Pegozafermin for the Treatment of Severe Hypertriglyceridemia: A Randomized, Double-blind, Placebo-controlled Phase 2 Study (ENTRIGUE STUDY)

DEEPAK L. BHATT, HAROLD E. BAYS, MICHAEL MILLER, TERESA PARLI, SHIBAO FENG, LULU STERLING, CYNTHIA L. HARTSFIELD, GERMAINE D. AGOLLAH, HANK MANSBACH, JOHN J. P. KASTELEIN ON BEHALF OF THE ENTRIGUE INVESTIGATORS

INTRODUCTION

- Fibroblast growth factor 21 (FGF21) is an endogenous hormone regulating lipid and glucose metabolism and energy expenditure.
- Pegozafermin (PGZ) is a glycoPEGylated FGF21 analog designed to have a longer half-life than native FGF21.
- PGZ is currently being developed for treatment of severe hypertriglyceridemia (SHTG) and non-alcoholic steatohepatitis (NASH).
- Data from a Phase 1b/2a POC study in subjects with NASH demonstrated overall metabolic benefit with improvements in lipids (TG, LDL, non-HDL and HDL), insulin resistance, HbA1c, body weight, and liver fat.

BACKGROUND

- SHTG ≥500mg/dL increases the risk of acute pancreatitis and cardiovascular disease.
- Current approved therapies reduce TG levels, however many SHTG patients do not attain desired levels (TG levels <500 mg/dl) leaving residual risk for acute pancreatitis and highlighting the need for new therapeutic options.
- SHTG is commonly associated with obesity, metabolic syndrome, insulin resistance, type 2 diabetes mellitus (T2DM) and non-alcoholic fatty liver disease (NAFLD).
- An ideal therapy would not only lower TG levels, but provide benefit for other metabolic comorbidities.

OBJECTIVE

- The ENTRIGUE trial was designed to investigate PGZ as a novel therapeutic agent for the treatment of SHTG

METHODS

Randomized, Double-Blind, Phase 2 Trial of Patients with SHTG (ENTRIGUE)

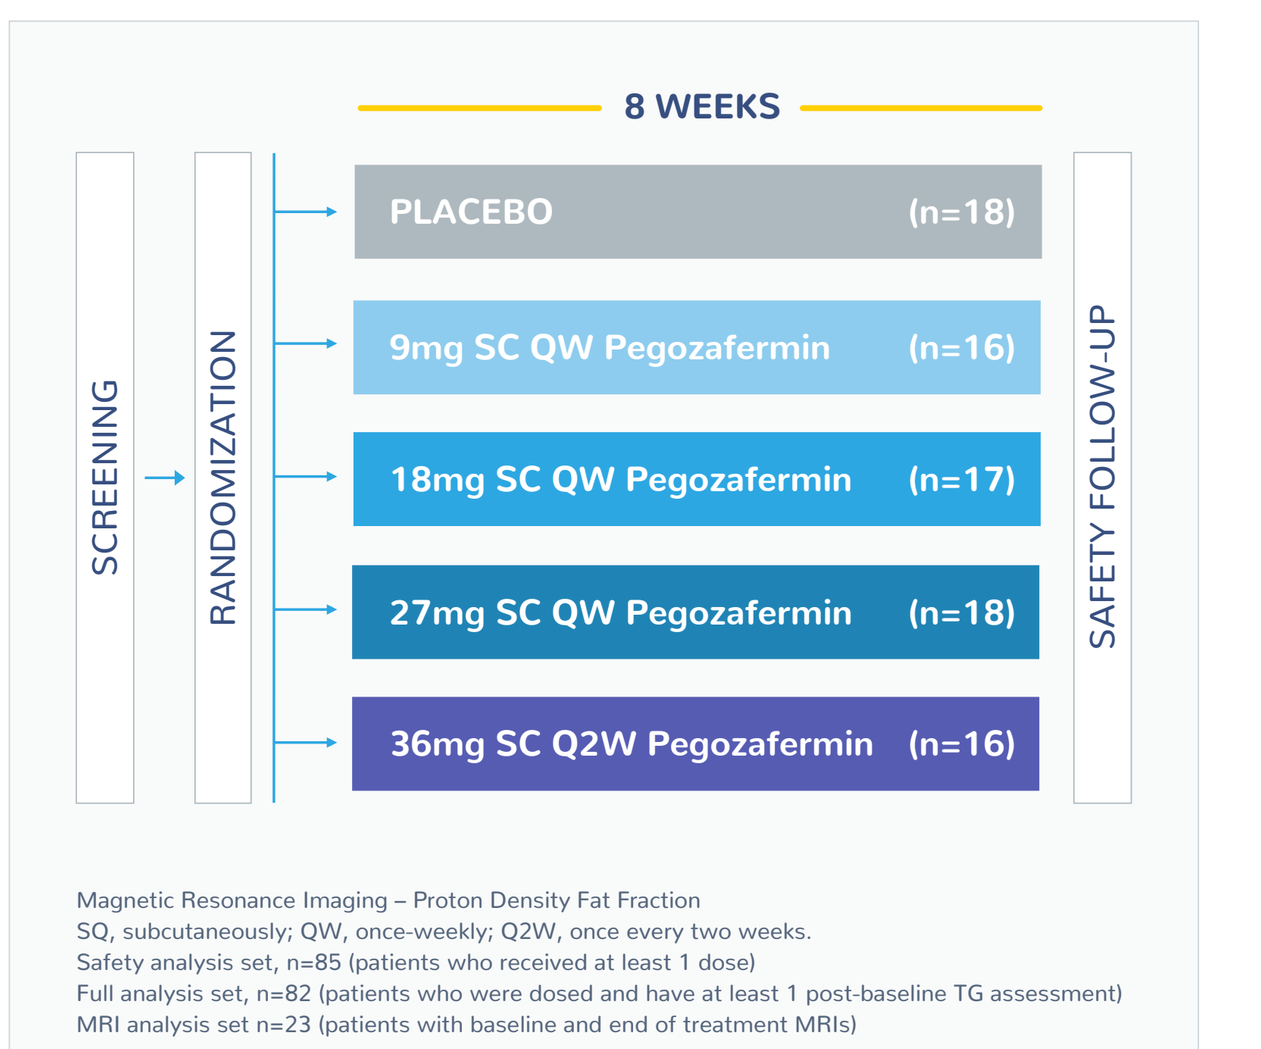
8 WEEKS
SCREENING
RANDOMIZATION
PLACEBO (n=18)
9mg SC QW Pegozafermin (n=16)
18mg SC QW Pegozafermin (n=17)
27mg SC QW Pegozafermin (n=18)
36mg SC Q2W Pegozafermin (n=16)
SAFETY FOLLOW-UP

Magnetic Resonance Imaging – Proton Density Fat Fraction
SQ, subcutaneously; QW, once-weekly; Q2W, once every two weeks.
Safety analysis set, n=85 (patients who received at least 1 dose)
Full analysis set, n=82 (patients who were dosed and have at least 1 post-baseline TG assessment)
MRI analysis set n=23 (patients with baseline and end of treatment MRIs)

KEY INCLUSION CRITERIA

- TG ≥500 mg/dL and ≤2,000 mg/dL
- Background therapy: statins and/or prescription omega-3 fatty acids, and/or fibrates OR none

PRIMARY ENDPOINT

- Primary endpoint: % Change in TGs from baseline

KEY SECONDARY ENDPOINTS

- Lipids: non-HDL-C, HDL-C, Apo-B
- Liver fat (MRI-PDFF)
- Glycemic control

RESULTS

Baseline Characteristics

Represents a population at high risk for cardiovascular disease

Parameter Mean or %	Placebo (n=18)	PGZ Pooled (n=67)	PGZ 9mg QW (n=16)	PGZ 18mg QW (n=17)	PGZ 27mg QW (n=18)	PGZ 36mg Q2W (n=16)	Total (n=85)
Age (years)	57.5	52.7	54.6	49.2	53.9	53.1	53.7
Male (%)	66.7	77.6	68.8	82.4	72.2	87.5	75.3
BMI (kg/m²)	33.1	33.1	32.9	32.3	34.2	32.9	33.1
Type 2 Diabetes (%)	61.1	47.8	56.3	35.3	55.6	43.8	50.6
Triglyceride (mg/dL)	720.3	735.8	721.7	709.5	680.3	840.3	732.5
non-HDL cholesterol (mg/dL)	219.6	209.3	216.2	203.2	203.4	215.4	211.5
HDL cholesterol (mg/dL)	28.3	28.4	30.7	27.3	30.6	24.8	28.4
LDL cholesterol (mg/dL)	87.9	89.4	91.6	88.3	97.3	79.5	89.1
Apolipoprotein B (ApoB) (mg/dL)	116.3	115.3	120.1	115.3	119.3	105.9	115.5
HbA1c ≥6.5%, n (%)	7 (38.9)	30 (44.8)	9 (56.3)	6 (35.3)	9 (50.0)	6 (37.5)	37 (43.5)
ALT (U/L)	29.1	33.9	36.3	36.9	33.0	29.2	32.8
Any background lipid-modifying therapy (LMT), n (%)	11 (61.1)	36 (53.7)	8 (50.0)	9 (52.9)	11 (61.1)	8 (50.0)	47 (55.3)
Liver fat content (%) (n=24)	16.5 [n=6]	21.3 [n=18]	19.8 [n=3]	18.0 [n=5]	22.4 [n=7]	25.5 [n=3]	20.1 [n=24]

PGZ Significantly Reduces Triglycerides Across All Dose Groups

Primary endpoint

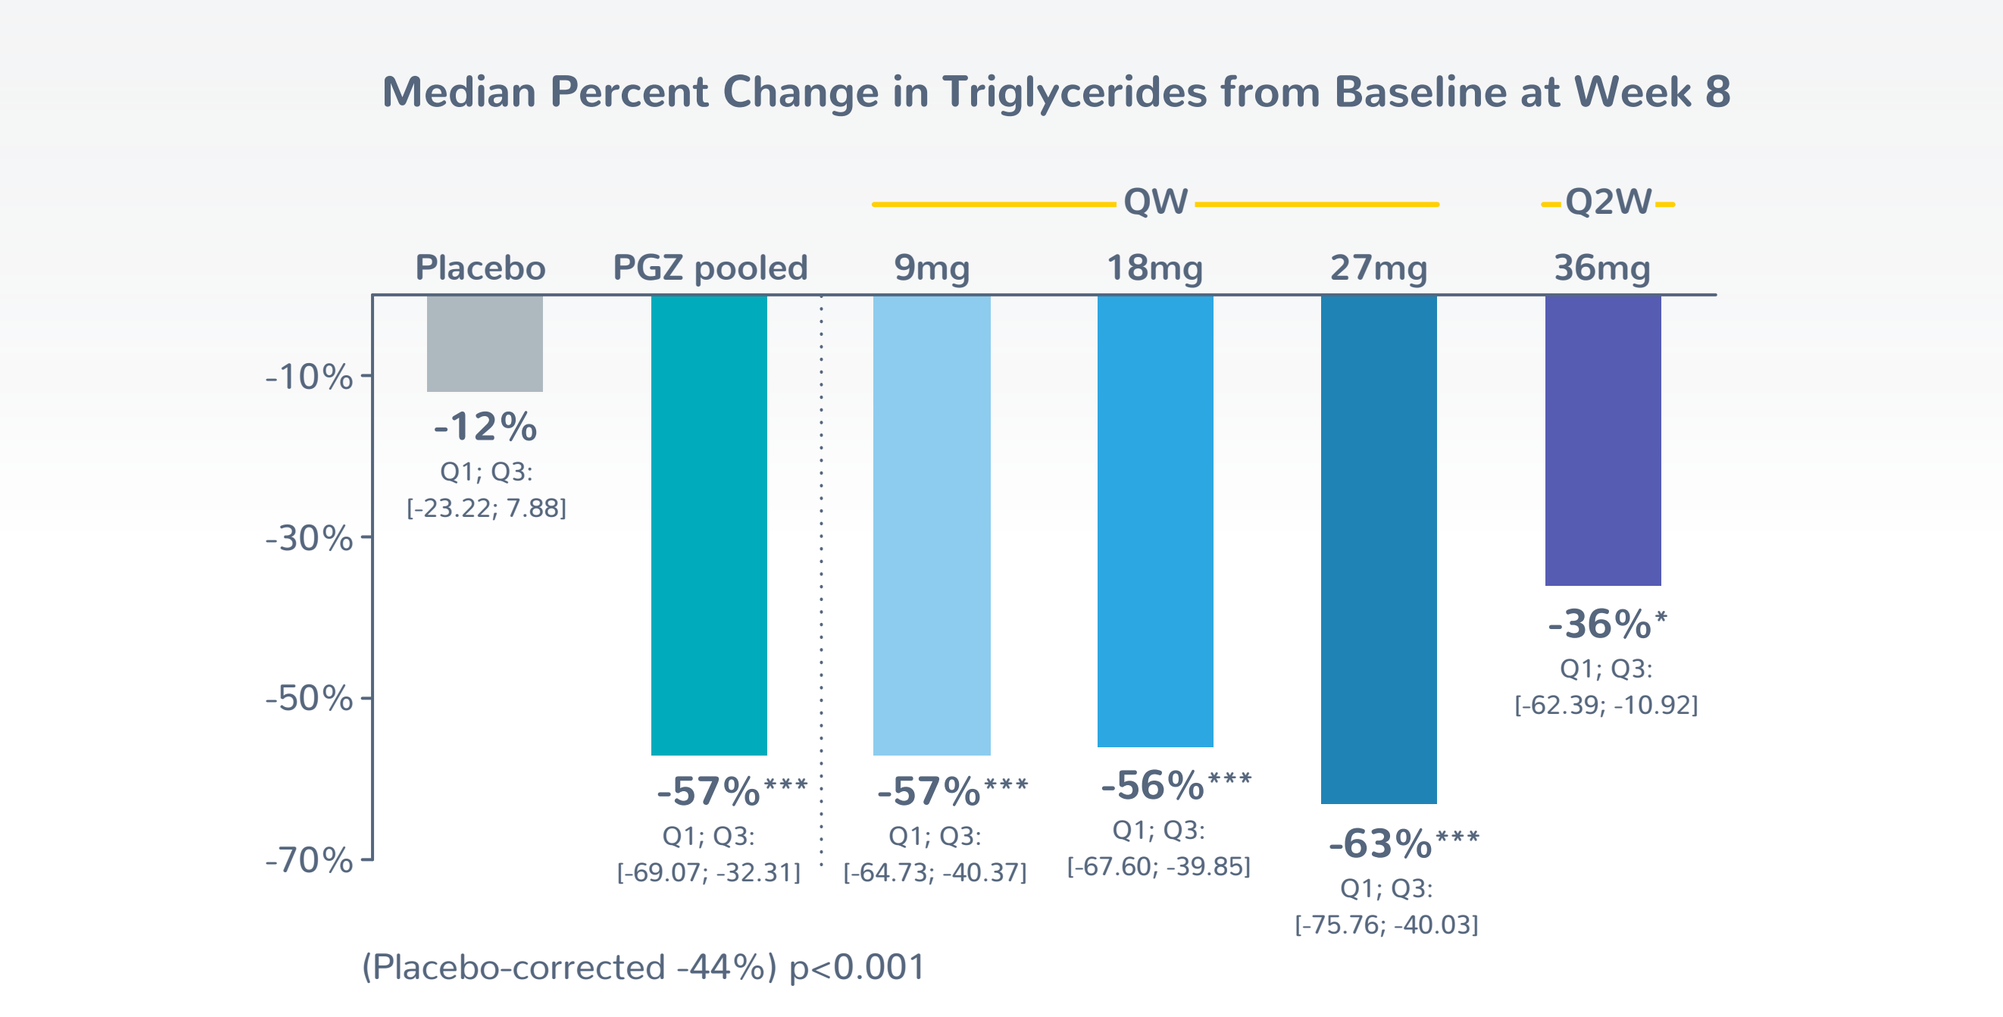
Median Percent Change in Triglycerides from Baseline at Week 8
QW
Q2W
Placebo
PGZ pooled
9mg
18mg
27mg
36mg
-10%
-30%
-50%
-70%
-12%
Q1; Q3: [-23.22; 7.88]
-57%***
Q1; Q3: [-69.07; -32.31]
-57%***
Q1; Q3: [-64.73; -40.37]
-56%***
Q1; Q3: [-67.60; -39.85]
-63%***
Q1; Q3: [-75.76; -40.03]
-36%*
Q1; Q3: [-62.39; -10.92]
(Placebo-corrected -44%) p<0.001

QW: Every week; Q2W: Every 2 weeks
Van Elteren Test for pooled PGZ; Wilcoxon Rank Sum for individual PGZ groups
Full Analysis Set; * p<0.05; *** p<0.001 versus placebo

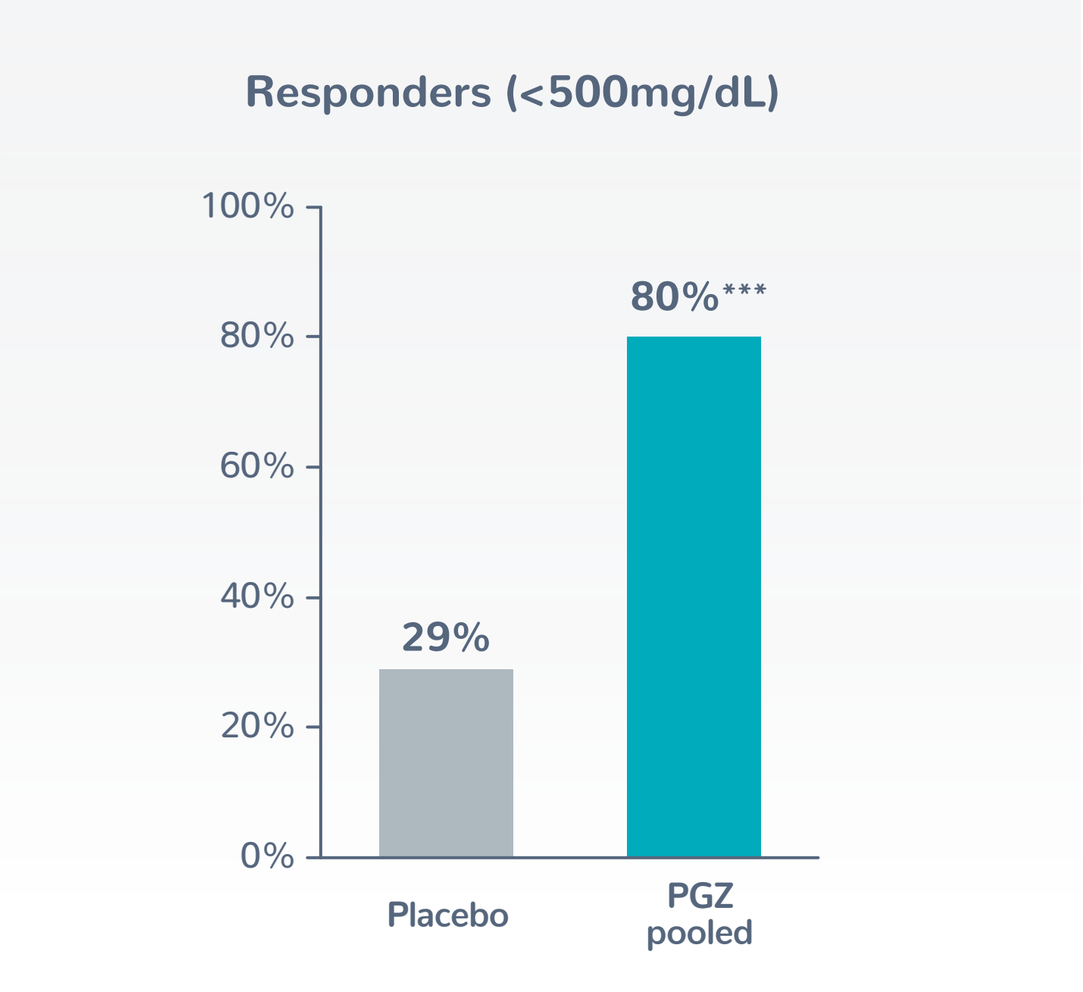
Responders (<500mg/dL)
100%
80%
60%
40%
20%
0%
29%
80%***
Placebo
PGZ pooled

Analysis via Chi-square test comparing PGZ vs placebo
Full Analysis Set; * p<0.05; ** p<0.01; *** p<0.001
TG Responders defined as patients who achieve TG <500 mg/dL

PGZ Treatment Led to a Significant Reduction in Triglycerides Independent of Background LMT or T2DM Status

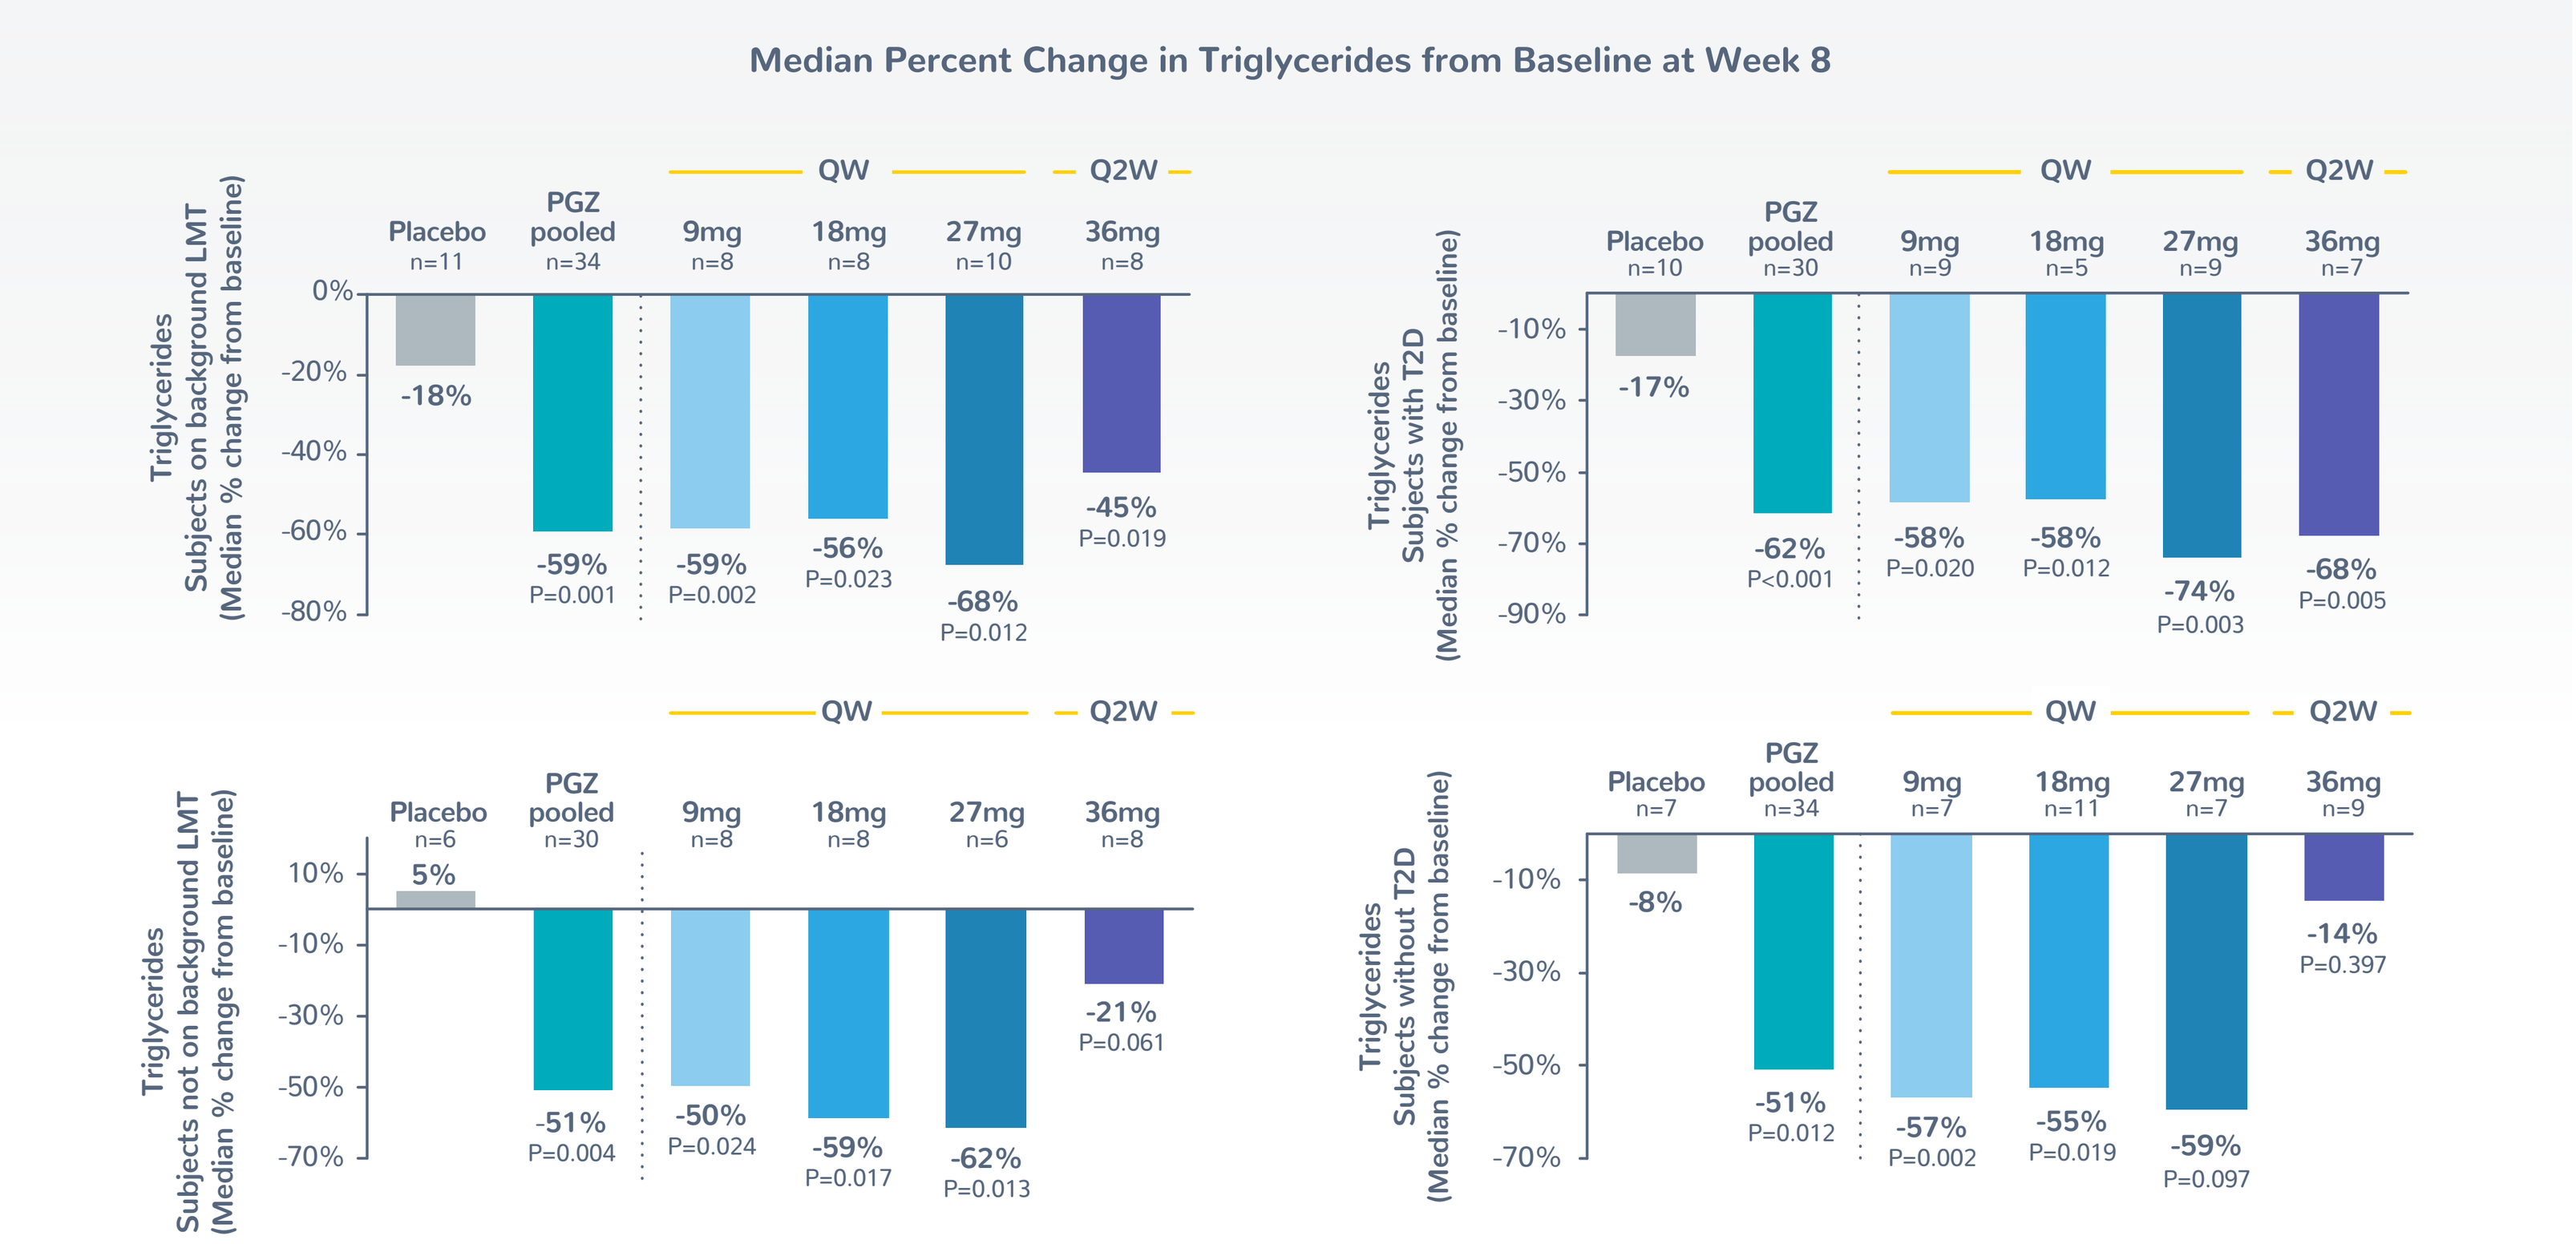
Median Percent Change in Triglycerides from Baseline at Week 8
Triglycerides Subjects on background LMT (Median % change from baseline)
Placebo n=11: -18%
PGZ pooled n=34: -59% P=0.001
9mg n=8: -59% P=0.002
18mg n=8: -56% P=0.023
27mg n=10: -68% P=0.012
36mg n=8: -45% P=0.019
Triglycerides Subjects with T2D (Median % change from baseline)
Placebo n=10: -17%
PGZ pooled n=30: -62% P<0.001
9mg n=9: -58% P=0.020
18mg n=5: -58% P=0.012
27mg n=9: -74% P=0.003
36mg n=7: -68% P=0.005
Triglycerides Subjects not on background LMT (Median % change from baseline)
Placebo n=6: 5%
PGZ pooled n=30: -51% P=0.004
9mg n=8: -50% P=0.024
18mg n=8: -59% P=0.017
27mg n=6: -62% P=0.013
36mg n=8: -21% P=0.061
Triglycerides Subjects without T2D (Median % change from baseline)
Placebo n=7: -8%
PGZ pooled n=34: -51% P=0.012
9mg n=7: -57% P=0.002
18mg n=11: -55% P=0.019
27mg n=7: -59% P=0.097
36mg n=9: -14% P=0.397
QW
Q2W

PGZ Significantly Decreases Triglycerides Across Various Threshold Levels

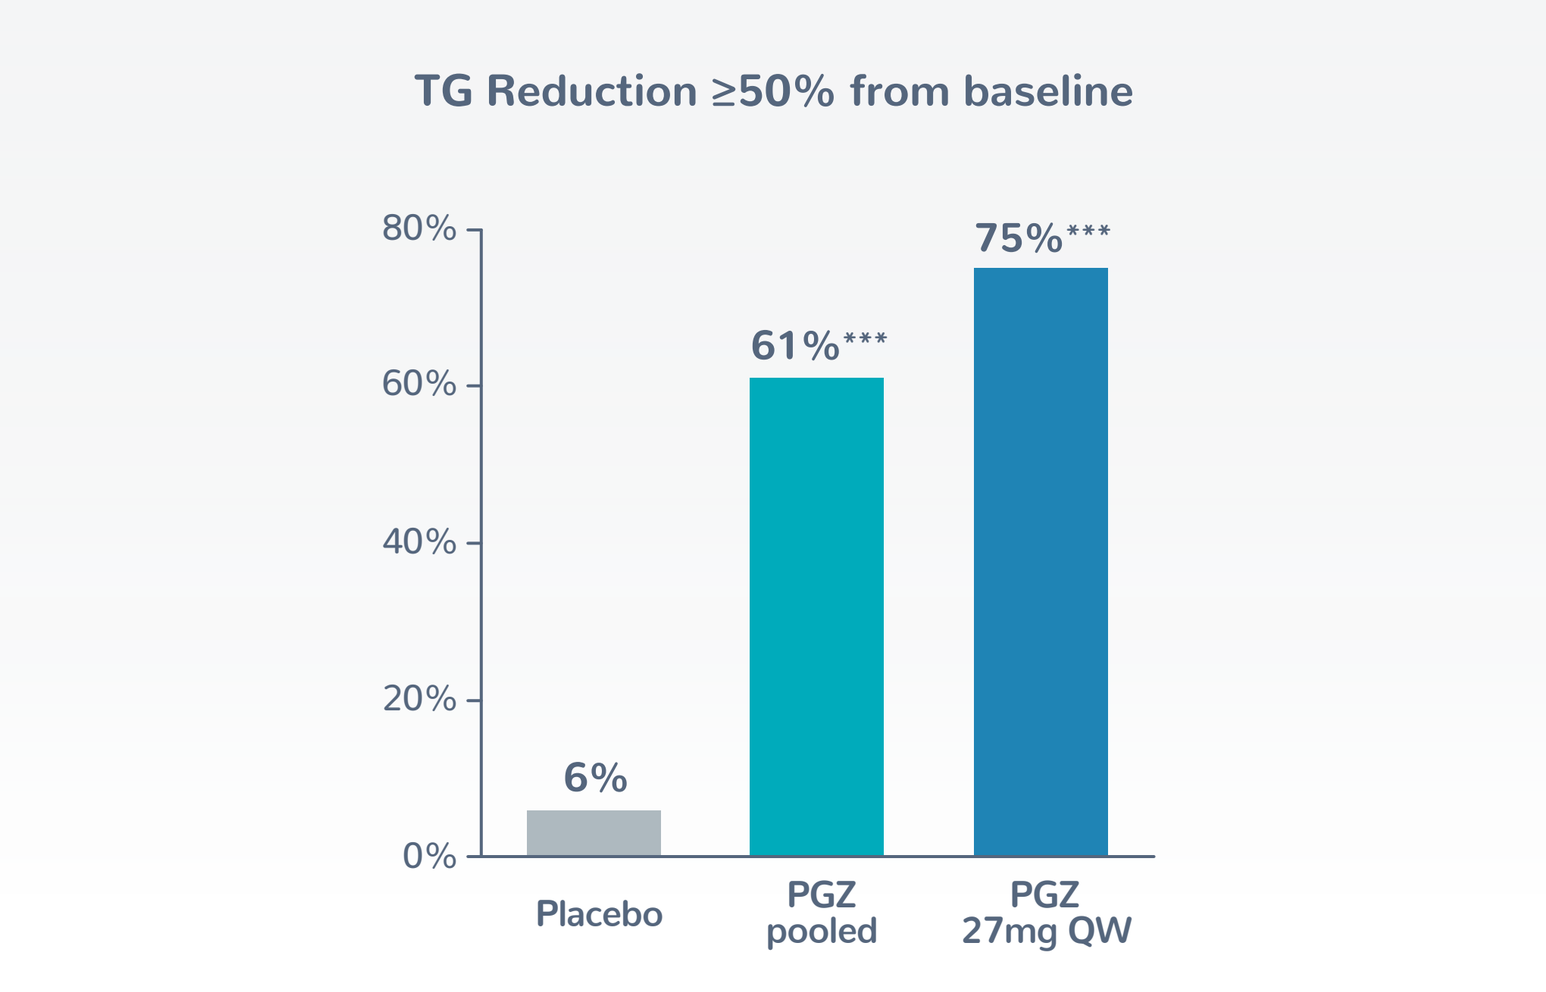
TG Reduction ≥50% from baseline
80%
60%
40%
20%
0%
6%
61%***
75%***
Placebo
PGZ pooled
PGZ 27mg QW

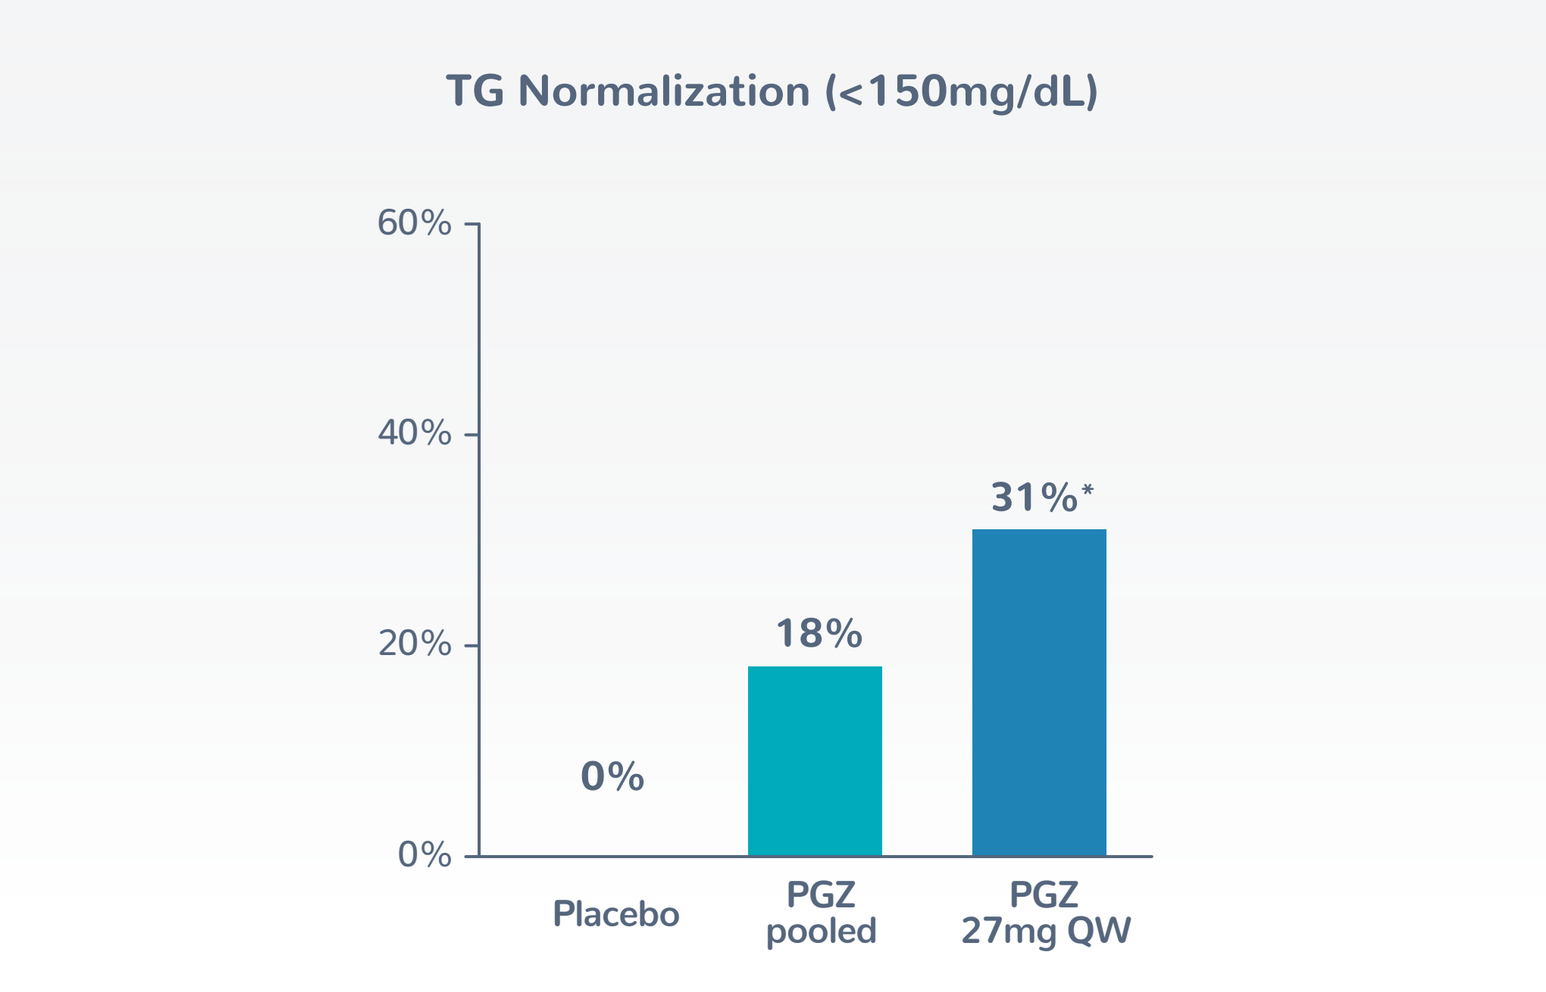
TG Normalization (<150mg/dL)
60%
40%
20%
0%
0%
18%
31%*
Placebo
PGZ pooled
PGZ 27mg QW

Analysis via CMH and Chi-square test comparing the pooled and individual PGZ groups vs placebo respectively
Full Analysis Set; * p<0.05; ** p<0.01; *** p<0.001

PGZ Demonstrated Reduction in Atherogenic Lipid Particles (Non-HDL-C and ApoB)

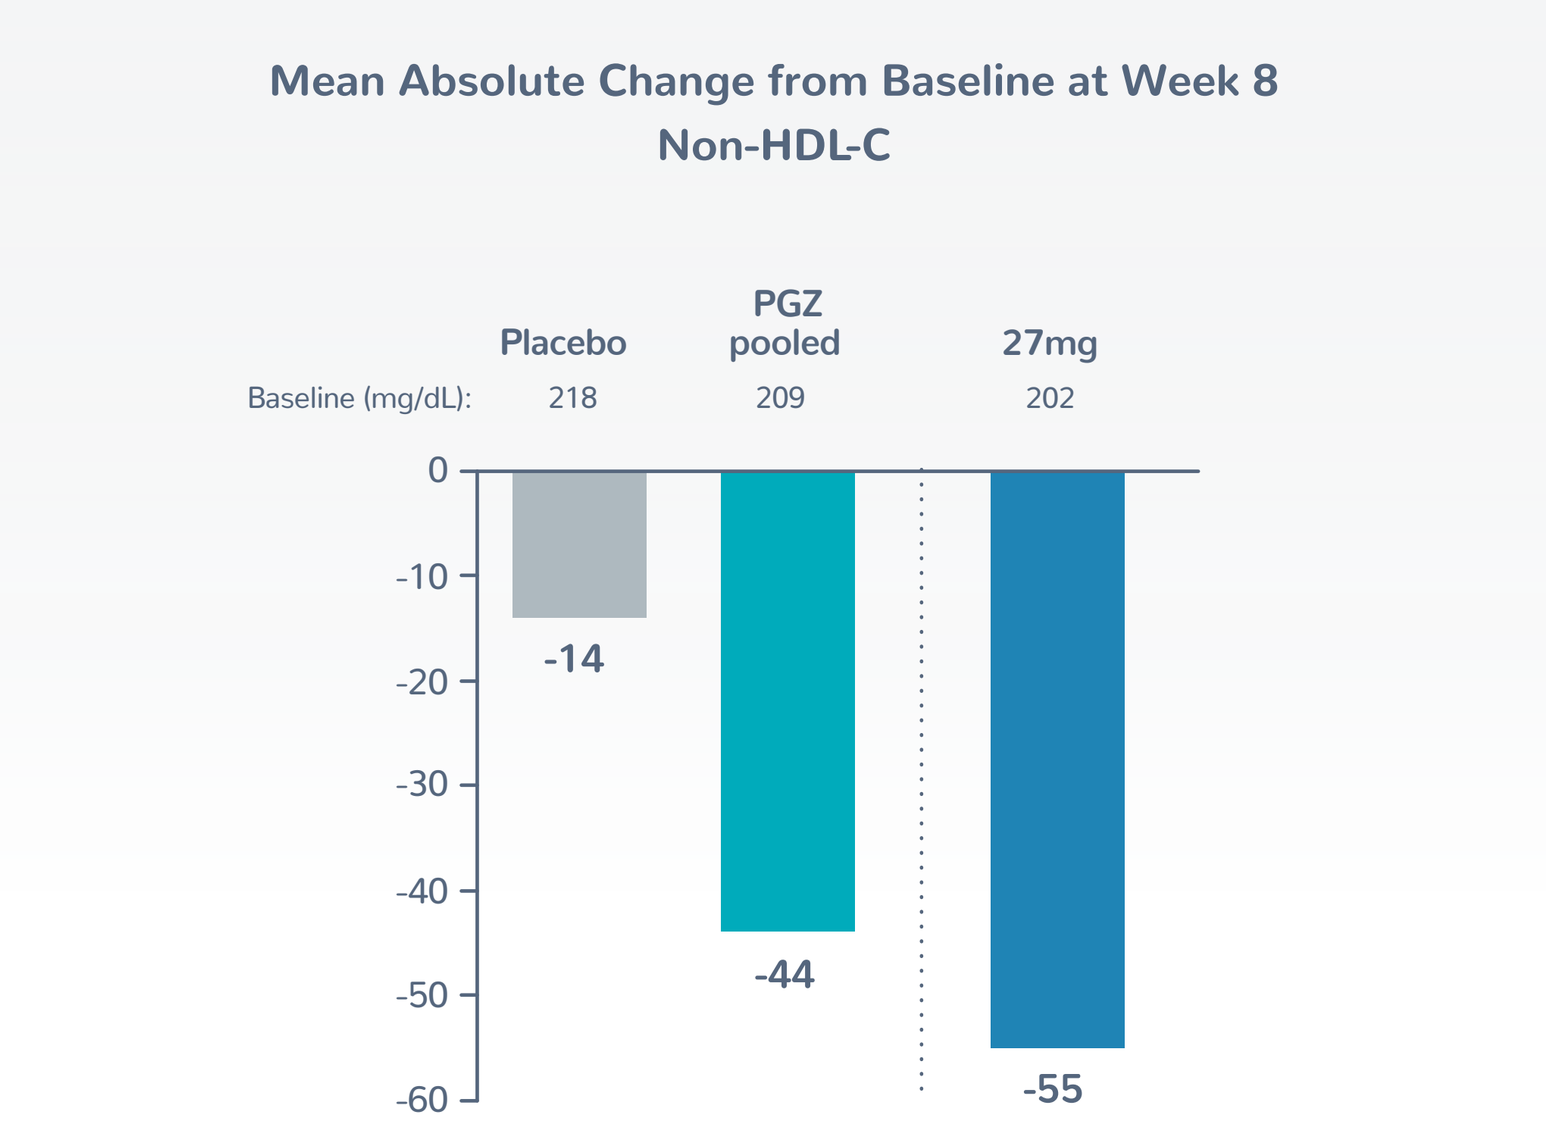
Mean Absolute Change from Baseline at Week 8
Non-HDL-C
Baseline (mg/dL):
Placebo 218
PGZ pooled 209
27mg 202
-14
-44
-55

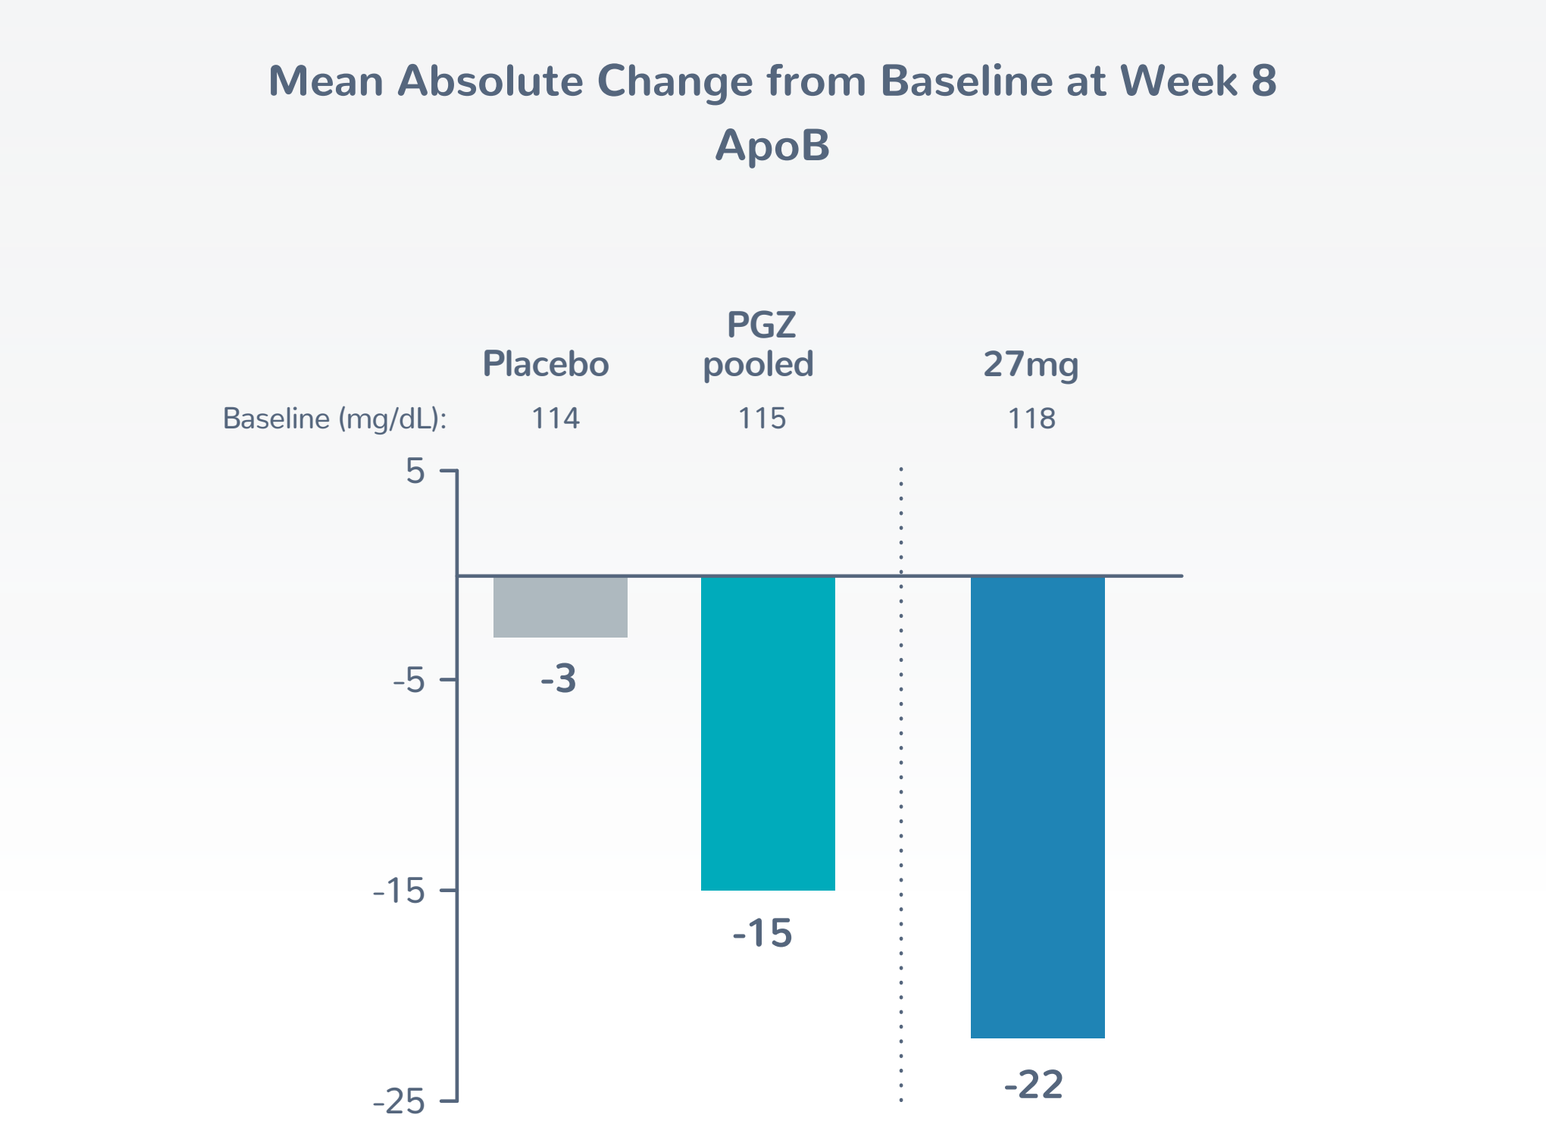
Mean Absolute Change from Baseline at Week 8
ApoB
Baseline (mg/dL):
Placebo 114
PGZ pooled 115
27mg 118
-3
-15
-22

Descriptive analysis was performed for mean absolute change

PGZ Decreases ApoC3 and Increases HDL-C with Minimal Impact on LDL-C

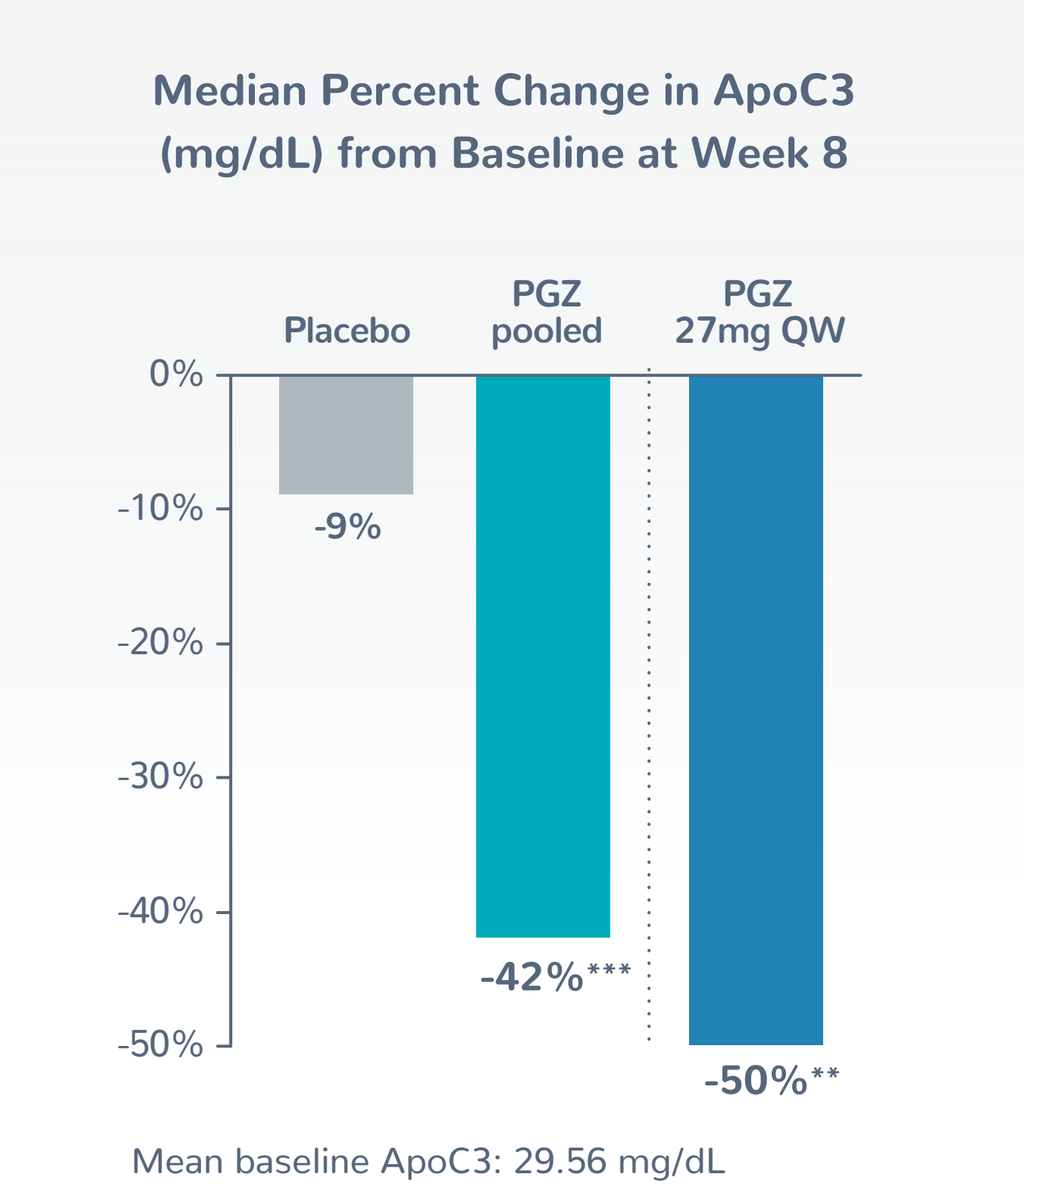
Median Percent Change in ApoC3 (mg/dL) from Baseline at Week 8
Placebo
PGZ pooled
PGZ 27mg QW
-9%
-42%***
-50%**
Mean baseline ApoC3: 29.56 mg/dL

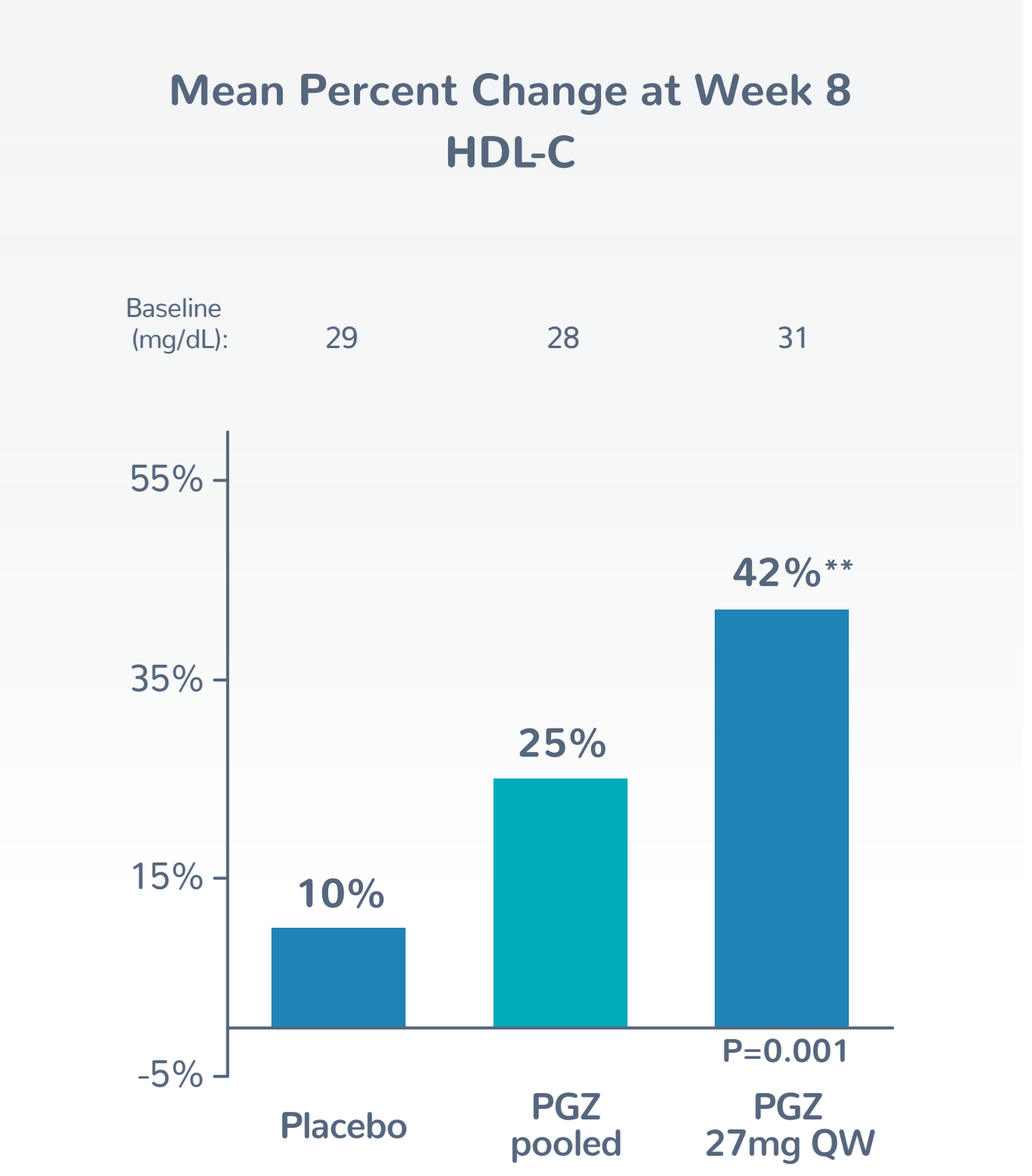
Mean Percent Change at Week 8
HDL-C
Baseline (mg/dL): 29 28 31
10%
25%
42%**
P=0.001
Placebo
PGZ pooled
PGZ 27mg QW

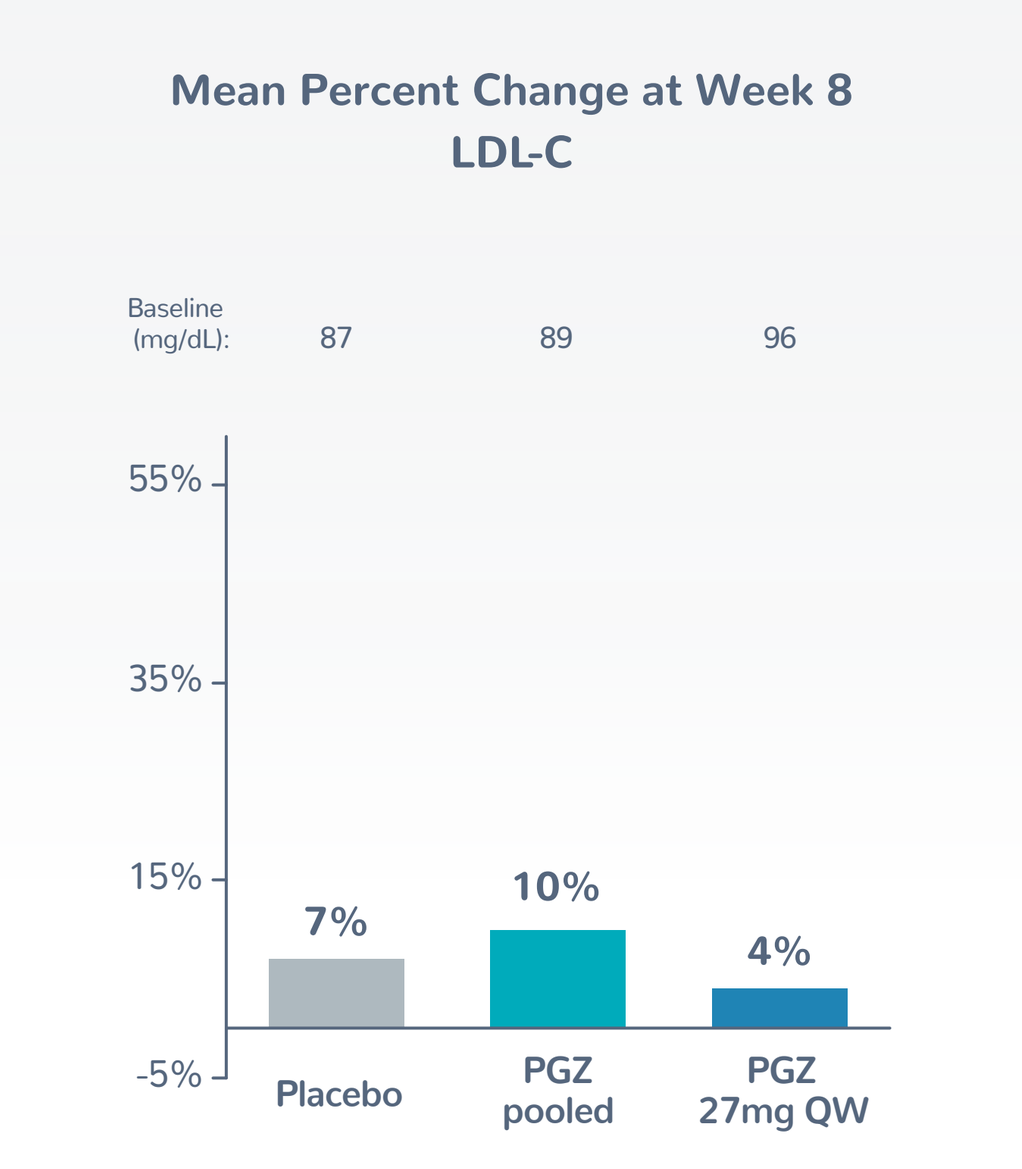
Mean Percent Change at Week 8
LDL-C
Baseline (mg/dL): 87 89 96
7%
10%
4%
Placebo
PGZ pooled
PGZ 27mg QW

Full Analysis Set; ** p<0.01; *** p<0.001; Van Elteren Test for pooled PGZ; Wilcoxon Rank-sum for individual PGZ groups

Full Analysis Set; ** p=0.01 versus placebo based on MMRM analysis

PGZ Demonstrated Significant Reduction in Liver Fat

Liver fat is an important potentiator of cardiovascular risk

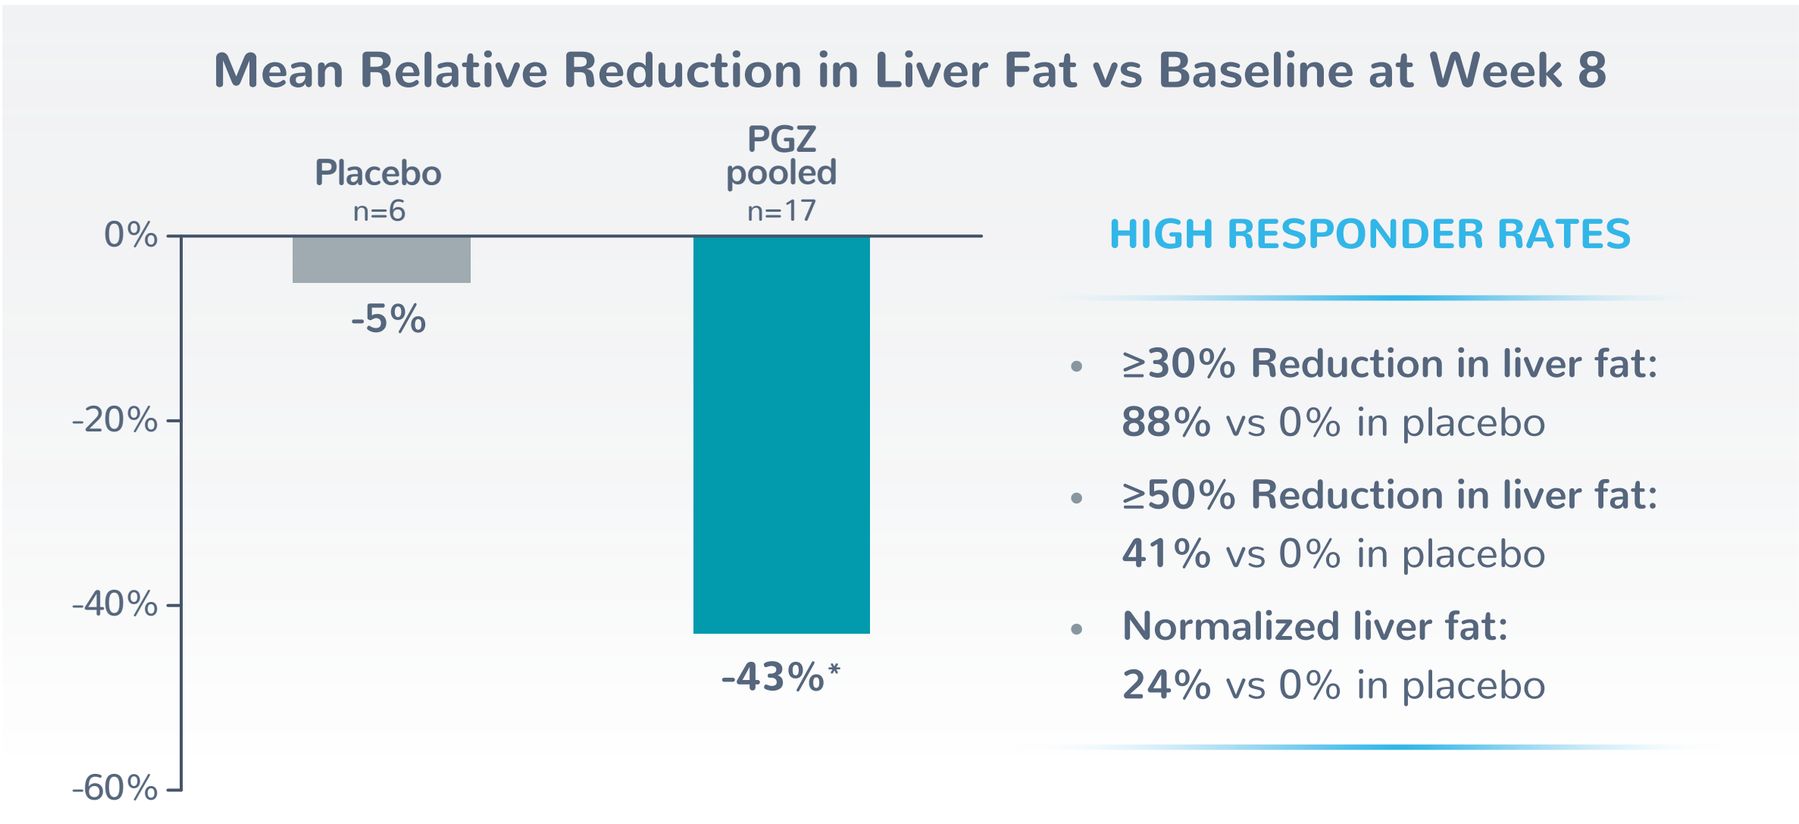
Mean Relative Reduction in Liver Fat vs Baseline at Week 8
Placebo n=6
PGZ pooled n=17
-5%
-43%*
HIGH RESPONDER RATES
≥30% Reduction in liver fat: 88% vs 0% in placebo
≥50% Reduction in liver fat: 41% vs 0% in placebo
Normalized liver fat: 24% vs 0% in placebo

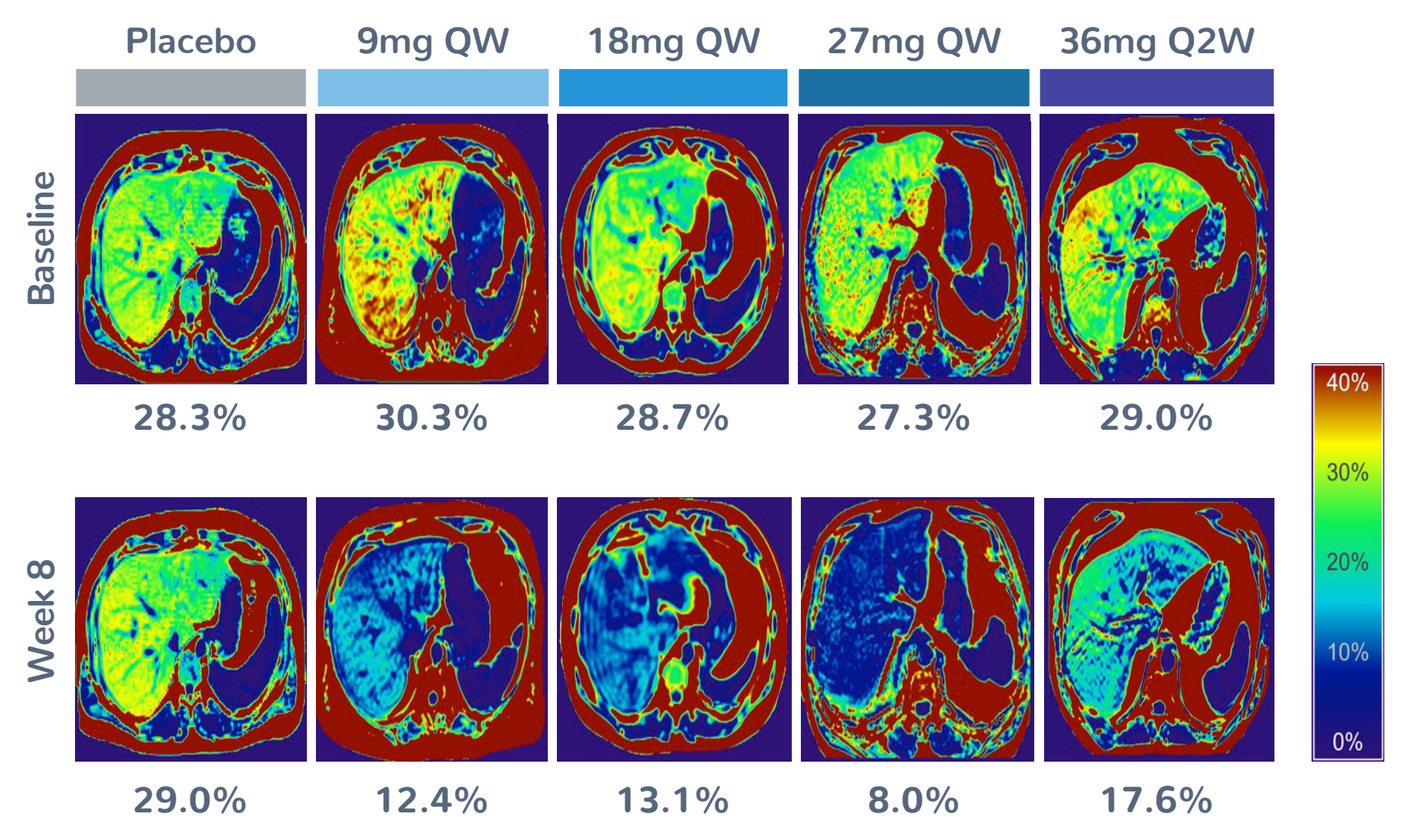
Placebo
9mg QW
18mg QW
27mg QW
36mg Q2W
Baseline
28.3%
30.3%
28.7%
27.3%
29.0%
Week 8
29.0%
12.4%
13.1%
8.0%
17.6%

MRI Analysis Set; p value vs placebo based on ANCOVA analysis
*p <0.05 vs placebo

Higher Dose of PGZ Improves Markers of Insulin Sensitivity

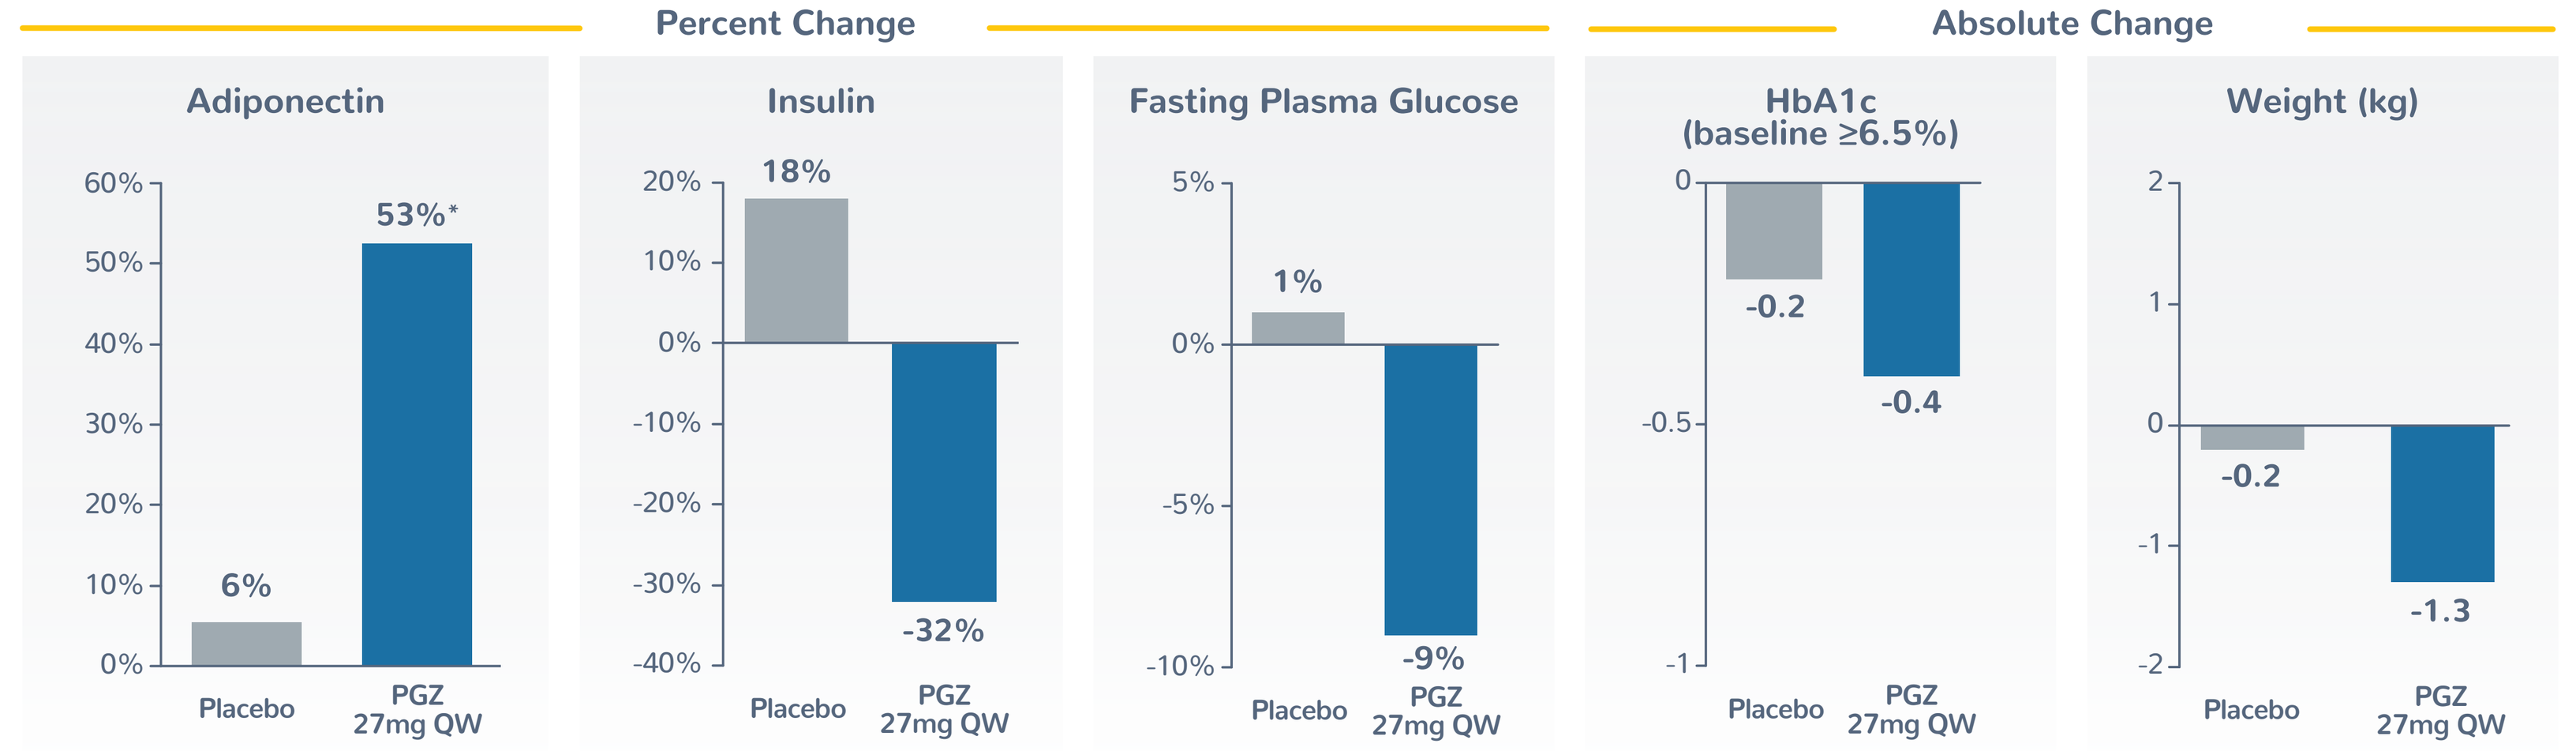
Percent Change
Absolute Change
Adiponectin: Placebo 6%; PGZ 27mg QW 53%*
Insulin: Placebo 18%; PGZ 27mg QW -32%
Fasting Plasma Glucose: Placebo 1%; PGZ 27mg QW -9%
HbA1c (baseline ≥6.5%): Placebo -0.2; PGZ 27mg QW -0.4
Weight (kg): Placebo -0.2; PGZ 27mg QW -1.3

Full Analysis Set; Descriptive analysis
Mean Changes at Week 8; *p=0.017

SAFETY

PGZ Was Well Tolerated Across Doses

Low incidence of treatment-related AEs in ≥7.5% of pooled PGZ group.

	Placebo (n=18)	PGZ Pooled (n=67)	PGZ 9mg QW (n=16)	PGZ 18mg QW (n=17)	PGZ 27mg QW (n=18)	PGZ 36mg Q2W (n=16)
Nausea	0	10%	0%	5%	22%	13%
Diarrhea	0	9%	17%	5%	17%	13%
Injection site reaction	0	9%	8%	10%	6%	13%

All AEs were Grade 1 or 2; No Grade 3 or higher TEAEs reported.
No transaminase elevation AEs reported.

	Placebo (n=18)	PGZ Pooled (n=67)	PGZ 9mg QW (n=16)	PGZ 18mg QW (n=17)	PGZ 27mg QW (n=18)	PGZ 36mg Q2W (n=16)
Serious adverse event (unrelated)	0	1*	0	0	1	0
Treatment emergent discontinuations (related/ unrelated)	0	2^/2	0	0	2^/2	0

*Unrelated SAE of Grade 2 hypertension in a subject with undiagnosed, uncontrolled hypertension at baseline; patient withdrew
^Grade 2 abdominal cramps (1) and Grade 2 nausea/vomiting (1)
Safety Analysis Set; patients reported on as treated basis

CONCLUSIONS

- PGZ significantly reduced:
 - Triglycerides by ˜50-60%
 - Non-HDL-Cholesterol by ˜20-30%
 - ApoB by ˜10-20%
 - Liver fat fraction by ˜40%
- Additional cardiometabolic improvements potentially make pegozafermin an attractive therapy in SHTG to address multiple co-morbidities simultaneously, including cardiac, glycemic, and hepatic risks.
- These data appear very promising for the planned Phase 3 trial utilizing the higher weekly dose(s) given for a longer duration.
- A randomized double-blind placebo-controlled phase 2b study in NASH is fully enrolled with data expected in 1Q23 (NCT04929483).

© 2022 89bio, Inc.