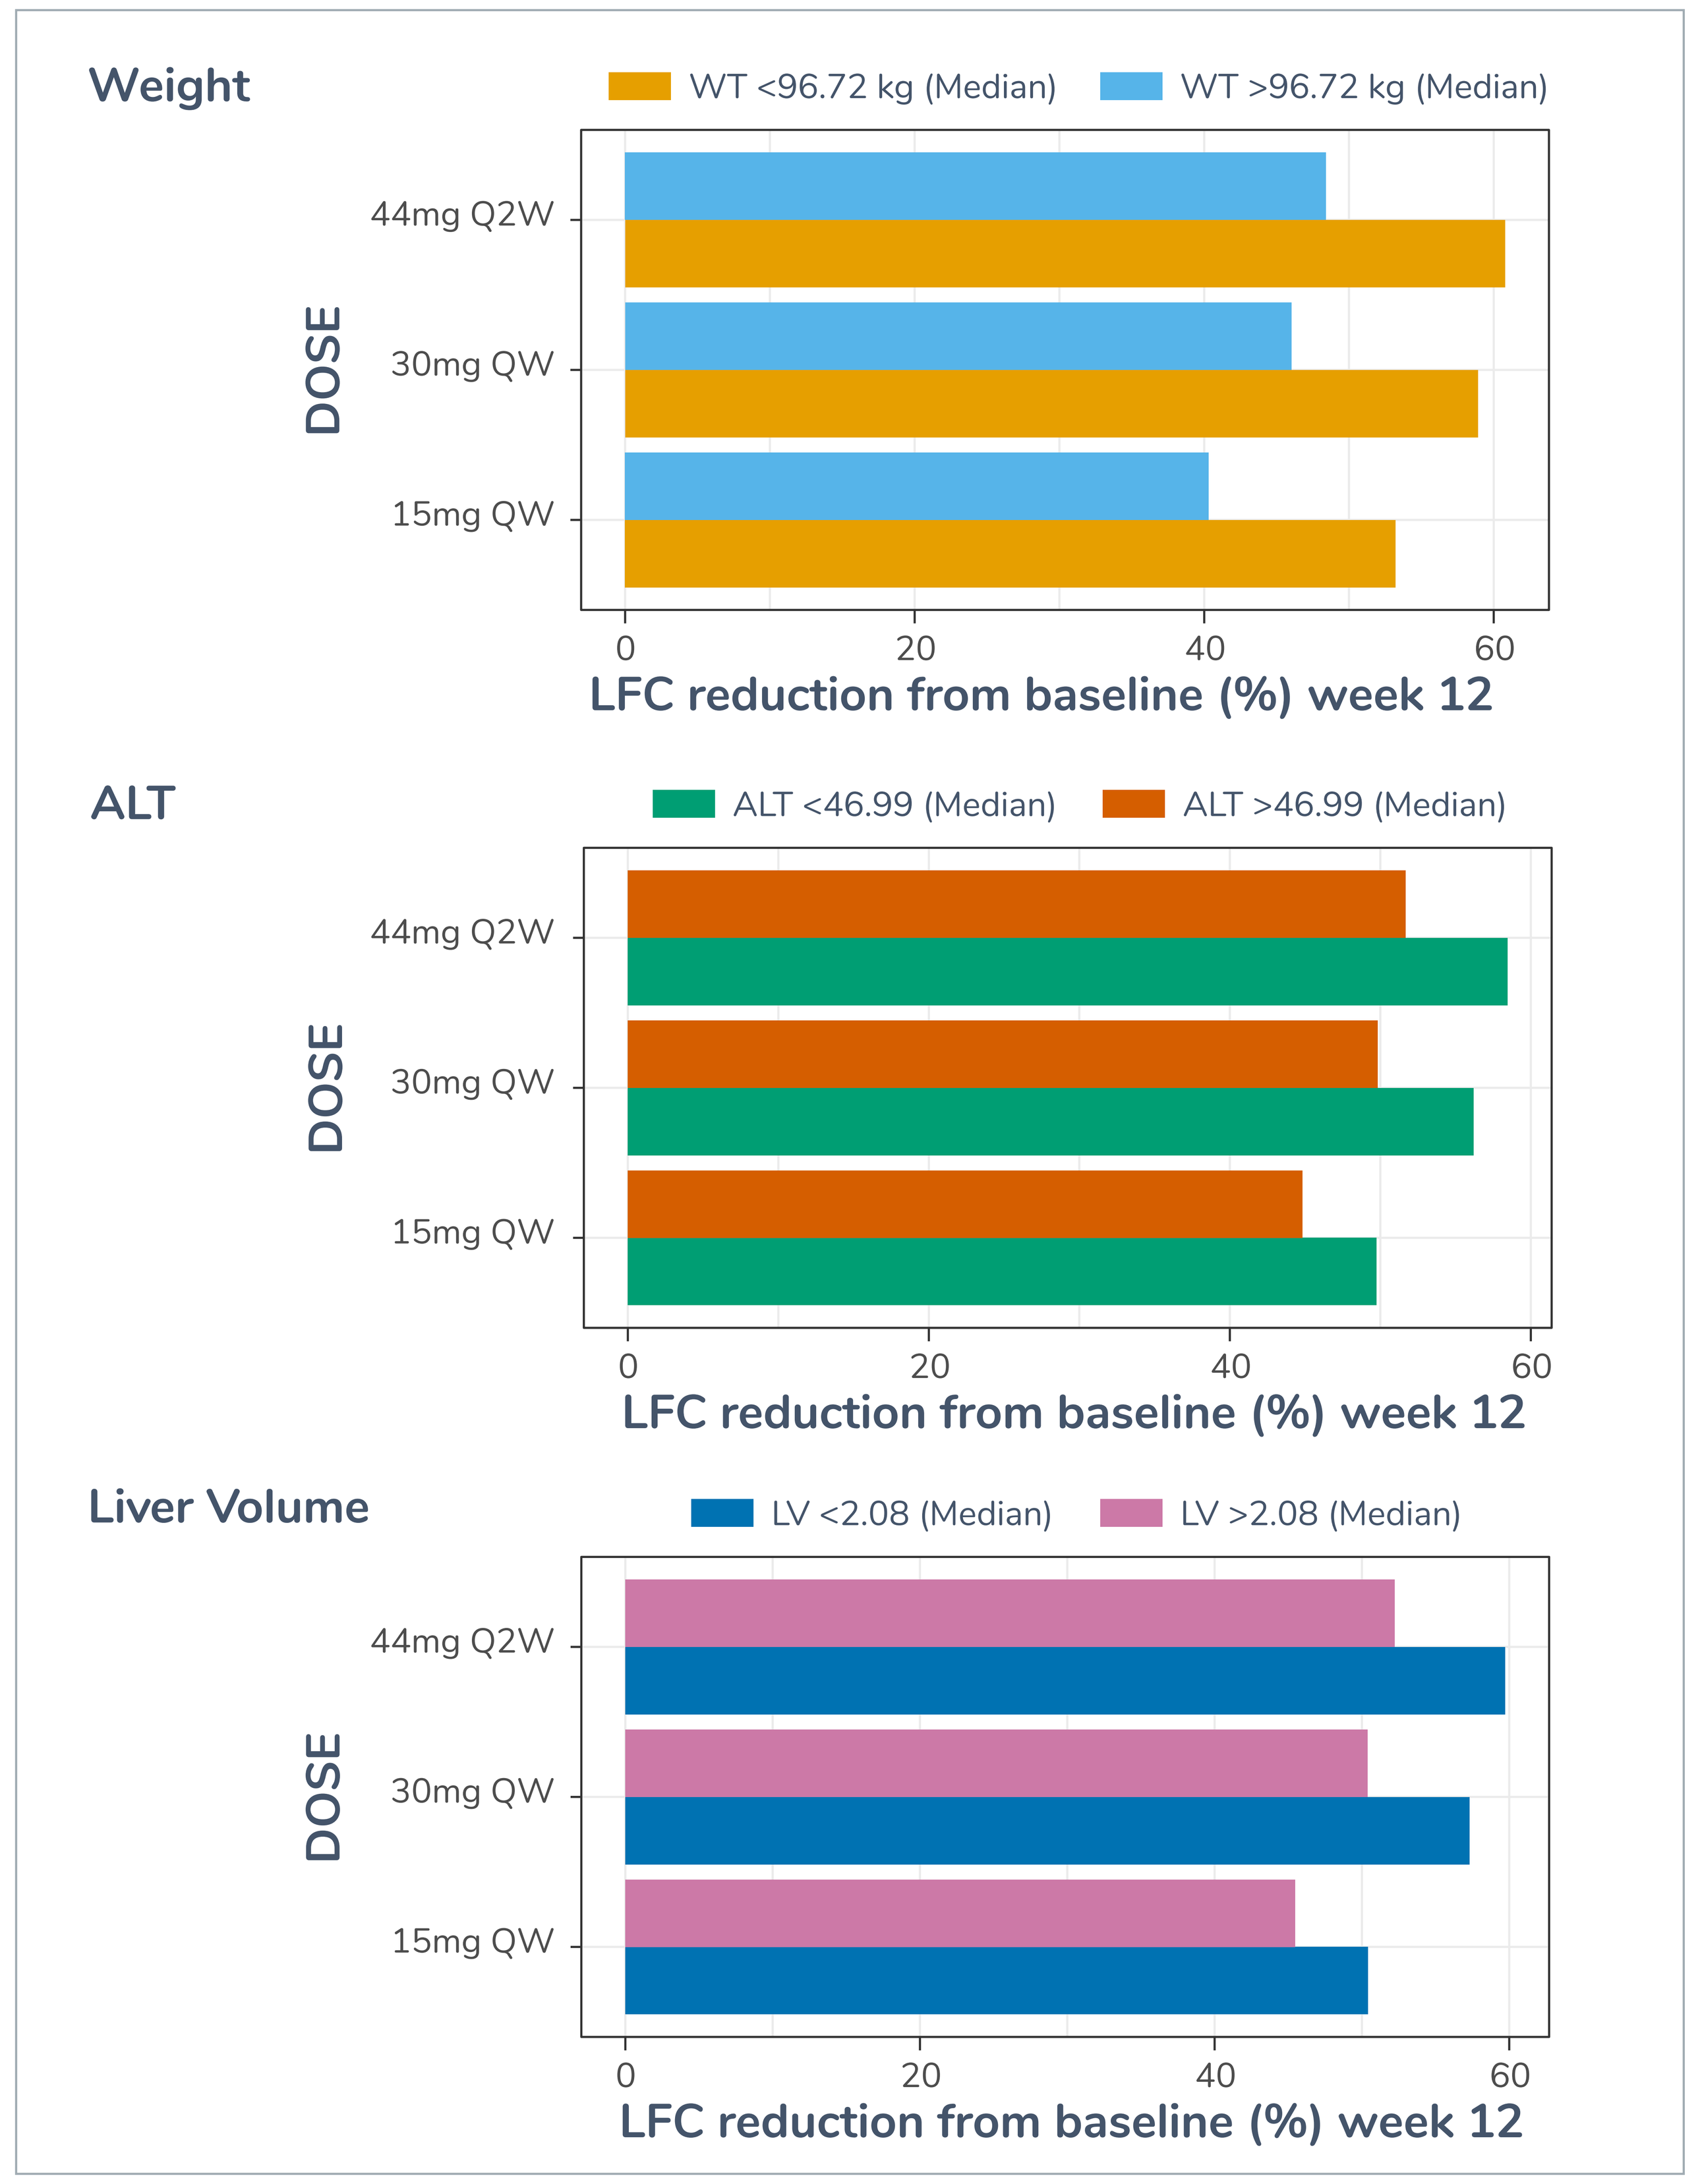
Weight
WT <96.72 kg (Median)
WT >96.72 kg (Median)
DOSE
44mg Q2W
30mg QW
15mg QW
0
20
40
60
LFC reduction from baseline (%) week 12
ALT
ALT <46.99 (Median)
ALT >46.99 (Median)
Liver Volume
LV <2.08 (Median)
LV >2.08 (Median)

ALT, alanine aminotransferase; LV, liver volume; QW, once a week; Q2W, twice a week; WT, weight.

- The 30 mg QW dose approximated E_{max} effect, while the 15 mg QW dose approximated suboptimal effect. Furthermore, 44 mg Q2W dosing (roughly equivalent to 22 mg QW) appeared to be an alternative and effective dose regimen.
- Regardless of covariate effects, patients can still benefit from BIO89-100.

CONCLUSIONS

- Population PK/PD modeling using clinical data from the Phase 1b/2a trial of BIO89-100 suggests that both QW and Q2W are feasible dosing regimens for patients with NASH and helps support dose selection for Phase 2b trial (ENLIVEN).
- Identified potential PK/PD covariates could help improve precision of treatment outcomes; however, these covariates require confirmation from larger study populations.

© 2021 89bio, Inc.