Weekly Subcutaneous Administration of BIO89-100, a Novel GlycoPEGylated-Fibroblast Growth Factor21 (FGF21) Analogue, Inhibits Sweetness Preference in Obese Cynomolgus Monkeys

M. ROSENSTOCK[1], Y. LIU[2], H. MANSBACH[1] and M. MARGALIT[1]

[1]89bio LTD, Departments of Preclinical and Clinical Development, Herzliya, Israel [2]Crown Bioscience Inc., Taicang City, Suzhou City, China

INTRODUCTION

BIO89-100, a novel glycoPEGylated analogue of FGF21, is being developed for the treatment of nonalcoholic steatohepatitis (NASH) and cardio-metabolic disorders. FGF21 regulates carbohydrate and lipid metabolism and has previously been shown to regulate sweetness and alcohol preference in mice and sweetness preference in monkeys.[1,2]

OBJECTIVES

The objective of the study was to assess the effect of BIO89-100 on sweetness preference in obese cynomolgus monkeys.

METHODS

In a 2-bottle sweetness preference test, BIO89-100 was administered to obese cynomolgus monkeys by subcutaneous injection once weekly (QW) for 3 weeks at doses of 0 (vehicle, phosphate-buffered saline) or 1 mg/kg, 3 animals per group. A 2-week washout period followed the last dose. The monkeys' preference for sweet (3% sucrose) or regular water was monitored by measuring fluid intake during the study, ie, before and during drug administration, and following drug's cessation. Other parameters examined included clinical observations, body weight, food intake, and clinical chemistry, including serum blood lipids.

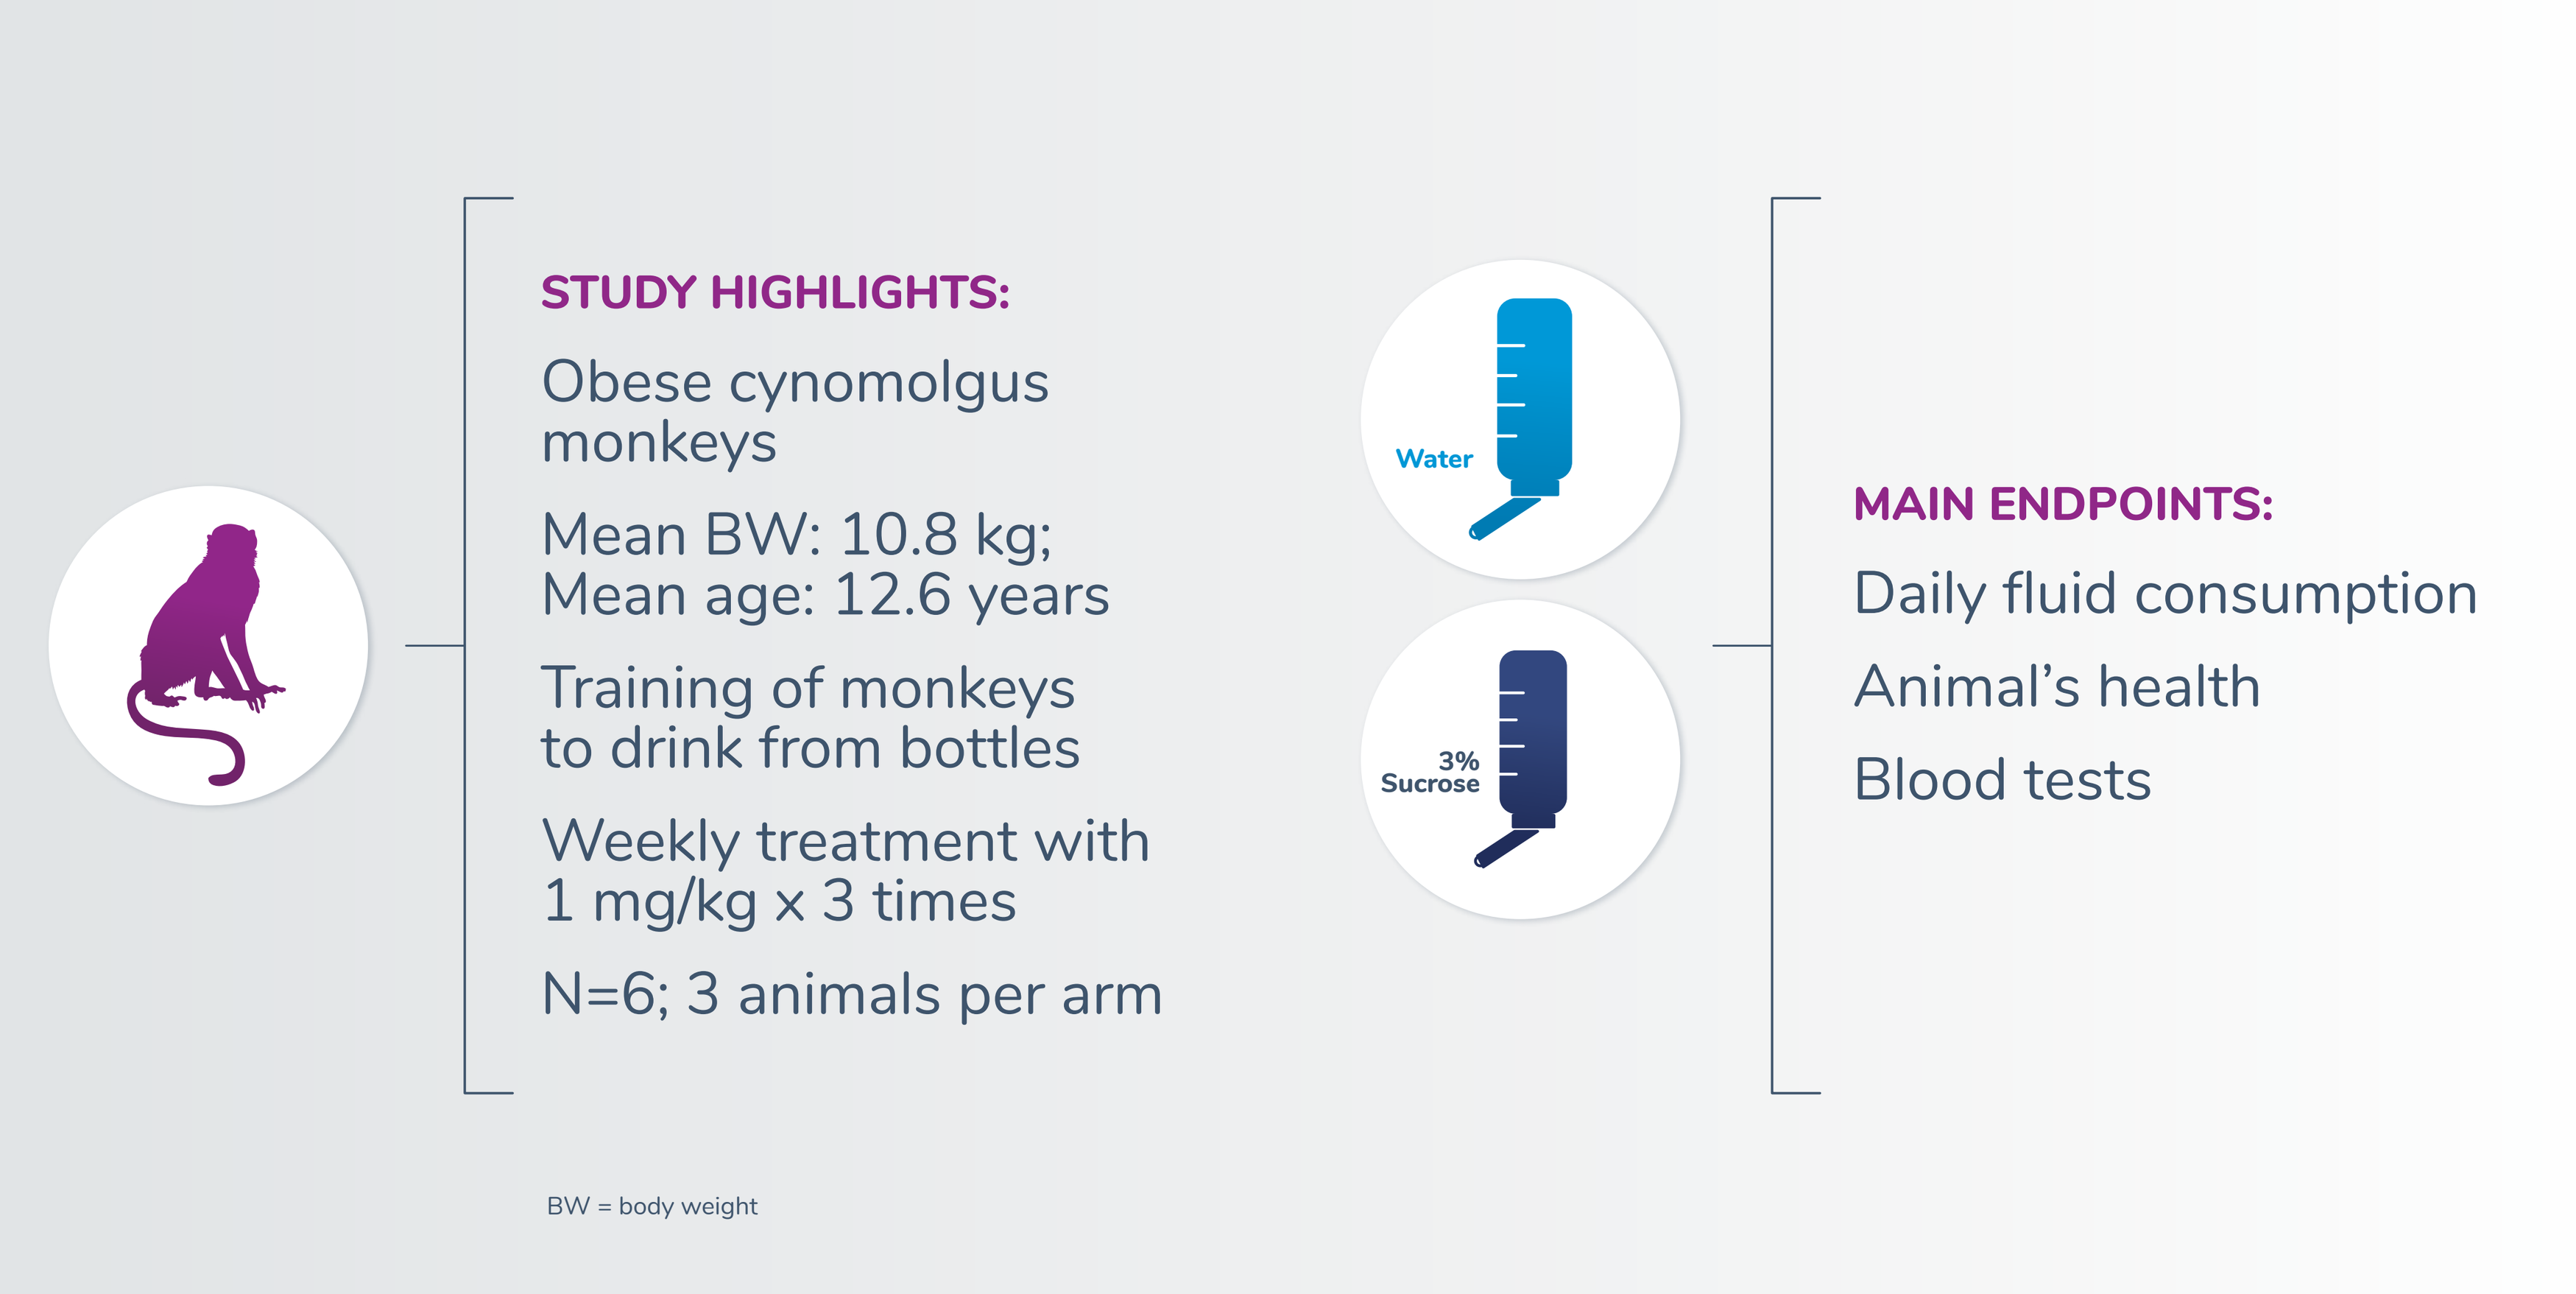
STUDY HIGHLIGHTS:
Obese cynomolgus monkeys
Mean BW: 10.8 kg; Mean age: 12.6 years
Training of monkeys to drink from bottles
Weekly treatment with 1 mg/kg x 3 times
N=6; 3 animals per arm
Water
3% Sucrose
MAIN ENDPOINTS:
Daily fluid consumption
Animal's health
Blood tests
BW = body weight

RESULTS

At baseline, animals who could choose either regular water or water containing 3% sucrose had a significant preference to sucrose-containing water, ie, an increase from mean of 250 mL/day of regular water to a mean of 650 mL/day of sweet water.

Following treatment with BIO89-100, preference for sweet water decreased significantly, to as low as mean of 1 mL/day, while preference for regular water increased, from mean of 37 to 184 mL/day. Once treatment with the compound ceased, the preference for sweet water gradually returned.

Vehicle-treated animals maintained a preference for sweet water throughout the treatment and washout periods.

Other noteworthy findings in obese animals treated with BIO89-100 included decreases in body weight, food intake, triglycerides (TG) and alanine transaminase (ALT), and increase in high-density lipoprotein (HDL) by a mean change from baseline of -13% (p=0.007), -44% (p=0.071), -78% (p=0.012), -41% (p=0.04) and +36% (p= 0.129), respectively, on Day 23.

Low-density lipoprotein (LDL) and glucose were unchanged relative to the vehicle group.

Except for food intake, none of these parameters returned to baseline values following the 2-week washout period.

Figure 1. Effect of BIO89-100 on Sugar Preference in Obese Monkeys in a 2-bottle Sweetness Preference Test

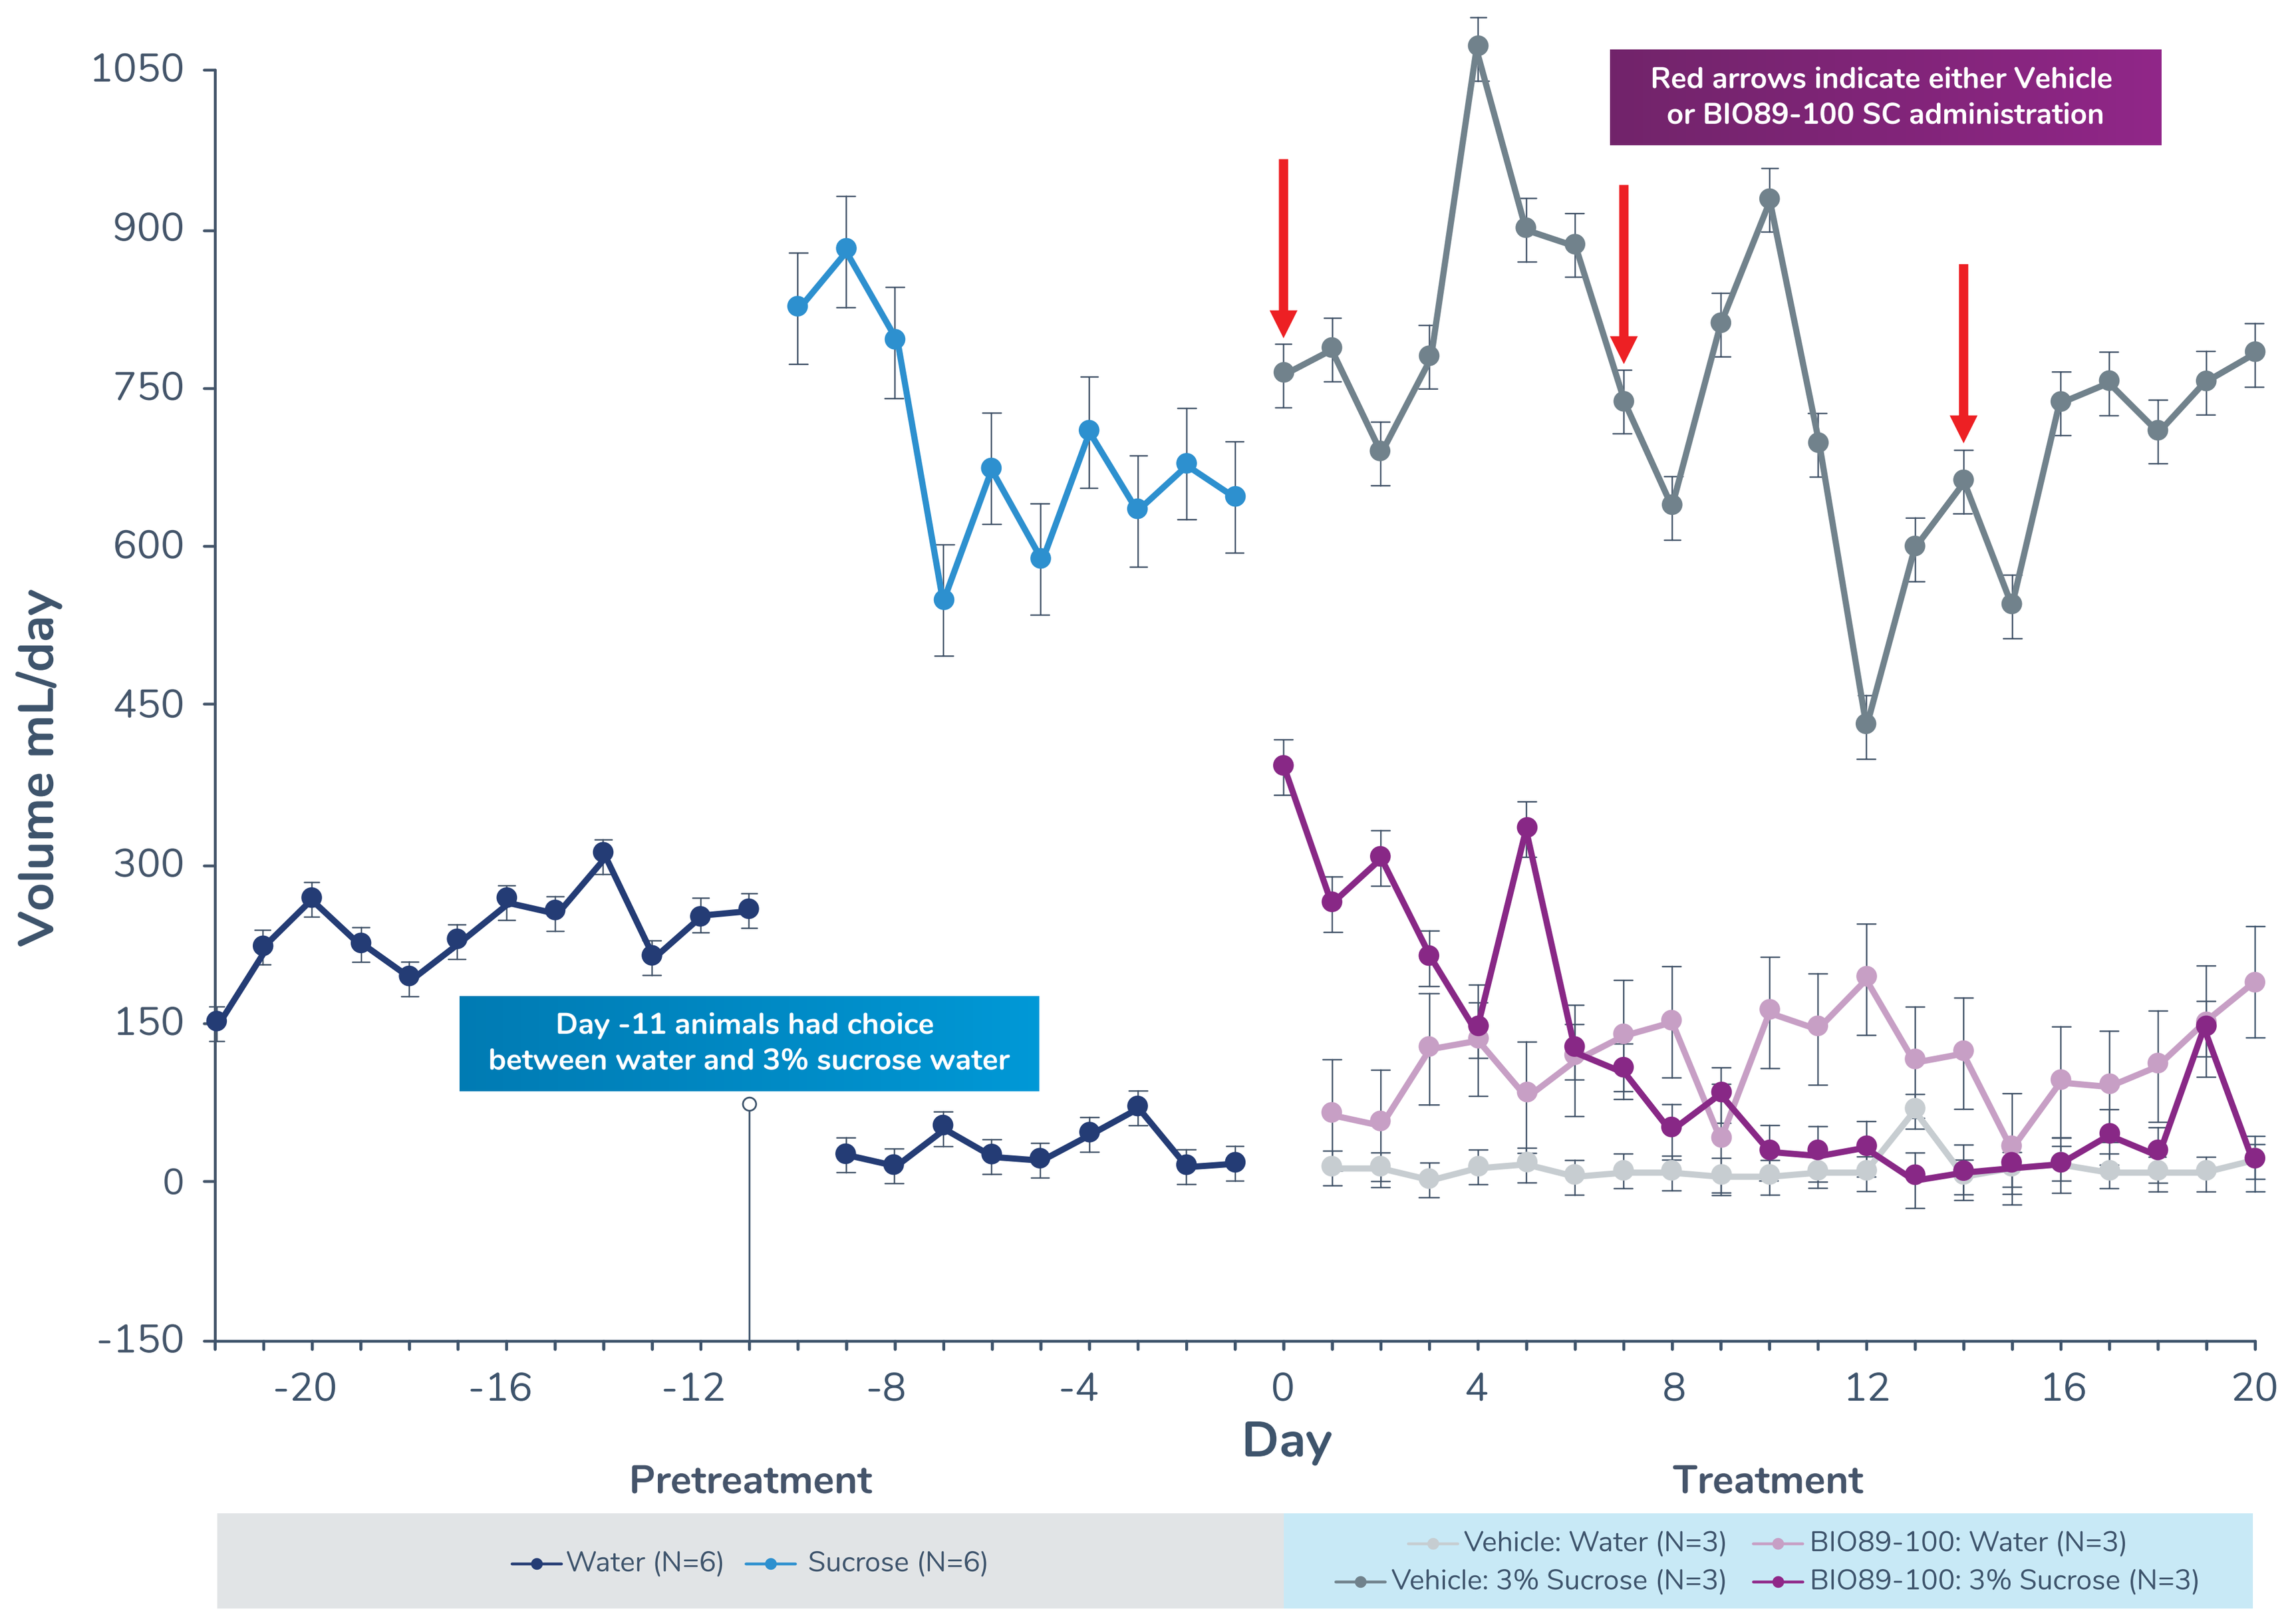
Red arrows indicate either Vehicle or BIO89-100 SC administration
Day -11 animals had choice between water and 3% sucrose water
Volume mL/day
1050
900
750
600
450
300
150
0
-150
-20
-16
-12
-8
-4
0
4
8
12
16
20
Day
Pretreatment
Treatment
Water (N=6)
Sucrose (N=6)
Vehicle: Water (N=3)
Vehicle: 3% Sucrose (N=3)
BIO89-100: Water (N=3)
BIO89-100: 3% Sucrose (N=3)

Figure 2. Effect of BIO89-100 on TG Levels, Body Weight, ALT Levels and HDL Levels in Obese Monkeys (% Change of Baseline)

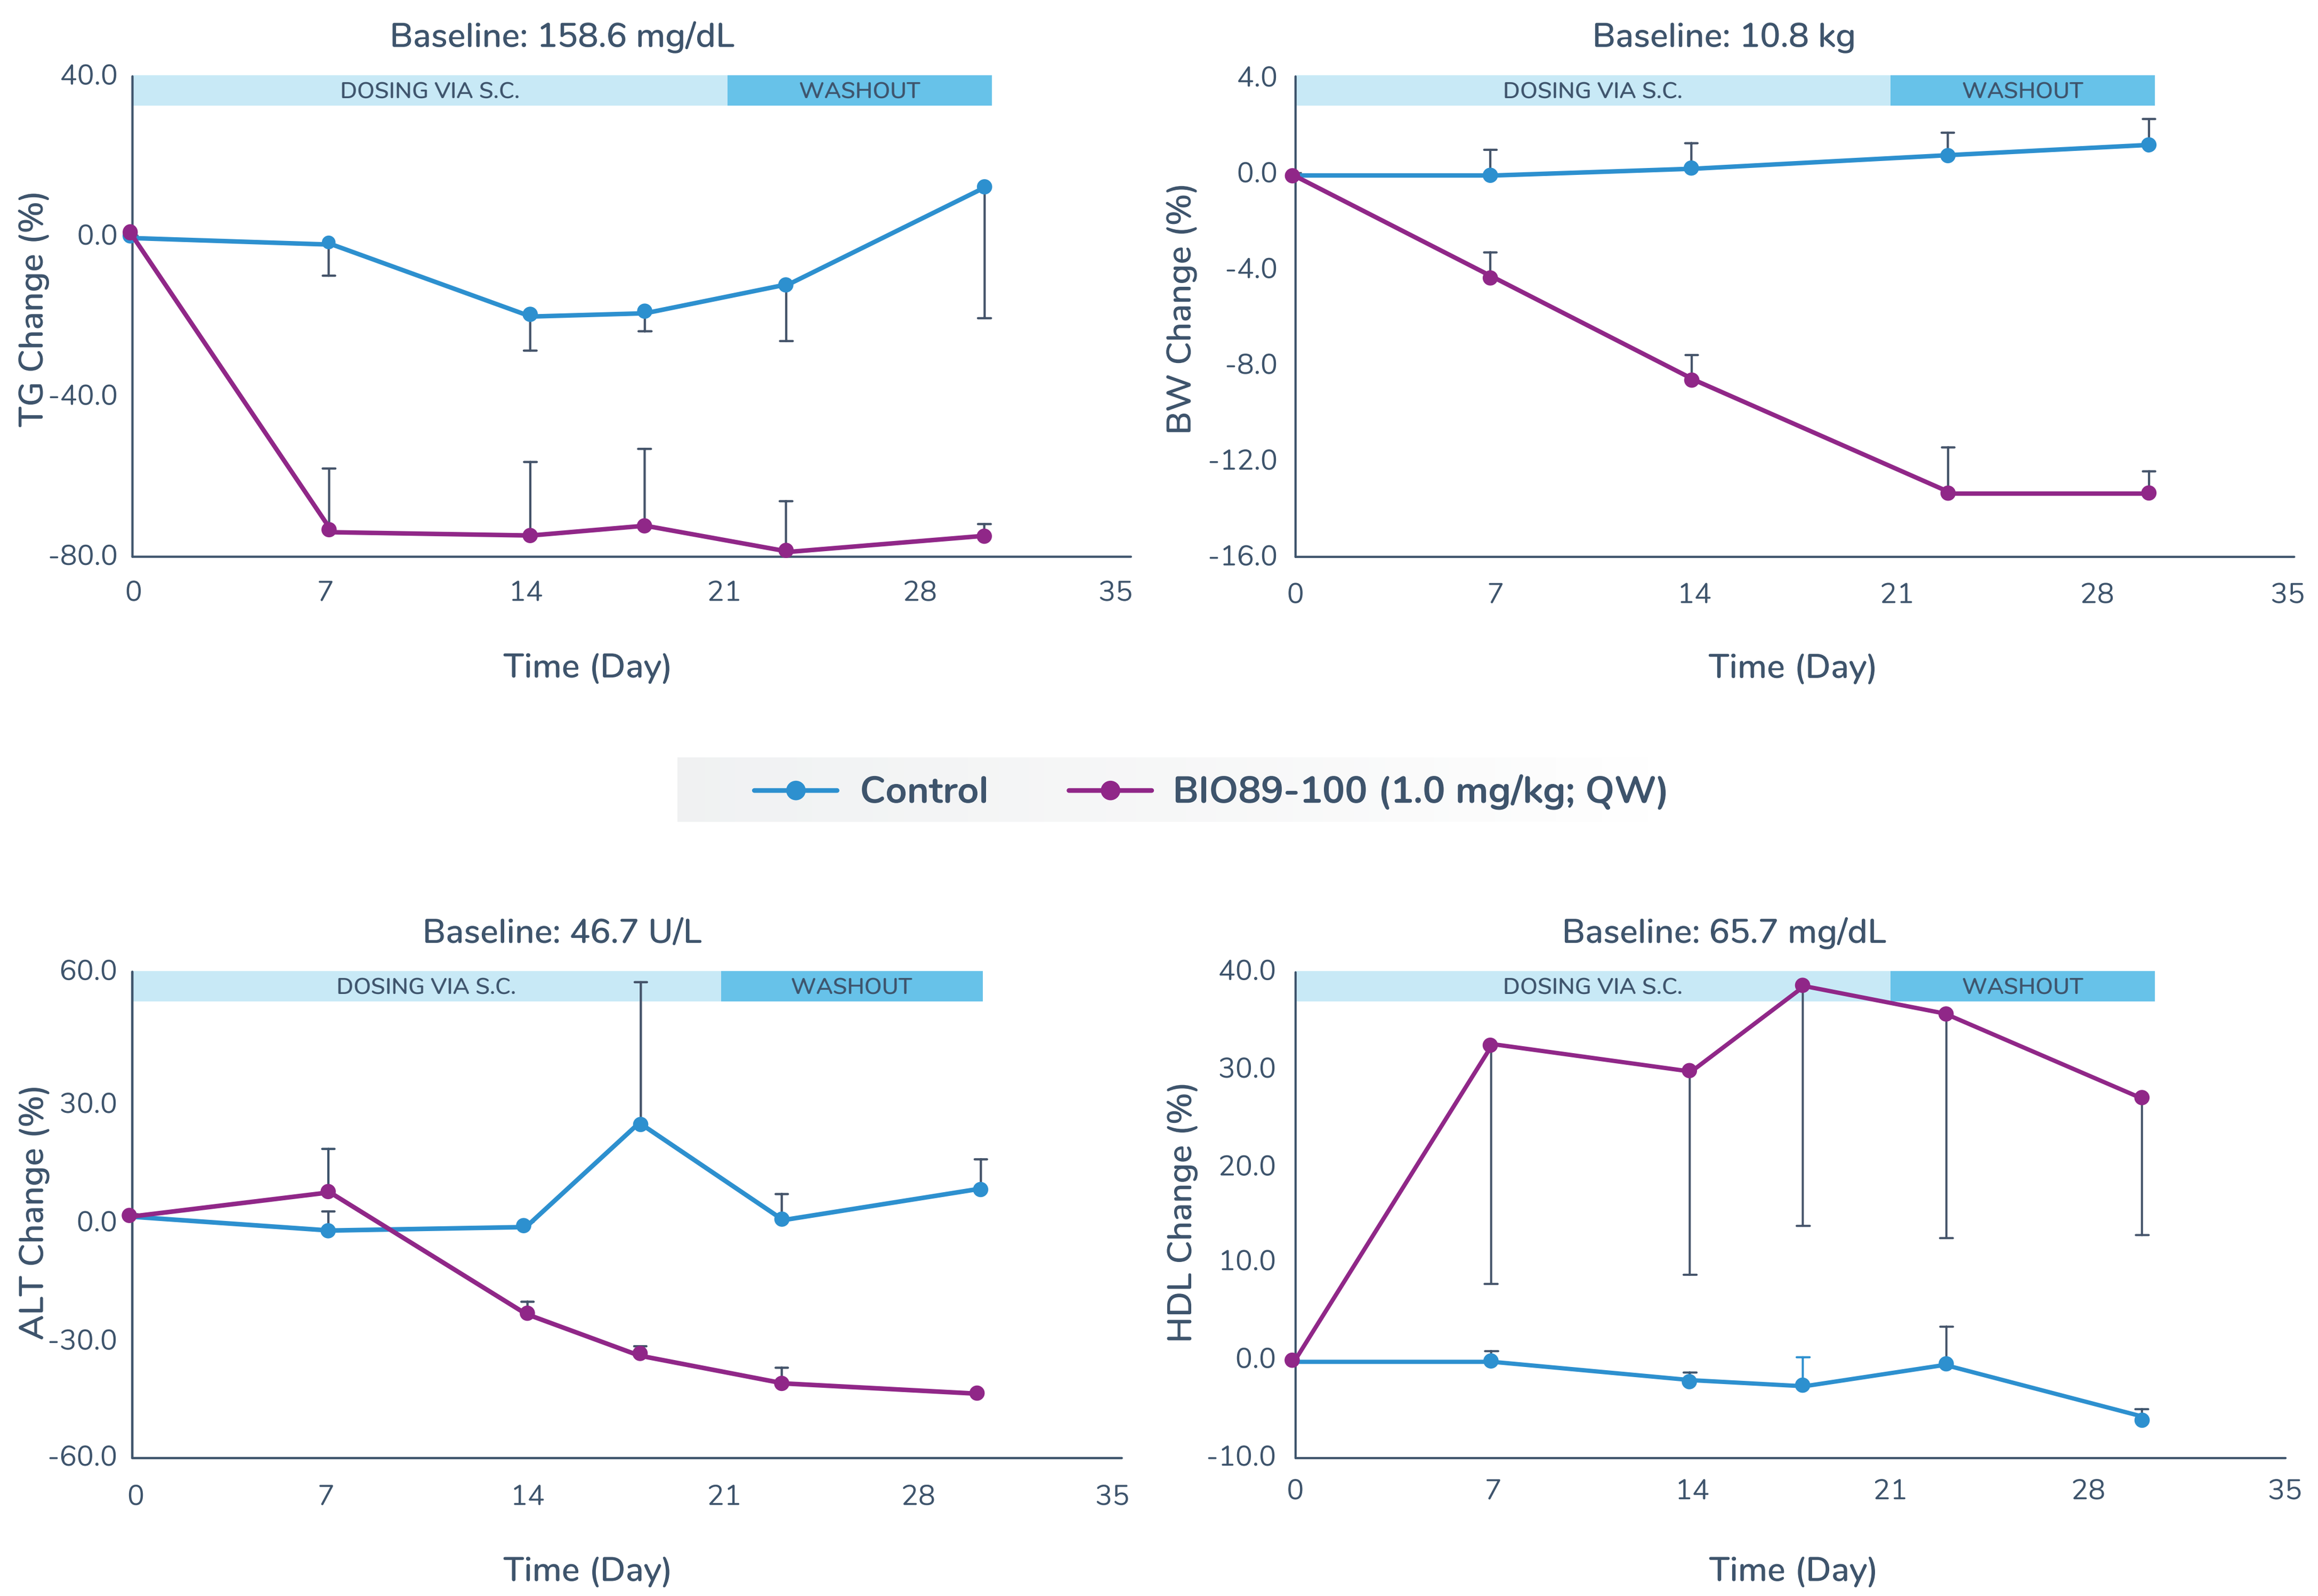
Baseline: 158.6 mg/dL
TG Change (%)
Baseline: 10.8 kg
BW Change (%)
Baseline: 46.7 U/L
ALT Change (%)
Baseline: 65.7 mg/dL
HDL Change (%)
DOSING VIA S.C.
WASHOUT
Time (Day)
Control
BIO89-100 (1.0 mg/kg; QW)

CONCLUSIONS

Weekly administration of BIO89-100 to obese monkeys resulted in a significant decrease in sweetness preference and in improvements in metabolic and liver-related lab parameters. FGF21 analogues such as BIO89-100 are a promising treatment modality for NASH. Decreased preference for sugar, if applicable to humans, may confer an additional important benefit patients with NASH and other cardiometabolic disorders.

Clinical studies are underway to evaluate the role of BIO89-100 in treatment of patients with NAFLD/NASH.

REFERENCES

1. Talukdar S, Owen BM, Song P, et al. FGF21 regulates sweet and alcohol preference. *Cell Metab.* 2016;23(2):344-349.
2. van Holstein-Rathlou S, BonDurant LD, Peltekian L. FGF21 mediates endocrine control of simple sugar intake and sweet taste preference by the liver. *Cell Metab.* 2016;23(2):335-343.

Trademarks, registered or otherwise, are the property of their respective owner(s). © 2019 89bio LTD